Comparative analysis of the CytoFLEX nano Flow Cytometer and the CytoFLEX LX Flow Cytometer

Evgenia Kim[1], Vera Tang[2], Samantha Bauchiero[1], Milan Popovic[1], Hoa Boi San[1], Anisha Jose[3], Sergei Gulnik[1]
[1]Research and Development, Beckman Coulter Life Sciences, Miami, FL 33196.
[2]University of Ottawa, Department of Biochemistry, Microbiology and Immunology.
[3]Commercial Excellence, Beckman Coulter Life Sciences.

Introduction

This work aims to demonstrate the performance of the CytoFLEX nano Flow Cytometer and provide a comparative analysis with the conventional CytoFLEX Flow Cytometer. A side-by-side comparison of scatter and fluorescence characteristics will provide insightful information on the beneficial features of the CytoFLEX nano Flow Cytometer as a dedicated instrument designed for nanoparticle characterization.

Methods

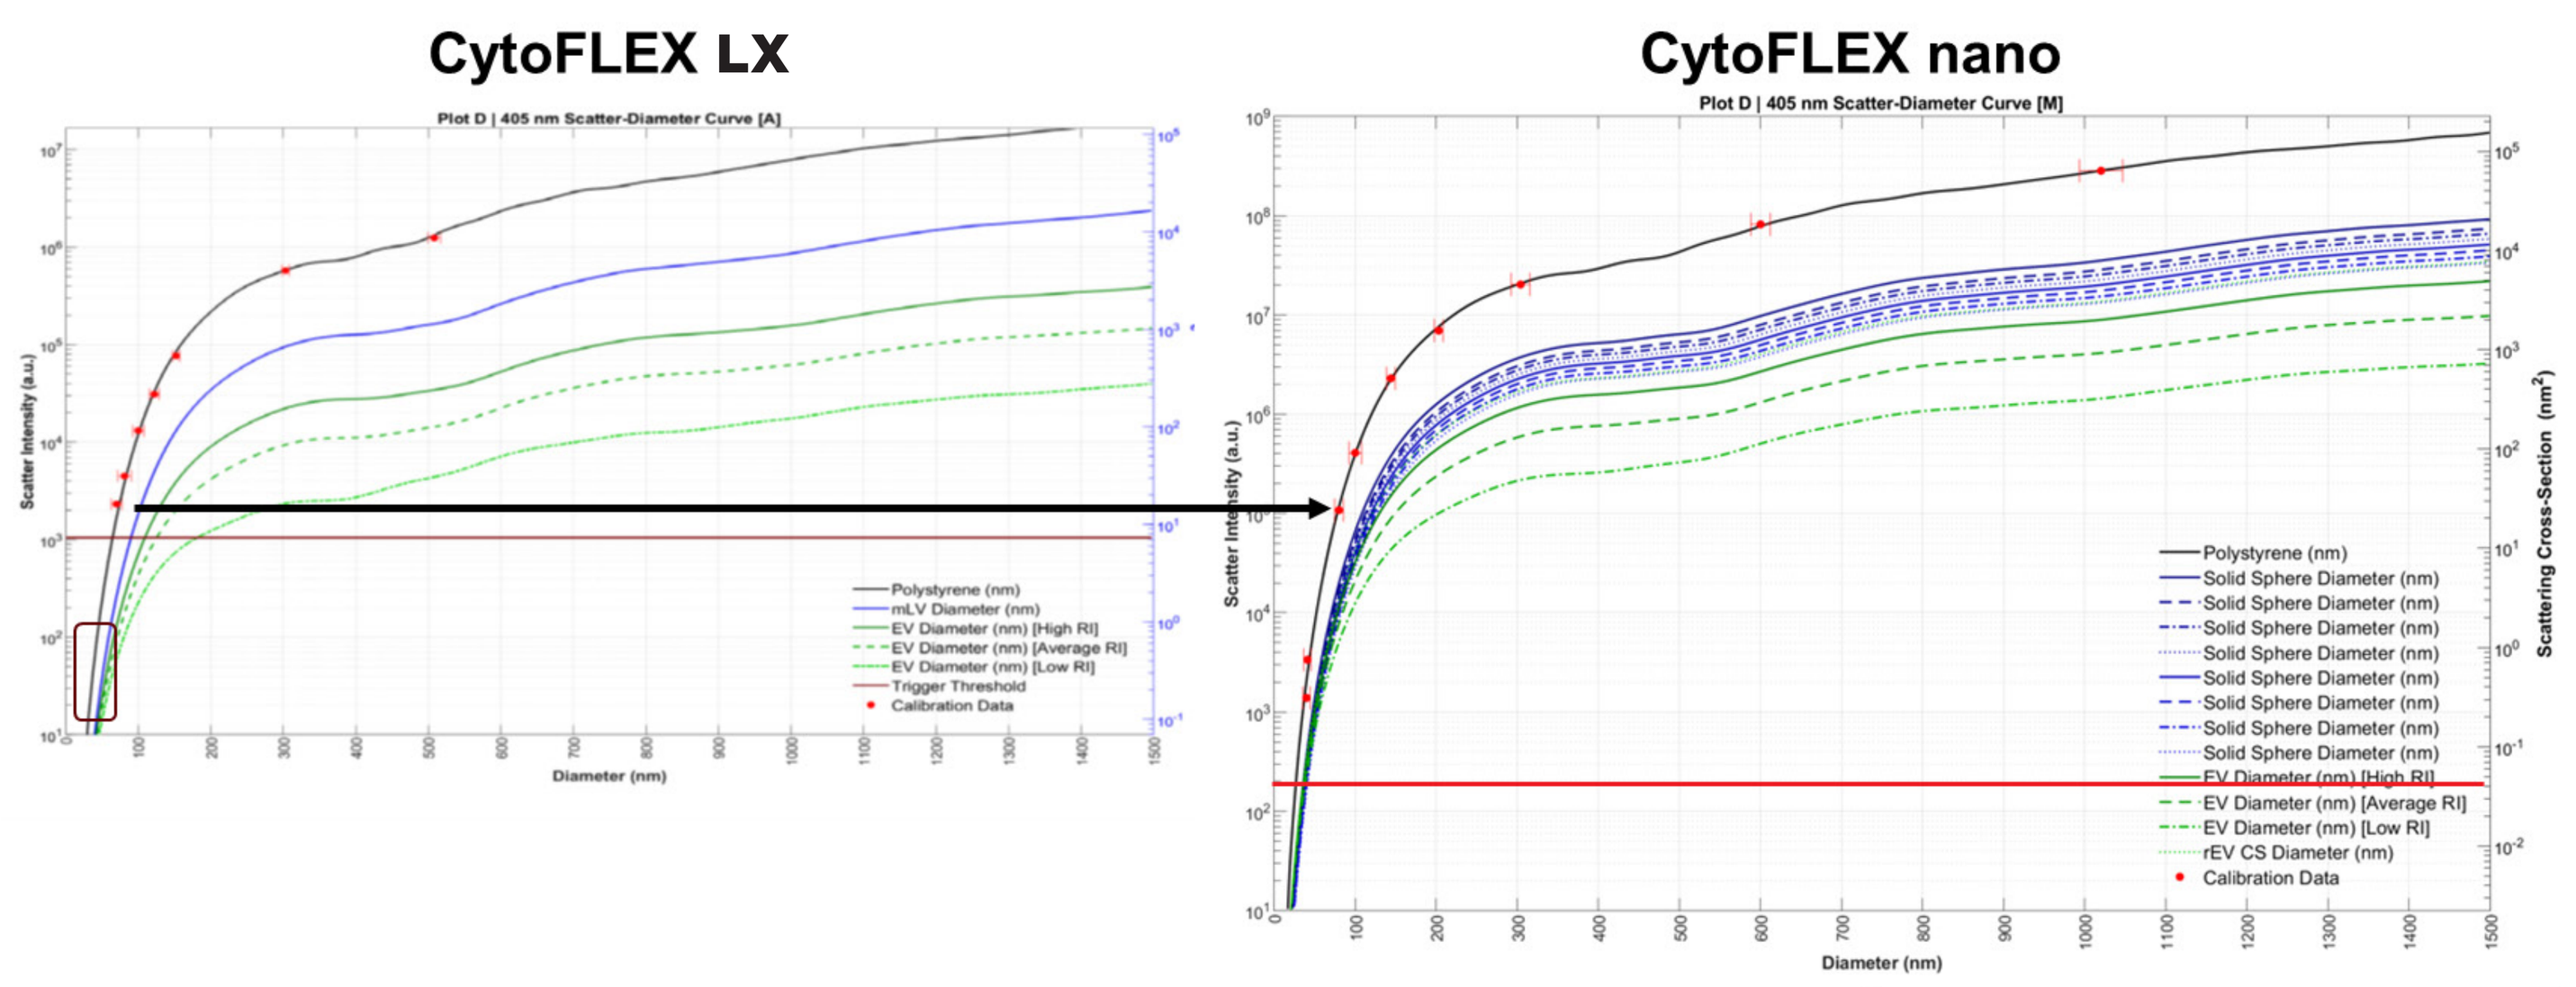
CytoFLEX LX
CytoFLEX nano
Plot D | 405 nm Scatter-Diameter Curve [A]
Plot D | 405 nm Scatter-Diameter Curve [M]
Diameter (nm)
Scatter Intensity (a.u.)
Scattering Cross-Section (nm²)

