Optimizing a Multicolor Flow Cytometry Panel Using the CytoFLEX mosaic Spectral Detection Module for Immune Cell Analysis

Fanuel Messaggio[1], Anisha Jose[2], Krittika Ralhan[2]
[1]Beckman Coulter Life Sciences, Miami, FL 33196.
[2]Beckman Coulter Life Sciences India Pvt Ltd, Karnataka, 562149.

INTRODUCTION

The CytoFLEX mosaic Spectral Detection Module enhances CytoFLEX instruments by integrating conventional and spectral detection methods. This module enables the detection of a broader range of markers and cell subpopulations, making it ideal for research with limited samples. A comprehensive flow cytometry panel has been designed to immunophenotype immune cell subsets, focusing on T-cell activation and differentiation. The panel includes 22 colors and 23 markers for a CytoFLEX mosaic 88 Spectral Detection Module paired with a CytoFLEX LX Flow Cytometer. It features overlapping fluorophores like APC and Alexa 647 and introduces new IR dyes, IR820 and IR870, to enhance performance.

METHODS

- Prepared 23 flow tubes: 22 single-color controls and one fully stained mix.
- Added antibodies to each tube, starting with SuperNova Buffer.
- Used whole blood cells as samples, lysed with Red Blood Cell Lysis Buffer pre-staining.
- Incubated cells with Fc blocker for 20 minutes in the dark.
- Added monocyte blocker without further incubation.
- Transferred cells per tube, vortexed, and incubated for 30 minutes at room temperature in the dark.
- Washed cells twice with staining buffer, centrifuged, decanted supernatant, and resuspended in staining buffer.
- For Versa-Comp Single-Color Staining: prepared 23 tubes, each with a drop of positive beads and the correspondent antibody.
- Incubated beads for 15 minutes at room temperature in the dark.
- Washed beads twice with staining buffer, centrifuged, decanted supernatant, and resuspended in staining buffer.

GATING STRATEGY

The antibodies used are displayed in Table 1. The gating strategy is based on the principles of removing noise signals from debris, discriminating doublets, and differentiating debris from CD45+ cells (all leukocytes) as illustrated in Figure 2. CD45+ cells are further differentiated into granulocytes, monocytes, and lymphocytes (B lymphocytes and T lymphocytes). An FSC-A vs. FSC-H pseudo color plot is employed to exclude cell doublets or aggregates, followed by SSC-A vs FSC-A plot to eliminate cell debris, which typically shows decreased forward scatter and increased side scatter. CD45+ white blood cell subsets are highlighted using a CD45 vs SSC-A dot plot. Monocytes are gated using the CD14 vs CD16 plot, while neutrophils and eosinophils are identified using the CD16 vs CD45 PerCP plot. Lymphocytes are further subdivided into CD3+CD4+ T cells using the HLA-DR vs CD11b plot. NK cells are gated out using the CD56 vs CD16 plot. This strategy ensures precise identification and analysis of various leukocyte subsets.