Figure 1. Comparison analysis between CytoFLEX LX and CytoFLEX nano. Scatter intensity as a function of the diameter of the particles showing difference in limits of detection. NIST Polystyrene beads were measured on both instruments.

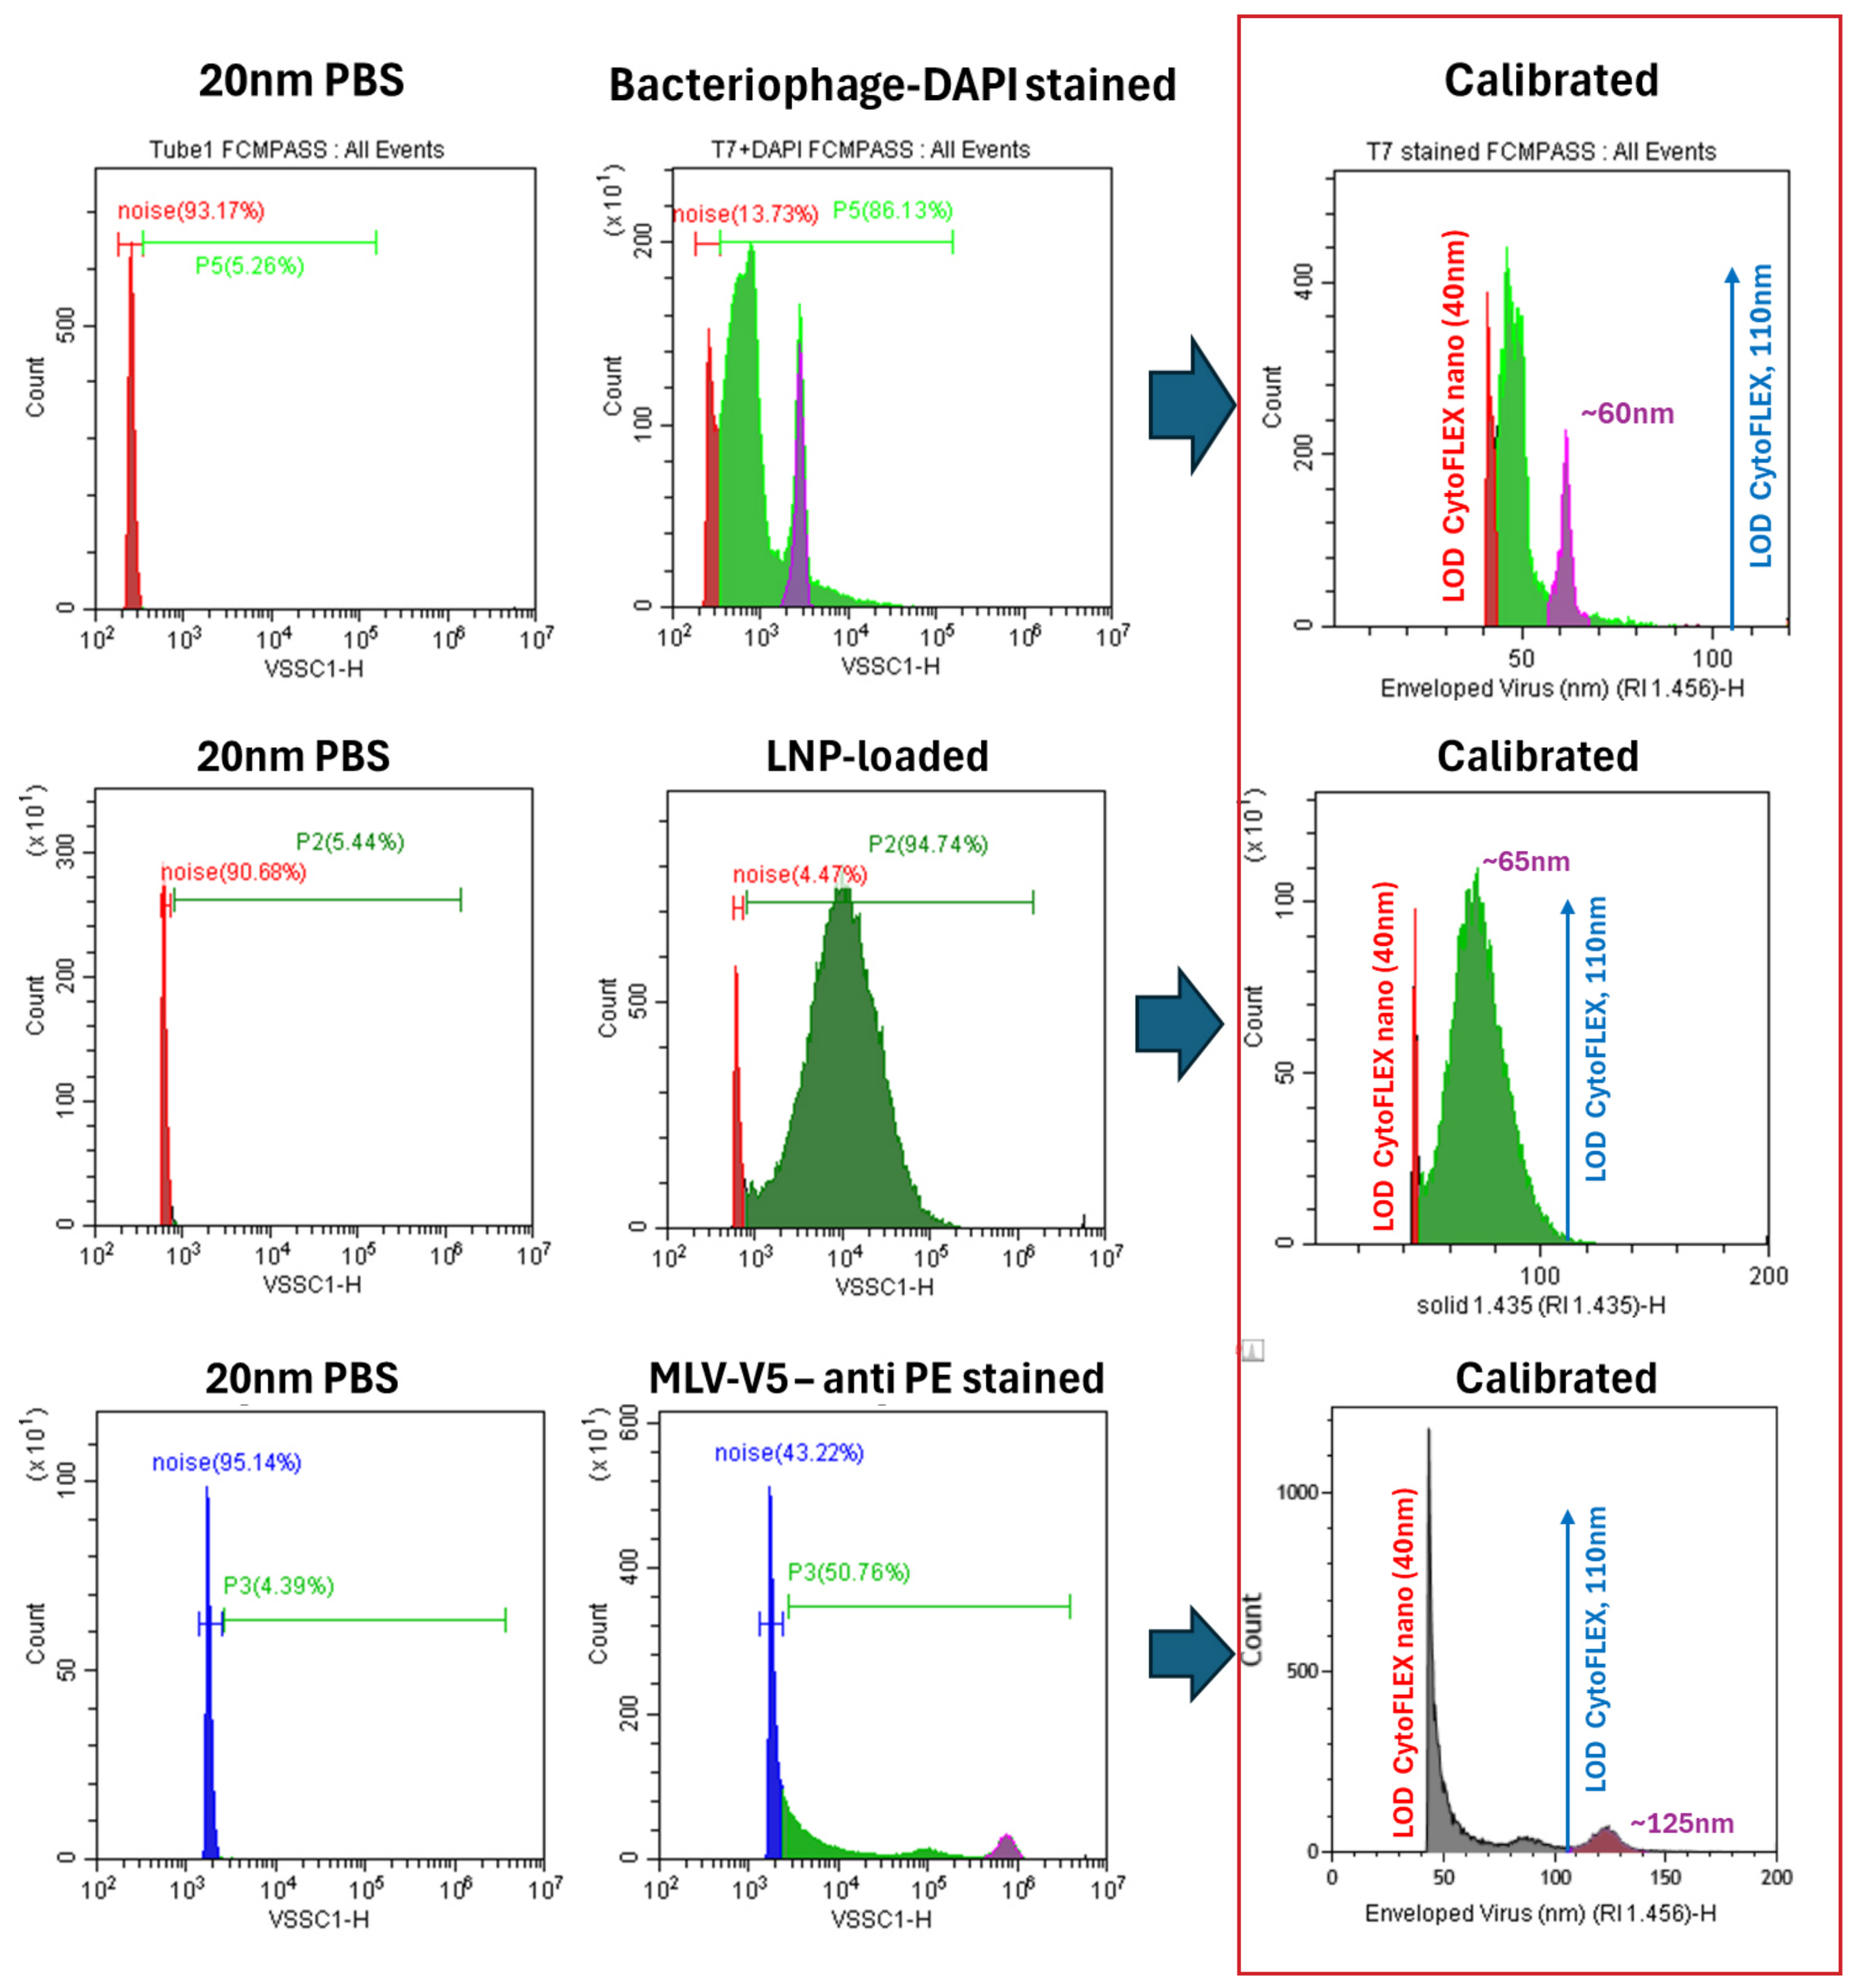
20nm PBS
Bacteriophage-DAPI stained
Calibrated
noise(93.17%)
P5(5.26%)
noise(13.73%)
P5(86.13%)
LOD CytoFLEX nano (40nm)
~60nm
LOD CytoFLEX, 110nm
Enveloped Virus (nm) (RI 1.456)-H
20nm PBS
LNP-loaded
Calibrated
noise(90.68%)
P2(5.44%)
noise(4.47%)
P2(94.74%)
~65nm
solid 1.435 (RI 1.435)-H
20nm PBS
MLV-V5 – anti PE stained
Calibrated
noise(95.14%)
P3(4.39%)
noise(43.22%)
P3(50.76%)
~125nm
VSSC1-H
Count

Figure 2. Analysis of biological samples in the CytoFLEX nano. Limits of detection of CytoFLEX LX and CytoFLEX Nano by scatter are displayed in accordance with the refractive index of the sample. 20 nm filtered PBS (a, d, g); Bacteriophage DAPI stained (b); calibrated data for Bacteriophage (c); Loaded lipid nanoparticles (LNPs) (e); calibrated data for LNPs (f); MLV-V5 anti-PE stained (h); calibrated MLV-V5 (i).
Conclusion. The CytoFLEX nano enables the detection of biological samples such as LNPs, Bacteriophage, and further characterization of samples such as MLV.

Step 1. Dilution tests for antibody controls only

Step 2. Isolation of EVs in PPP

Step 3. Dilution tests of the unstained sample

Step 4. Fractions identification

Step 5. Staining protocol

Figure 3. Isolation and staining of platelet-poor plasma (PPP) EVs. Typical workflow diagram of testing and preparation of PPP EVs following staining protocol.

1. Markers selection. CD9 and CD61 provide good staining for PPP EVs.

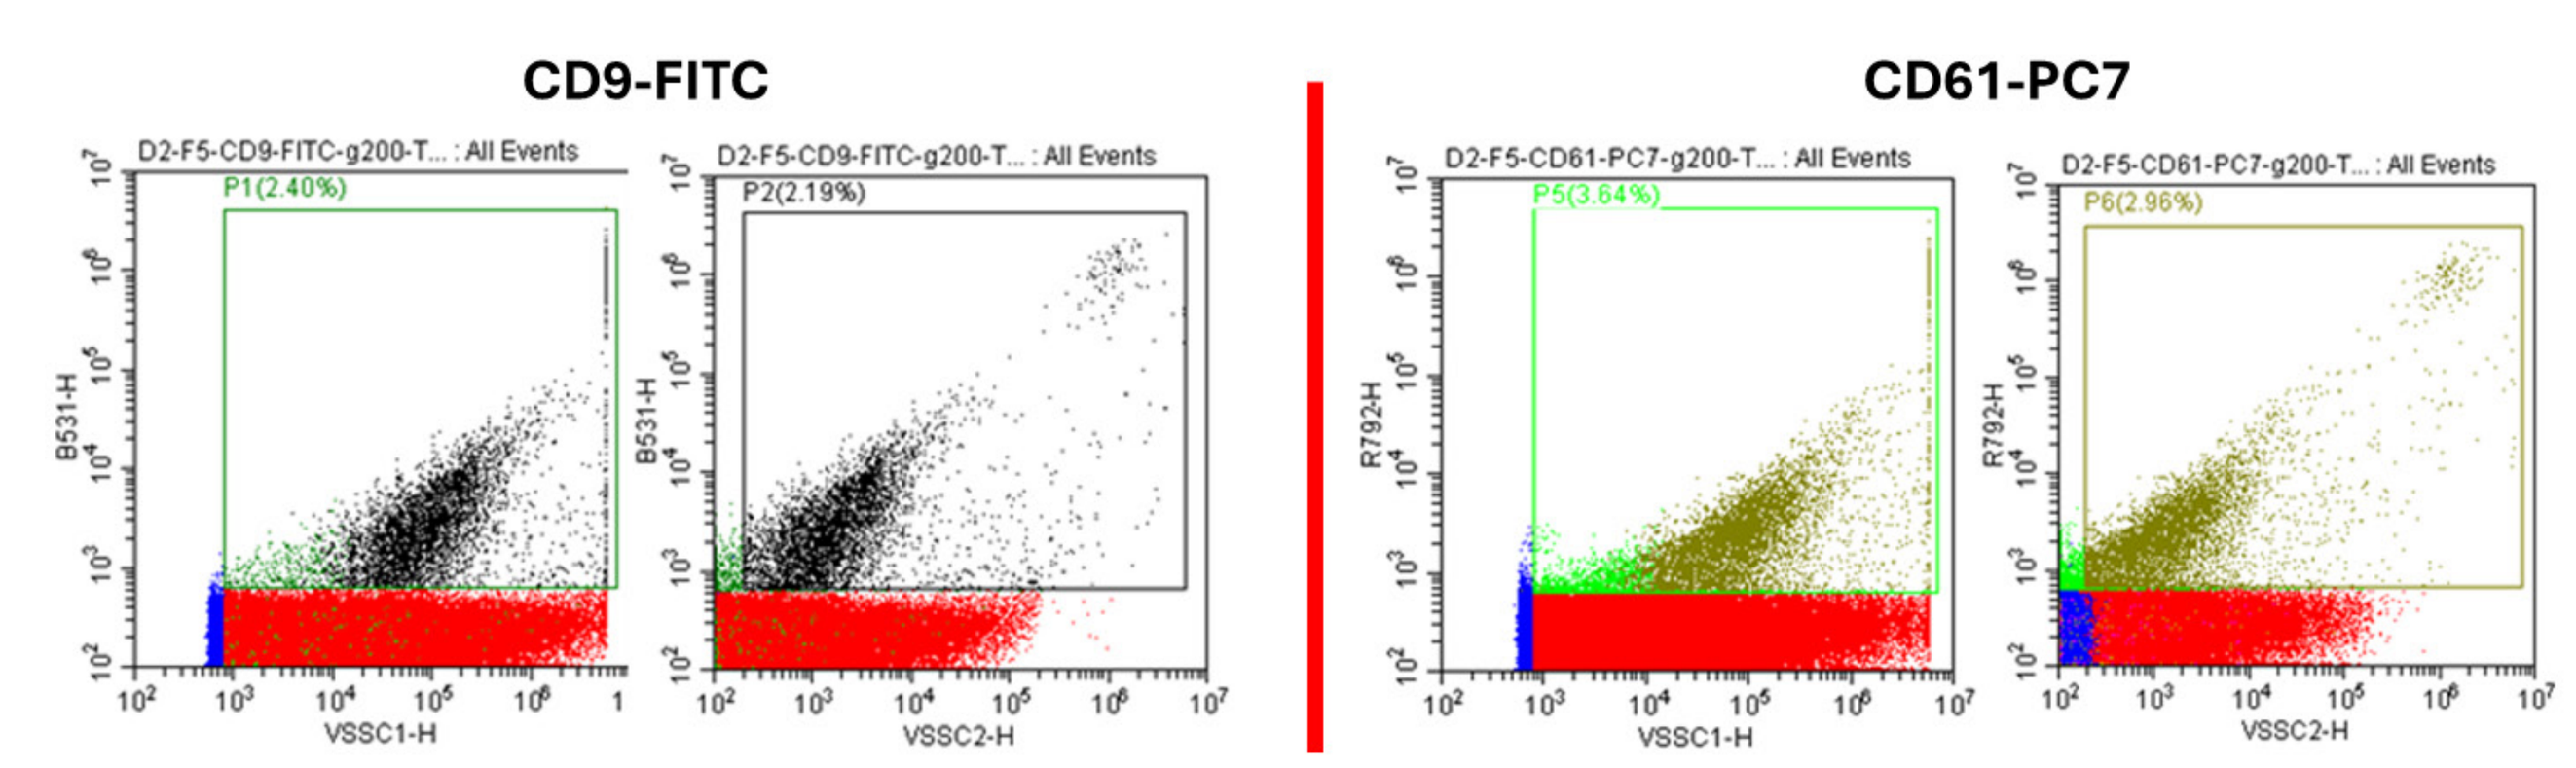
CD9-FITC
CD61-PC7
P1(2.40%)
P2(2.19%)
P5(3.64%)
P6(2.96%)
VSSC1-H
VSSC2-H