Marker	Clone	Fluorochrome	Vol	Catalog # (US Only)	Catalog # (OUS Only)	Function	Company
CD27	L128	BUV395	5 µL	563816	563816	Phenotyping of T cells, activation	BD
CD33	D3HL60.251	SNv428	5 µL	C76819	B30632	Monocytes	Beckman
CD19	B49213	PacBlue	5 µL	A86355	C78087	Leniage marker of B cells	Beckman
CD4	13B8.2	PacBlue	5 µL	A82789	A71116	Leniage marker for CD4+ T cells	Beckman
CD8	B9.11	KrO	5 µL	B00067	B36123	Lineage marker for CD8+ T cells	Beckman
CD38	LS198-4.3	SNv605	5 µL	C78077	B49197	Phenotyping of T cells and NK cells, activation	Beckman
CD25	B1.49.9	SNv650	5 µL	D05542	A64617	Phenotyping of T cells, Treg identification	Beckman
HLA-DR	IMMU357	SNv786	3 µL	C78087	C78077	Lineage marker for gd T cells	Beckman
CD183 (CXCR3)	GO25H7	AF488	5 µL	B68144	B68132	Phenotyping of T cells, migration	Beckman
CD45RO	UCHL1	RB545	5 µL	569778	569778	Phenotyping of T cells, naive versus memory	BD
CD16	3G8	ECD	5 µL	A33098	B49213	Lineage marker of monocytes	Beckman
CD45	2D1	PerCP	5 µL	368506	368506	Pan-Hematopoietic marker	BioLegend
CD197 (CCR7)	G043H7	PE	10 µL	B30632	B68144	Phenotyping of T cells, naive versus memory	Beckman
CD11b	ICRF44	AF594	5 µL	301340	301340	Phenotyping of DCs and monocytes	BioLegend
CD127	R34.34	AF594	5 µL	A64617	C76819	Phenotyping of T cells, Treg identification	Beckman
CD279 (PD-1)	PD.1	PC5.5	5 µL	B36123	B49216	Phenotyping of T cells, activation and	Beckman
CD196 (CCR6)	B-R35	PC7	5 µL	B68132	C86899	B lymphocytes to some extent to T lymphocyte	Beckman
CD23	9P25	APC	5 µL	A69964	B49186	B lymphocytes and monocytes	Beckman
CD56	1H11	AF647	5 µL	362513	362513	NK cells	BioLegend
CD45RA	2H4LDH11LD89	AF700	5 µL	B90396	C86914	Phenotyping of T cells, naive versus memory	Beckman
CD3	UCHT1	APC-A750	5 µL	A66329	A94680	Lineage marker for pan T cells	Beckman
CD20		IR820	10 µL	NPD	NPD	Lineage marker of B cells	Beckman
CD14		IR870	10 µL	NPD	NPD	Lineage marker of monocytes	Beckman

Table 1. Antibody Panel Details.

RESULTS

Similarity Matrix

	Krome Orange	SNv786	SNv605	SNv428	RB545	PerCP	PE-Cy7	PE-Cy5.5	PE-Cy5	PE	Pacific Blue	IR870	IR820	ECD	BV650	BUV395	APC-AF750	APC	Alexa fluor 6...	Alexa Fluor 5...	Alexa Fluor 4...	A700	Unstained_lymph
Unstained_lymph	0.48	0.05	0.16	0.19	0.21	0.06	0	0.05	0.03	0.06	0.21	0.1	0.01	0.05	0.03	0.38	0.01	0.02	0.02	0.04	0.21	0.01	1
A700	0.01	0.07	0.07	0	0	0.14	0.06	0.26	0.21	0.01	0	0.11	0.02	0.05	0.13	0	0.37	0.42	0.46	0.07	0	1	
Alexa Fluor 488	0.03	0	0.01	0.01	0.64	0.01	0.01	0.01	0.01	0.04	0.01	0.05	0.01	0.02	0.01	0.01	0	0	0	0	1		
Alexa Fluor 594	0.07	0.02	0.55	0	0.03	0.4	0.13	0.38	0.6	0.51	0	0.11	0.01	0.94	0.23	0	0.04	0.26	0.09	1			
Alexa fluor 647	0	0.01	0.04	0	0	0.28	0.02	0.16	0.34	0.02	0	0.07	0.02	0.06	0.16	0	0.22	0.88	1				
APC	0	0.02	0.17	0.01	0	0.33	0.04	0.22	0.49	0.06	0	0.09	0.02	0.19	0.27	0.02	0.23	1					
APC-AF750	0	0.17	0.04	0	0	0.07	0.23	0.1	0.1	0.01	0	0.18	0.08	0.03	0.06	0	1						
BUV395	0.04	0	0.01	0.06	0.01	0	0.01	0	0	0	0.02	0.03	0	0	0.02	1							
BV650	0.12	0.11	0.76	0.07	0	0.5	0.02	0.12	0.2	0.04	0.04	0.07	0.01	0.13	1								
ECD	0.04	0.01	0.44	0	0.08	0.37	0.13	0.38	0.57	0.57	0	0.11	0.01	1									
IR820	0.03	0.14	0.02	0.04	0.01	0.01	0.02	0.01	0.01	0	0.06	0.6	1										
IR870	0.1	0.14	0.11	0.1	0.07	0.11	0.09	0.12	0.14	0.06	0.13	1											
Pacific Blue	0.22	0.09	0.05	0.73	0.01	0	0	0	0	0	1												
PE	0.07	0	0.22	0	0.13	0.1	0.15	0.12	0.18	1													
PE-Cy5	0.01	0.02	0.32	0	0.05	0.71	0.13	0.71	1														
PE-Cy5.5	0.01	0.03	0.23	0	0.04	0.49	0.23	1															
PE-Cy7	0.01	0.1	0.08	0	0.02	0.08	1																
PerCP	0.05	0.07	0.49	0	0.03	1																	
RB545	0.09	0	0.03	0.01	1																		
SNv428	0.11	0.13	0.07	1																			
SNv605	0.3	0.13	1																				
SNv786	0.03	1																					
Krome Orange	1																						