2. Fractions selection and Donor dependance. Staining of different fractions could be affected depending on the donor.

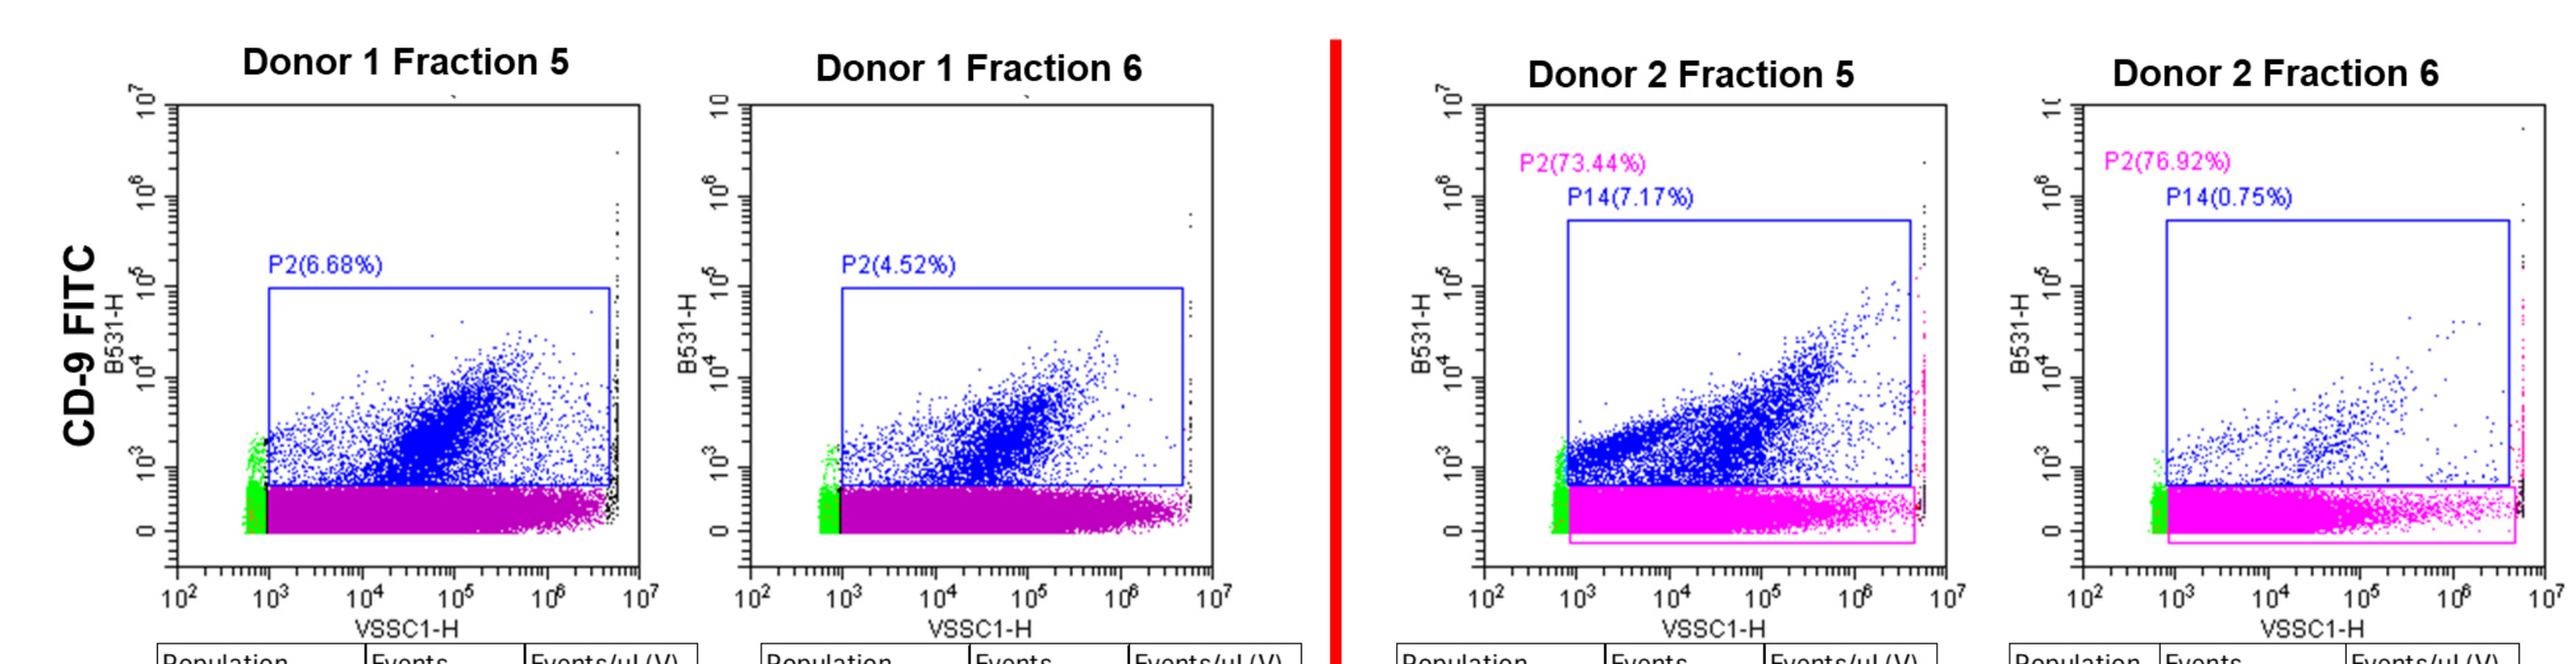
Donor 1 Fraction 5
Donor 1 Fraction 6
Donor 2 Fraction 5
Donor 2 Fraction 6
CD-9 FITC
P2(6.68%)
P2(4.52%)
P2(73.44%)
P14(7.17%)
P2(76.92%)
P14(0.75%)

Population	Events	Events/µL(V)
All Events	100000	13977.02
P2	6316	882.79

Population	Events	Events/µL(V)
All Events	100000	21677.63
P2	4235	918.05

Population	Events	Events/µL(V)
All Events	100000	13147.43
P14	7173	943.07

Population	Events	Events/µL(V)
All Events	100000	14636.36
P14	748	109.48

3. Clone selection. Staining brightness could be affected depending on the clone.

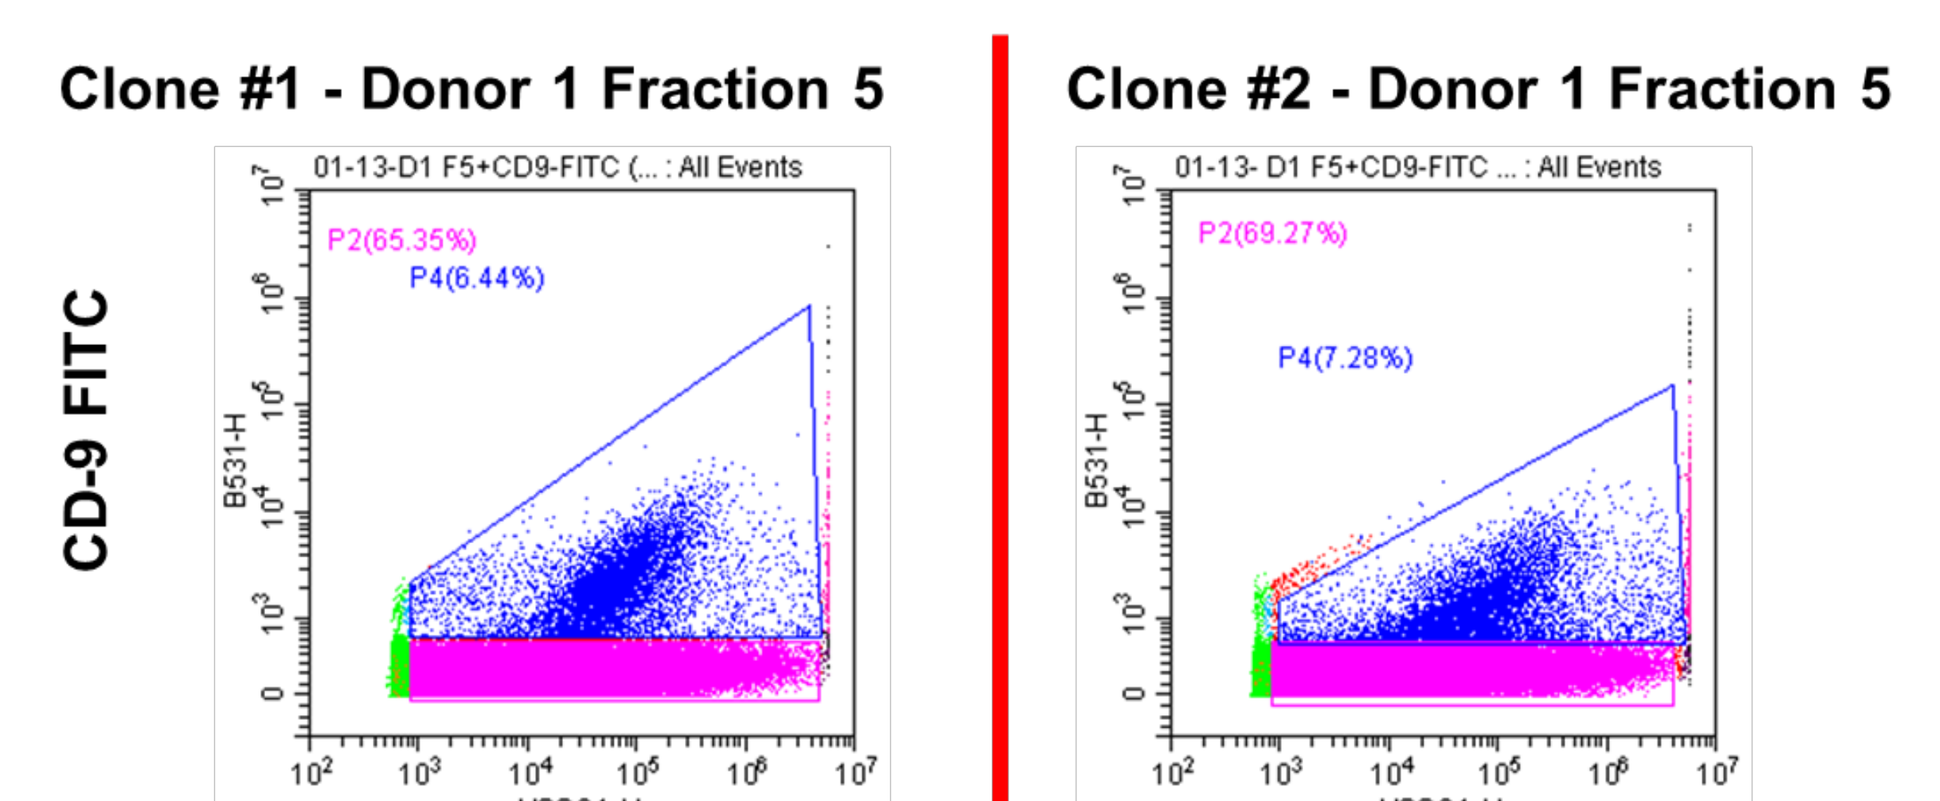
Clone #1 - Donor 1 Fraction 5
Clone #2 - Donor 1 Fraction 5
CD-9 FITC
P2(65.35%)
P4(6.44%)
P2(69.27%)
P4(7.28%)
VSSC1-H

Population	Events	Events/µL(V)	Median B531-H
All Events	100000	13977.02	231.9
P4	6438	899.84	2091.6

Population	Events	Events/µL(V)	Median B531-H
All Events	100000	11613.66	221.4
P4	7279	845.36	1126.2

4. Instrument and channel sensitivity dependence. Staining brightness could be affected depending on the instrument and specific channel.

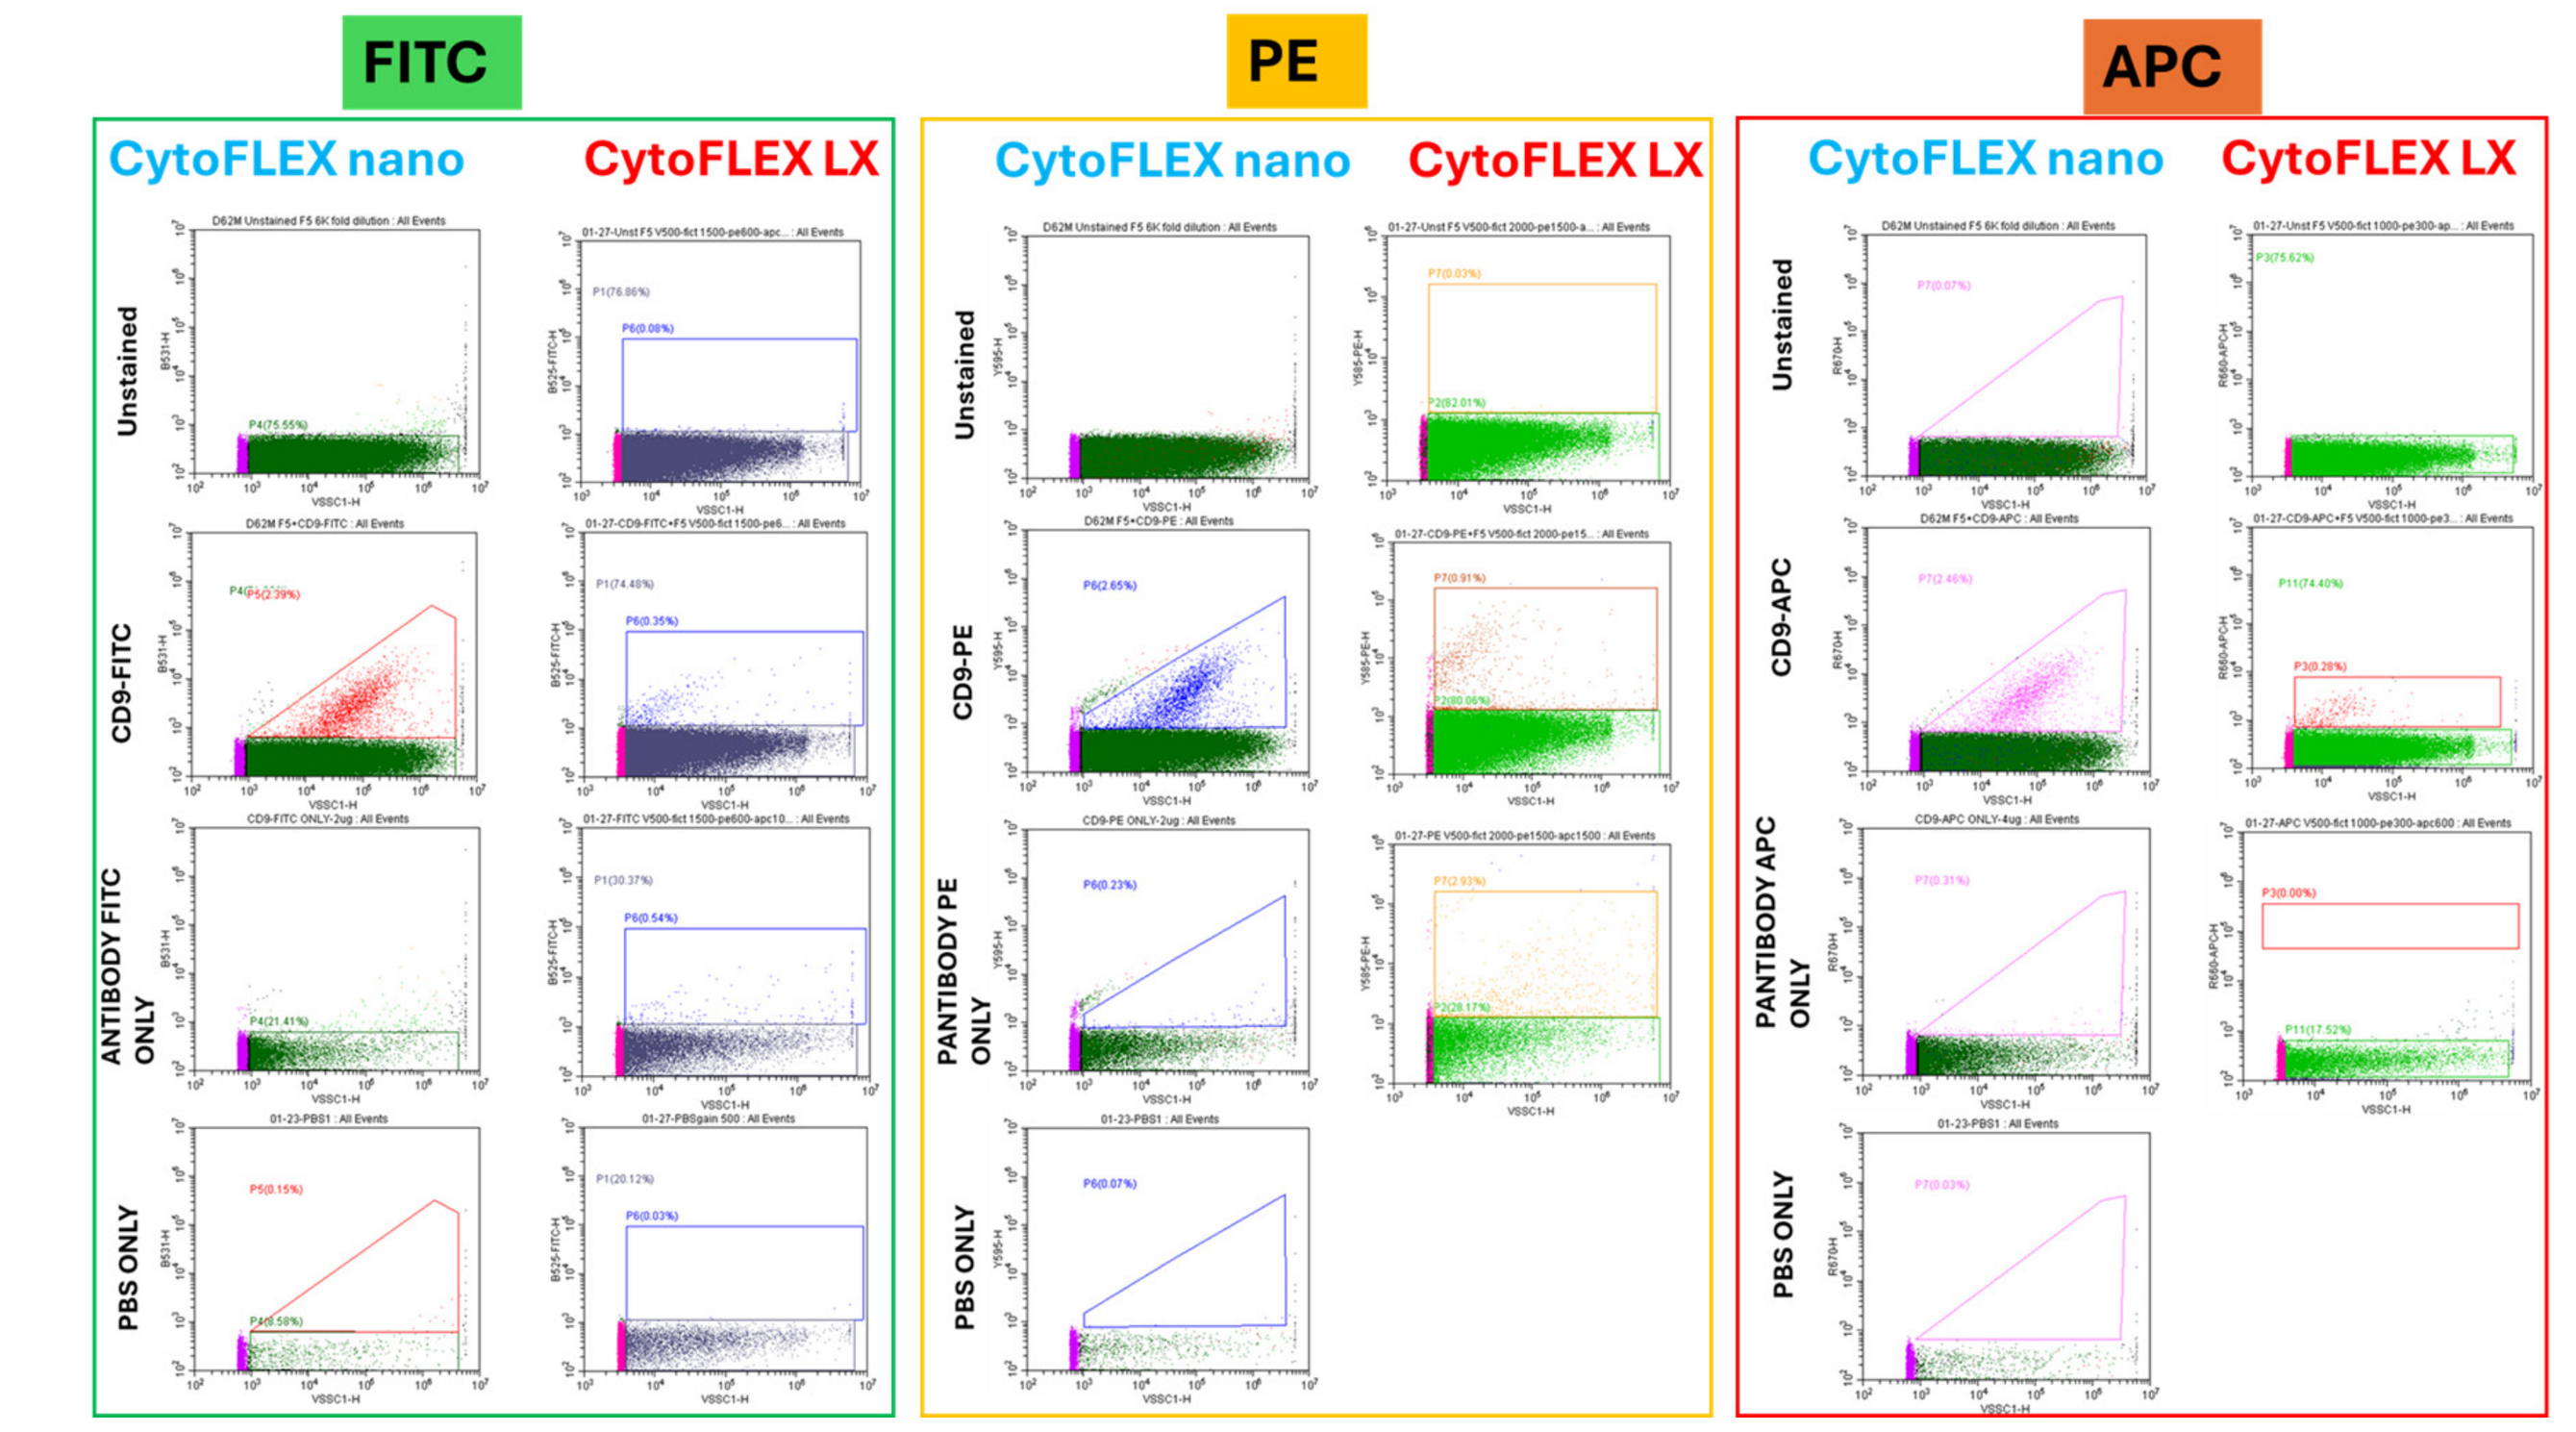
FITC
PE
APC
CytoFLEX nano
CytoFLEX LX
Unstained
CD9-FITC
ANTIBODY FITC ONLY
PBS ONLY
CD9-PE
PANTIBODY PE ONLY
CD9-APC
PANTIBODY APC ONLY