Figure 1: Similarity Index. The Similarity Index of the fluorochromes used in this panel resulted in a Complexity Score of 11.0. This was generated using CytExpert for Spectral software.

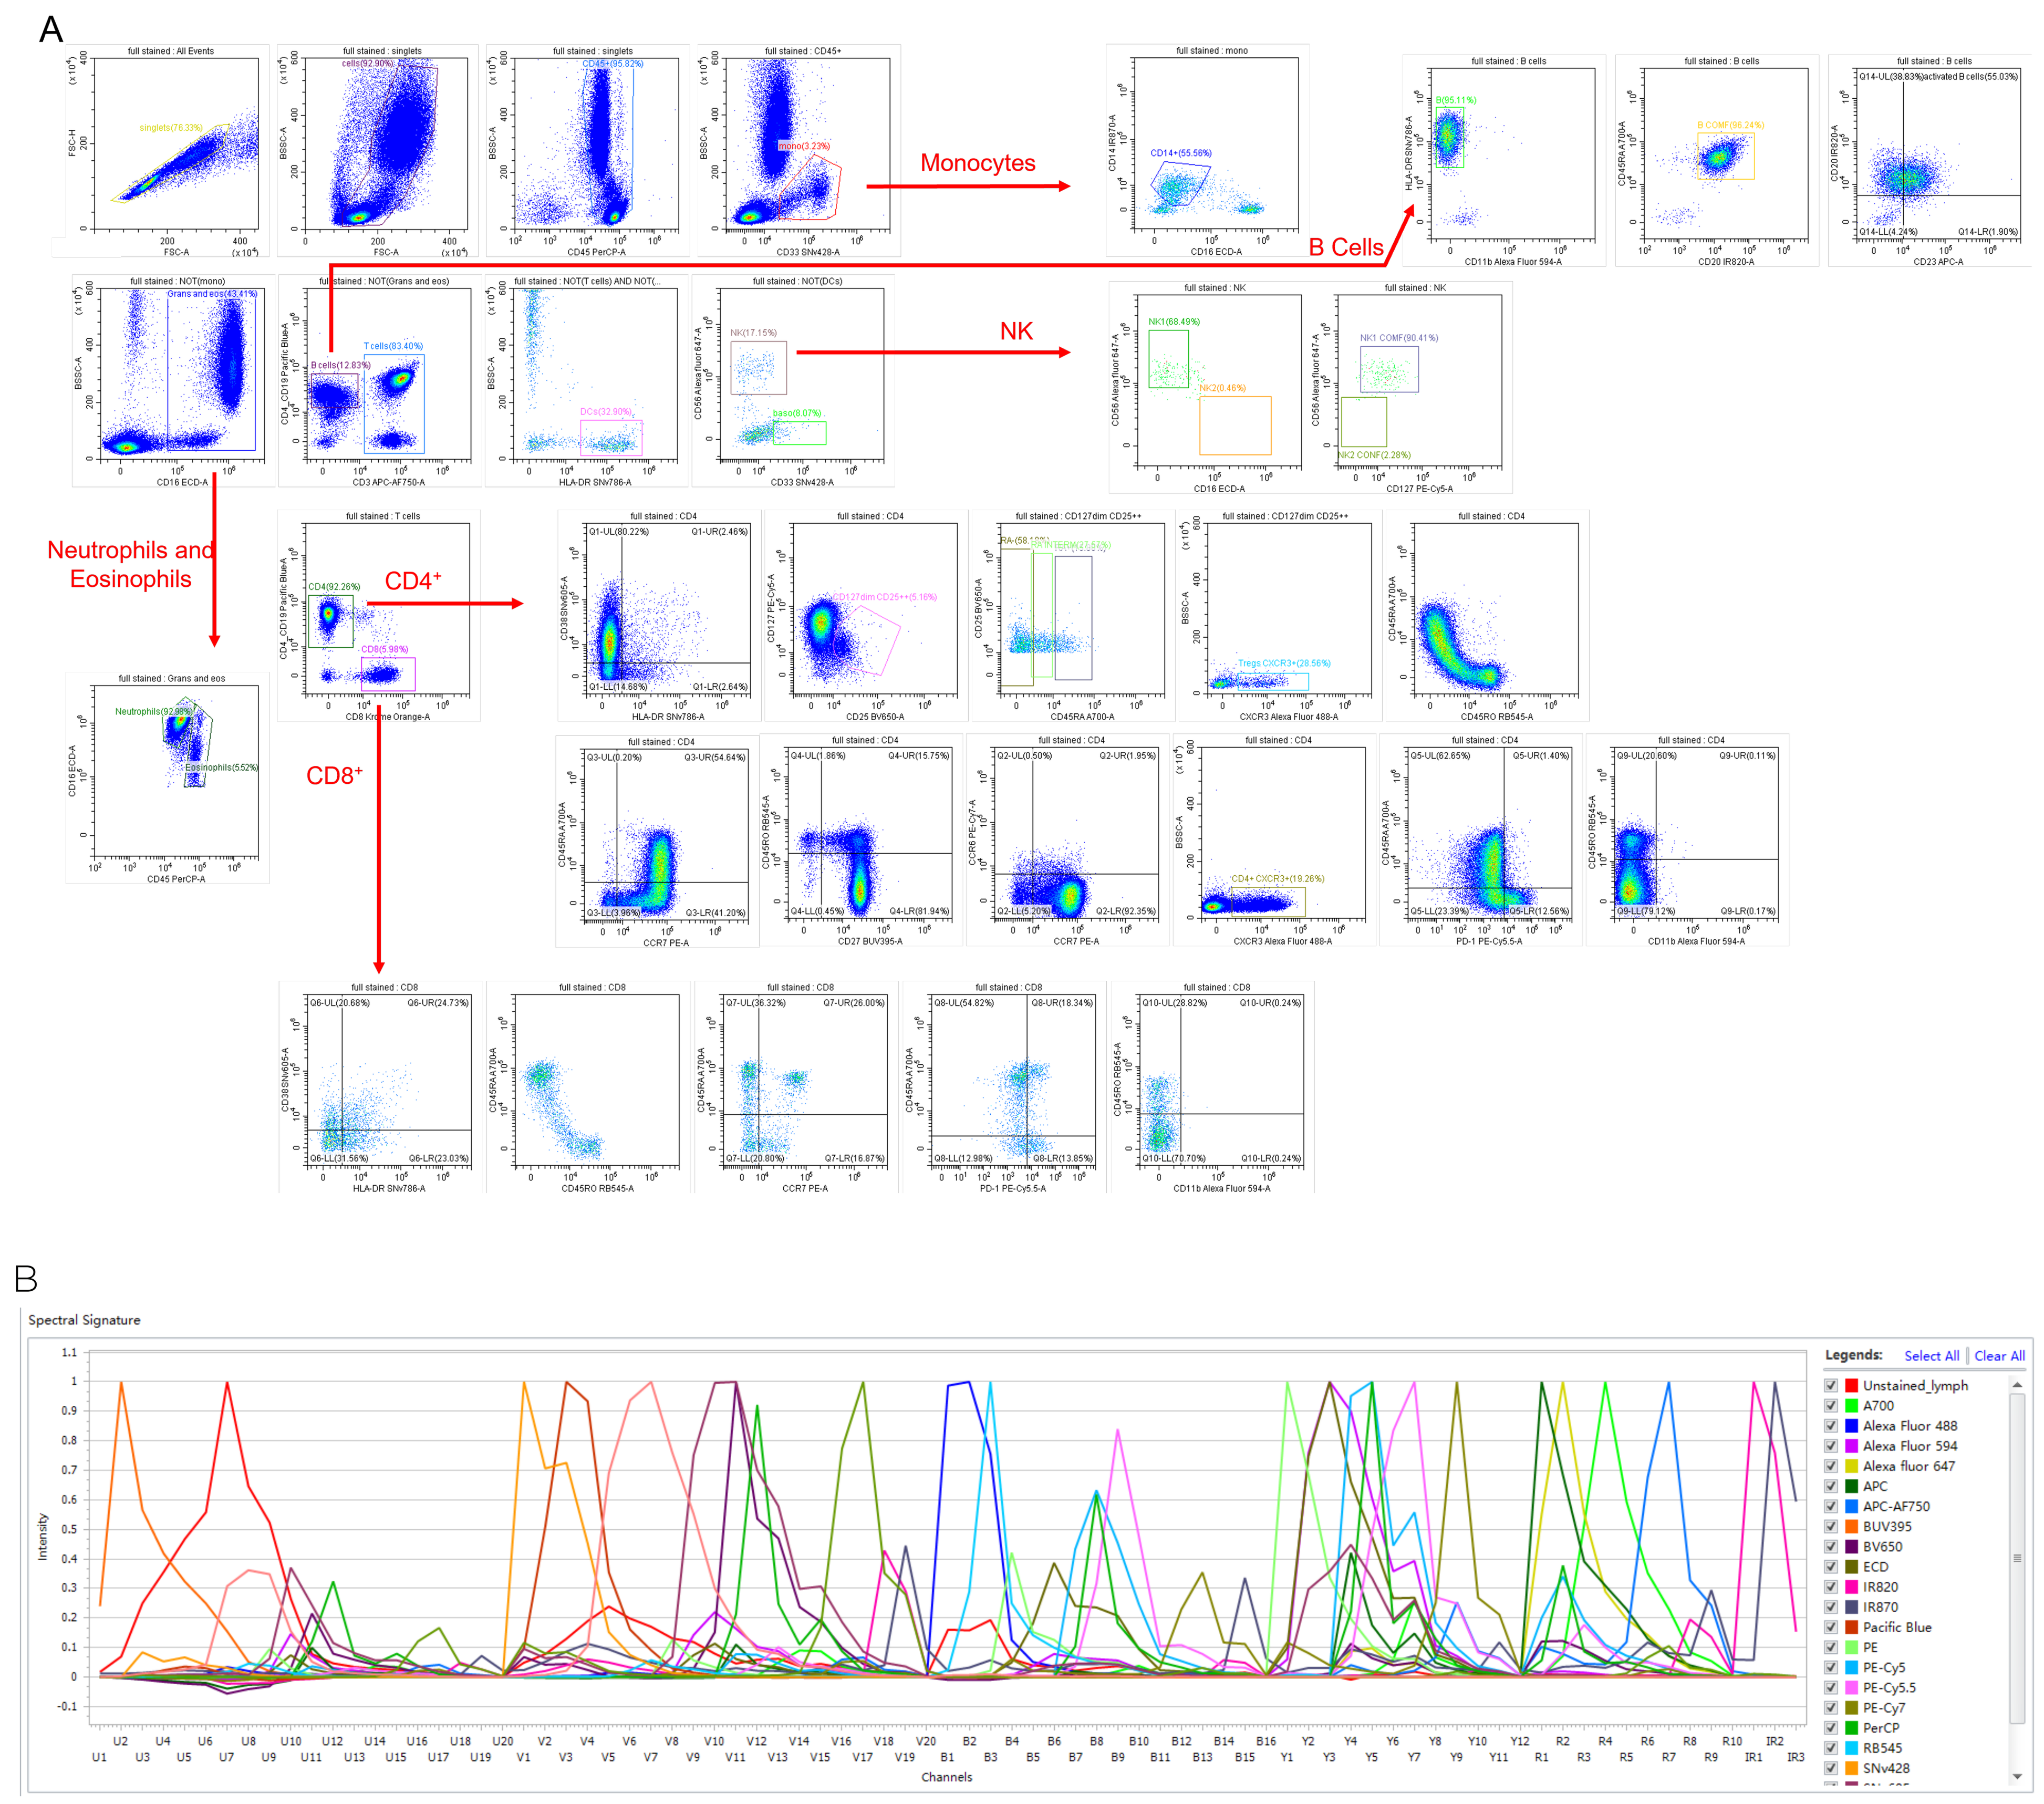
A
Monocytes
B Cells
NK
Neutrophils and Eosinophils
CD4+
CD8+
B
Spectral Signature
Intensity
Channels
Legends: Select All | Clear All
Unstained_lymph
A700
Alexa Fluor 488
Alexa Fluor 594
Alexa fluor 647
APC
APC-AF750
BUV395
BV650
ECD
IR820
IR870
Pacific Blue
PE
PE-Cy5
PE-Cy5.5
PE-Cy7
PerCP
RB545
SNv428

Figure 2: Gating and Spectral Signatures. A) The gating strategy, as explained in the description, ensures the identification of various cell subsets. Data was unmixed using the Poisson hybrid algorithm. B) Fluorescent spectral signature of each fluorochrome post unmixing, including the unstained autofluorescence signature. Spectra were generated using CytExpert for Spectral software.

SUMMARY

The panel demonstrated the capabilities of the CytoFLEX mosaic Spectral Detection Module in managing a 23-color panel. It highlighted key features, such as Poisson hybrid unmixing algorithms that effectively distinguish overlapping colors and a user-friendly setup that streamlines the process. With its autofluorescence extraction and control definition, CytExpert for Spectral software ensured precise spectral experiment unmixing, further simplifying user interaction. We successfully identified all key leukocyte populations and their subsets, as shown in Figure 2A. These features collectively underscore the system's performance and reliability in detailed cell population analysis.

Pacific Blue ("PB") and Alexa Fluor ("AF") are trademarks or registered trademarks of Thermo Fisher Scientific Inc. Brilliant Violet ("BUV"). PerCP is trademark or registered trademark of Becton, Dickinson and Company. PE-Cy5 is a trademark or registered trademark of GE Healthcare.

For Research Use Only. Not for use in diagnostic procedures.
© 2025 Beckman Coulter, Inc. All rights reserved. Beckman Coulter, the stylized logo, and the Beckman Coulter product and service marks mentioned herein are trademarks or registered trademarks of Beckman Coulter, Inc. in the United States and other countries. All other trademarks are the property of their respective owners.

For Beckman Coulter's worldwide office locations and phone numbers, please visit contact us at **beckman.com**
2025-GBL-EN-107751-v2