Figure 4. Factors Influencing Single Color Optimization. 1) markers selection, 2) fractions selection and donor dependence, 3) clone selection, and 4) instrument sensitivity at specific channels.

APC

Instruments	Event number	LOD, #sites	LOD, nm
CytoFLEX LX, gate A	384	200	100 nm
CytoFLEX nano, gate A	685		
CytoFLEXLX nano, gate C	2303	20	50 nm

FITC

Instruments	Event number	LOD, #sites	LOD, nm
CytoFLEX LX, gate A	304	65	100 nm
CytoFLEX nano, gate A	811		
CytoFLEXLX nano, gate B	2365	25	50 nm

PE

Instruments	Event number	LOD, #sites	LOD, nm
CytoFLEX LX, gate A	609	208	100 nm
CytoFLEX nano, gate A	1213		
CytoFLEXLX nano, gate B	2412	80	50 nm

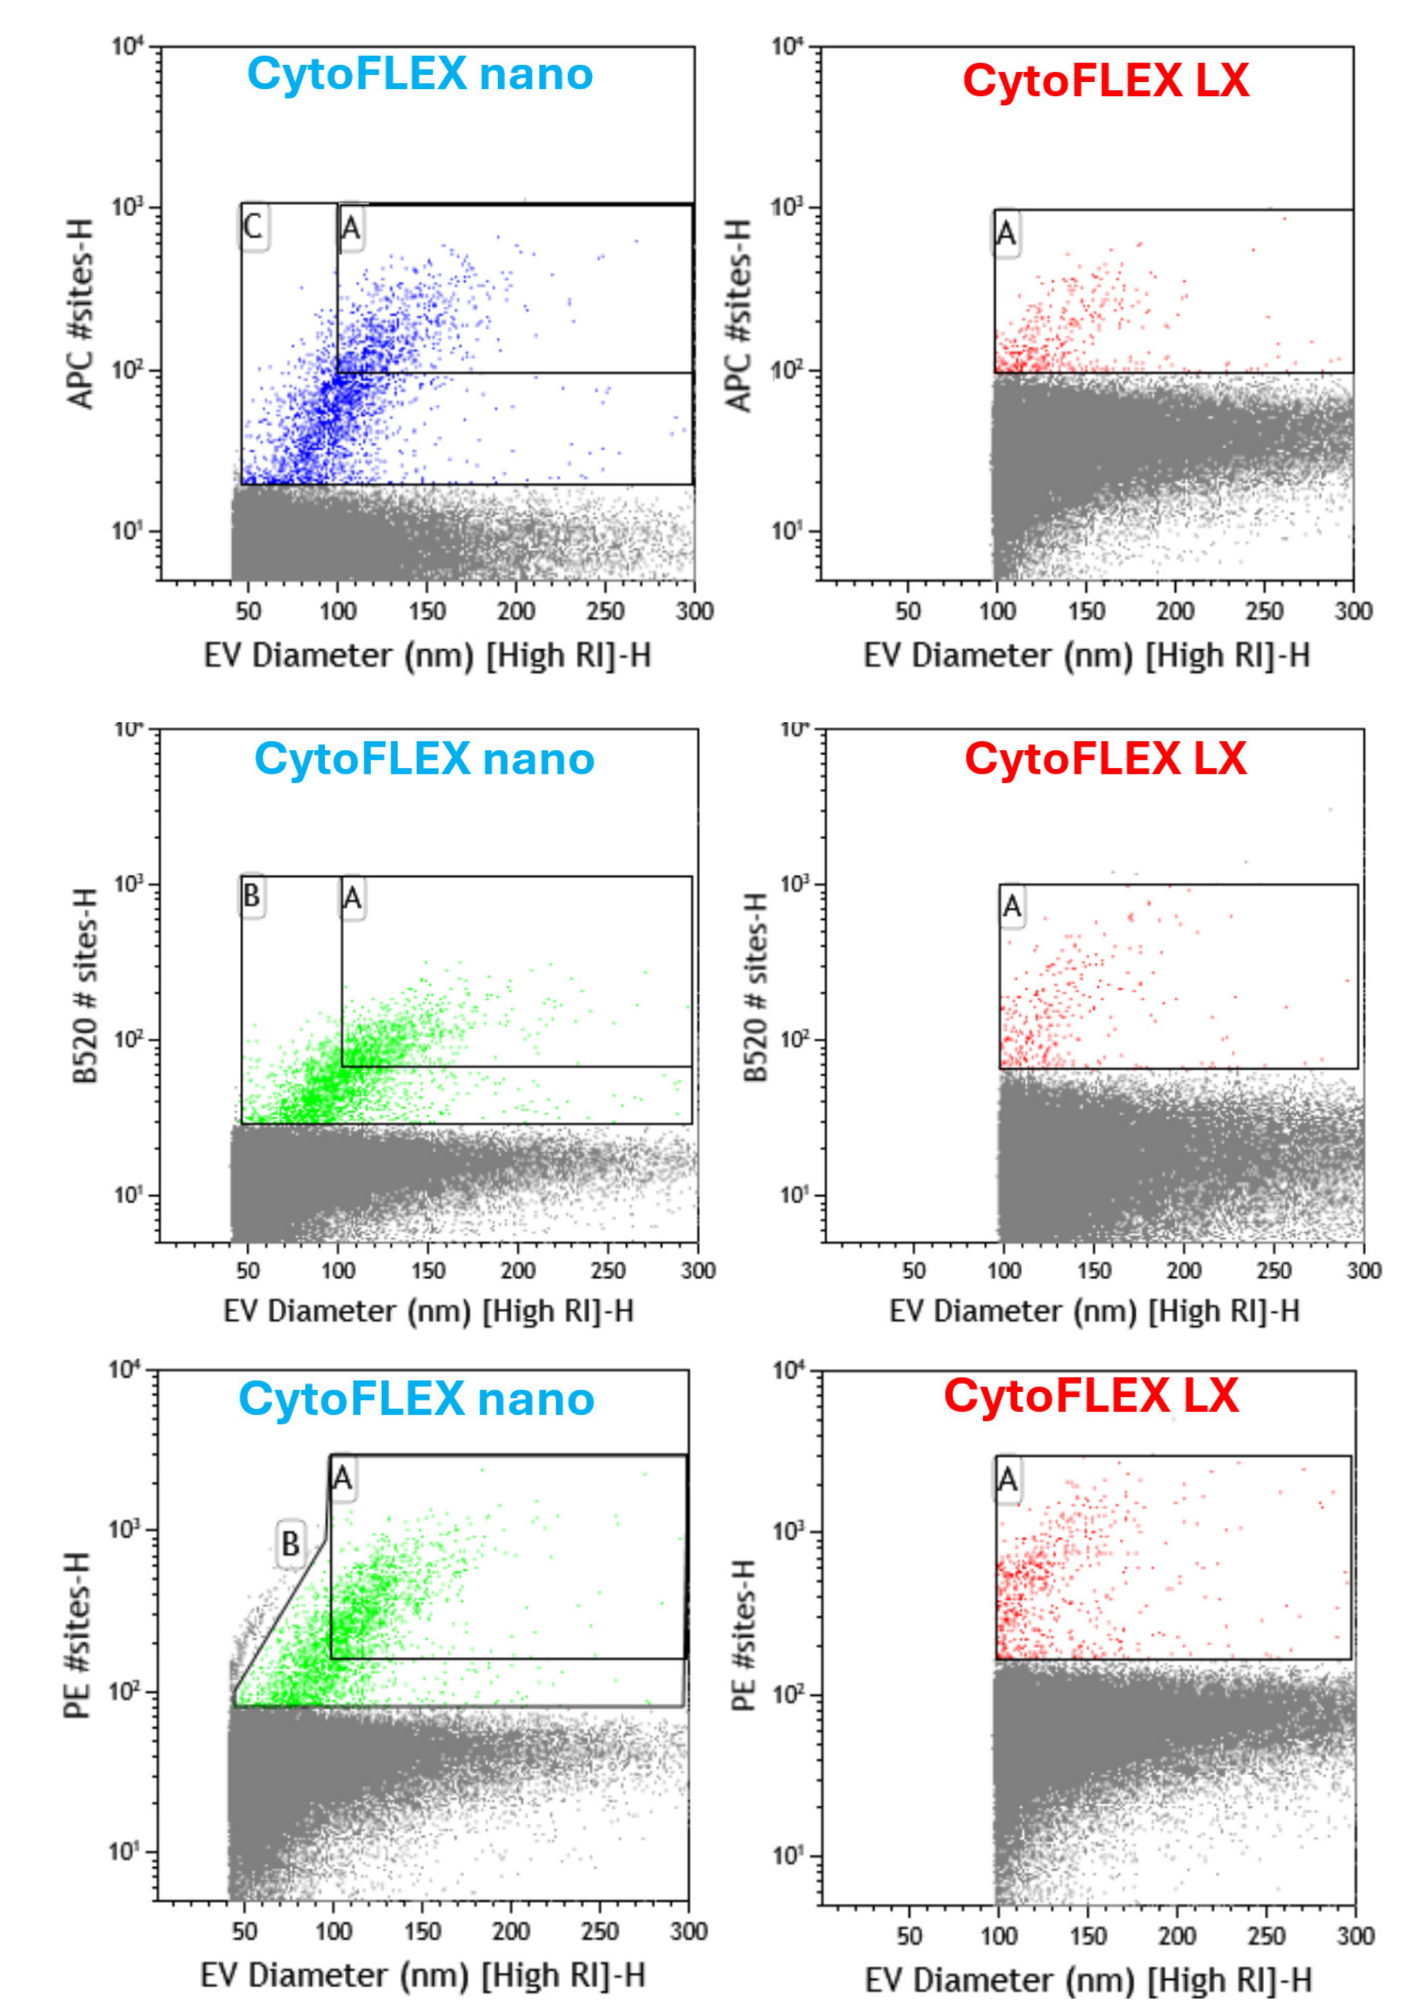
CytoFLEX nano
CytoFLEX LX
APC #sites-H
B520 # sites-H
PE #sites-H
EV Diameter (nm) [High RI]-H

Figure 5. Limits of detection of the CytoFLEX LX and the CytoFLEX nano by fluorescence for APC, PE, and FITC using optimized staining of PPP Evs. Based on antibody capture beads for APC, PE, and B520, respectively.

Conclusions

The CytoFLEX nano provides superior scatter sensitivity compared to the CytoFLEX LX, enabling the detection of biological samples approximately 50 nm in size with refractive indices around 1.4. Additionally, the CytoFLEX nano offers superior fluorescence sensitivity compared to the CytoFLEX LX, allowing the detection of approximately 20 antibody capture sites of fluorescent fluorochrome.

Acknowledgements

Cytiva R&D team Hailey Kim, Andrew Kondratowicz, Adam Crowe

For Research Use Only. Not for use in diagnostic procedures.
© 2025 Beckman Coulter, Inc. All rights reserved. Beckman Coulter, the stylized logo, and the Beckman Coulter product and service marks mentioned herein are trademarks or registered trademarks of Beckman Coulter, Inc. in the United States and other countries.
All other trademarks are the property of their respective owners.

For Beckman Coulter's worldwide office locations and phone numbers, please visit contact us at **beckman.com**
2025-GBL-EN-107674-v2