Label-free Characterization of Biological Nanoparticles

Evgenia Kim[1], Vera Tang[2], Hoa Boi San[1], Jen Bon Lui[1], Milan Popovic[1], Anisha Jose[3], Sergei Gulnik[1],
[1]Research and Development, Beckman Coulter Life Sciences, Miami, FL 33196.
[2]University of Ottawa, Department of Biochemistry, Microbiology and Immunology.
[3]Commercial Excellence, Beckman Coulter Life Sciences.

Introduction

Flow cytometry is a powerful method to analyze heterogeneous particle populations based on single particle detection and is widely used for cellular analysis. However, the sensitivity of most current conventional cytometers has limitations to detect nanoparticles with sizes below 200 nm, creating a challenge for extracellular vesicle (EV) analysis. This work aims to demonstrate the performance of the CytoFLEX nano Flow Cytometer as a sensitive **label-free tool** to characterize nanoparticles by leveraging its capability to collect and analyze scatter parameters at multiple wavelengths.

Methods

The methodology used is explained in Figure 1. We will demonstrate how the CytoFLEX nano, equipped with 5 scatter channels, can benefit the customer.

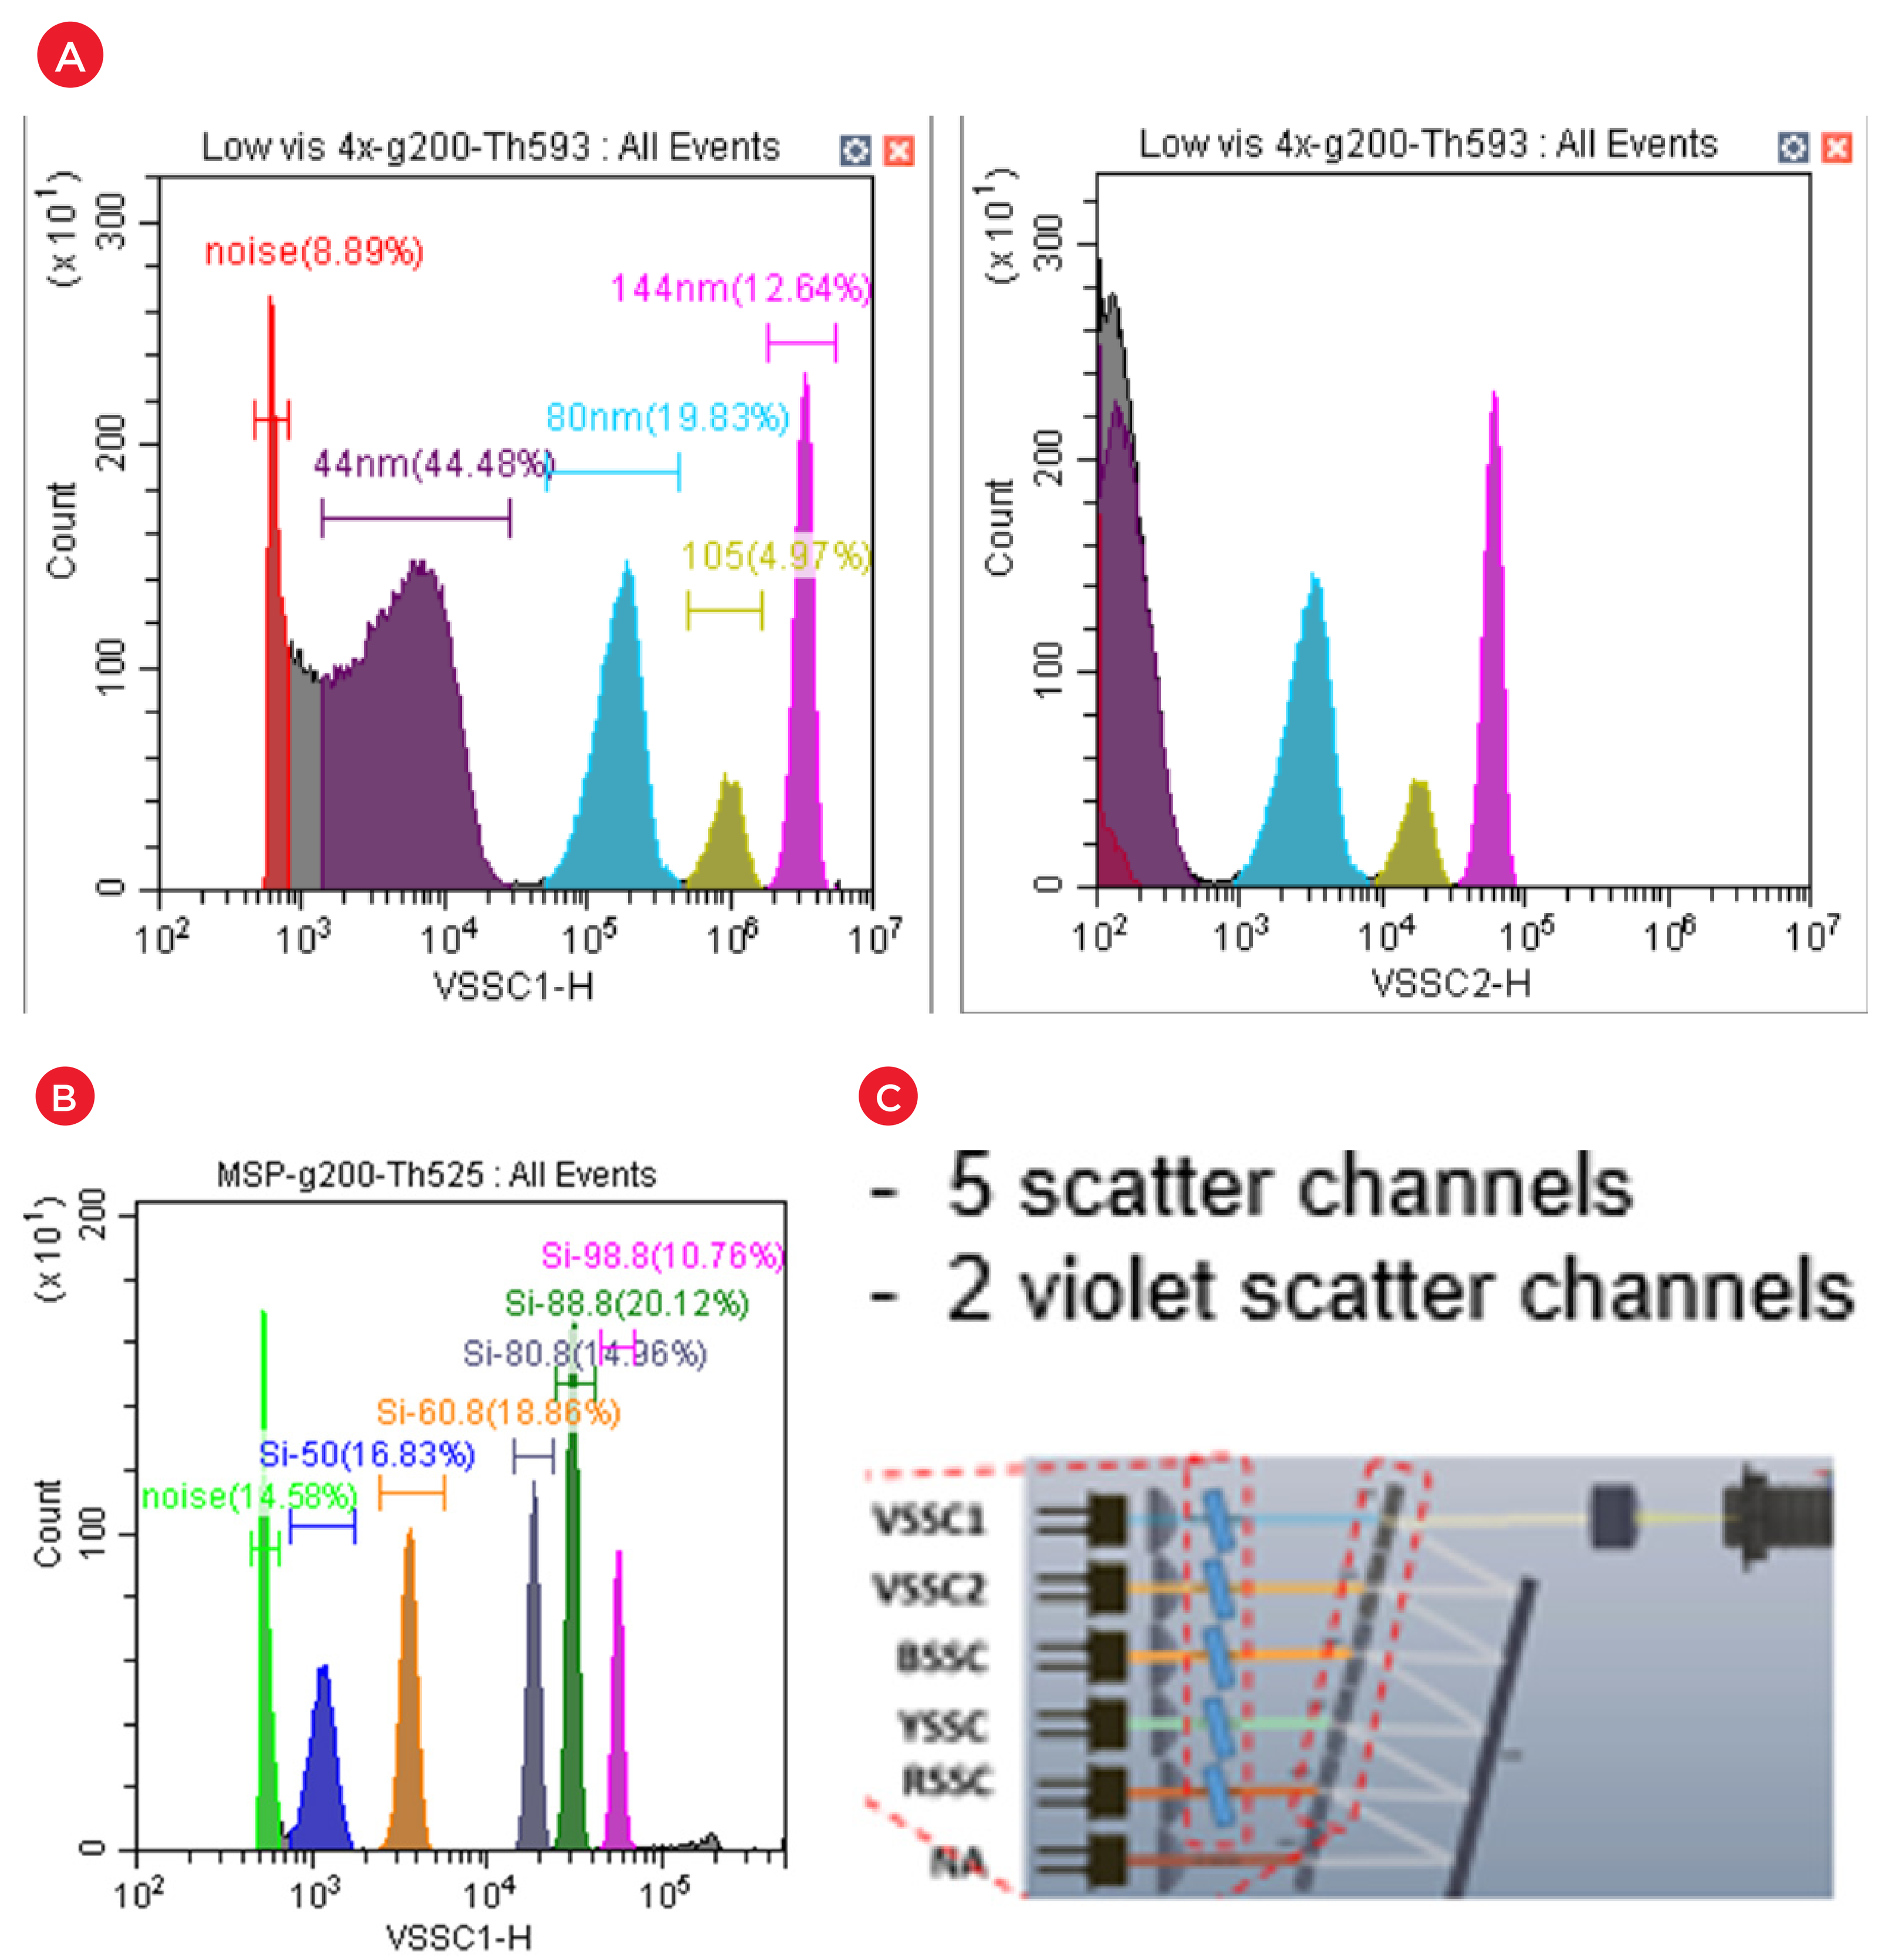
A
Low vis 4x-g200-Th593 : All Events
noise(8.89%)
144nm(12.64%)
80nm(19.83%)
44nm(44.48%)
105(4.97%)
Count
VSSC1-H
VSSC2-H
B
MSP-g200-Th525 : All Events
Si-98.8(10.76%)
Si-88.8(20.12%)
Si-80.8(14.96%)
Si-60.8(18.86%)
Si-50(16.83%)
noise(14.58%)
C
- 5 scatter channels
- 2 violet scatter channels
VSSC1
VSSC2
BSSC
YSSC
RSSC

Figure 1. Histograms of small particles run on Violet side scatter (VSSC). Polystyrene beads (a) and silica beads (b); schematic of the scatter channels.

Results

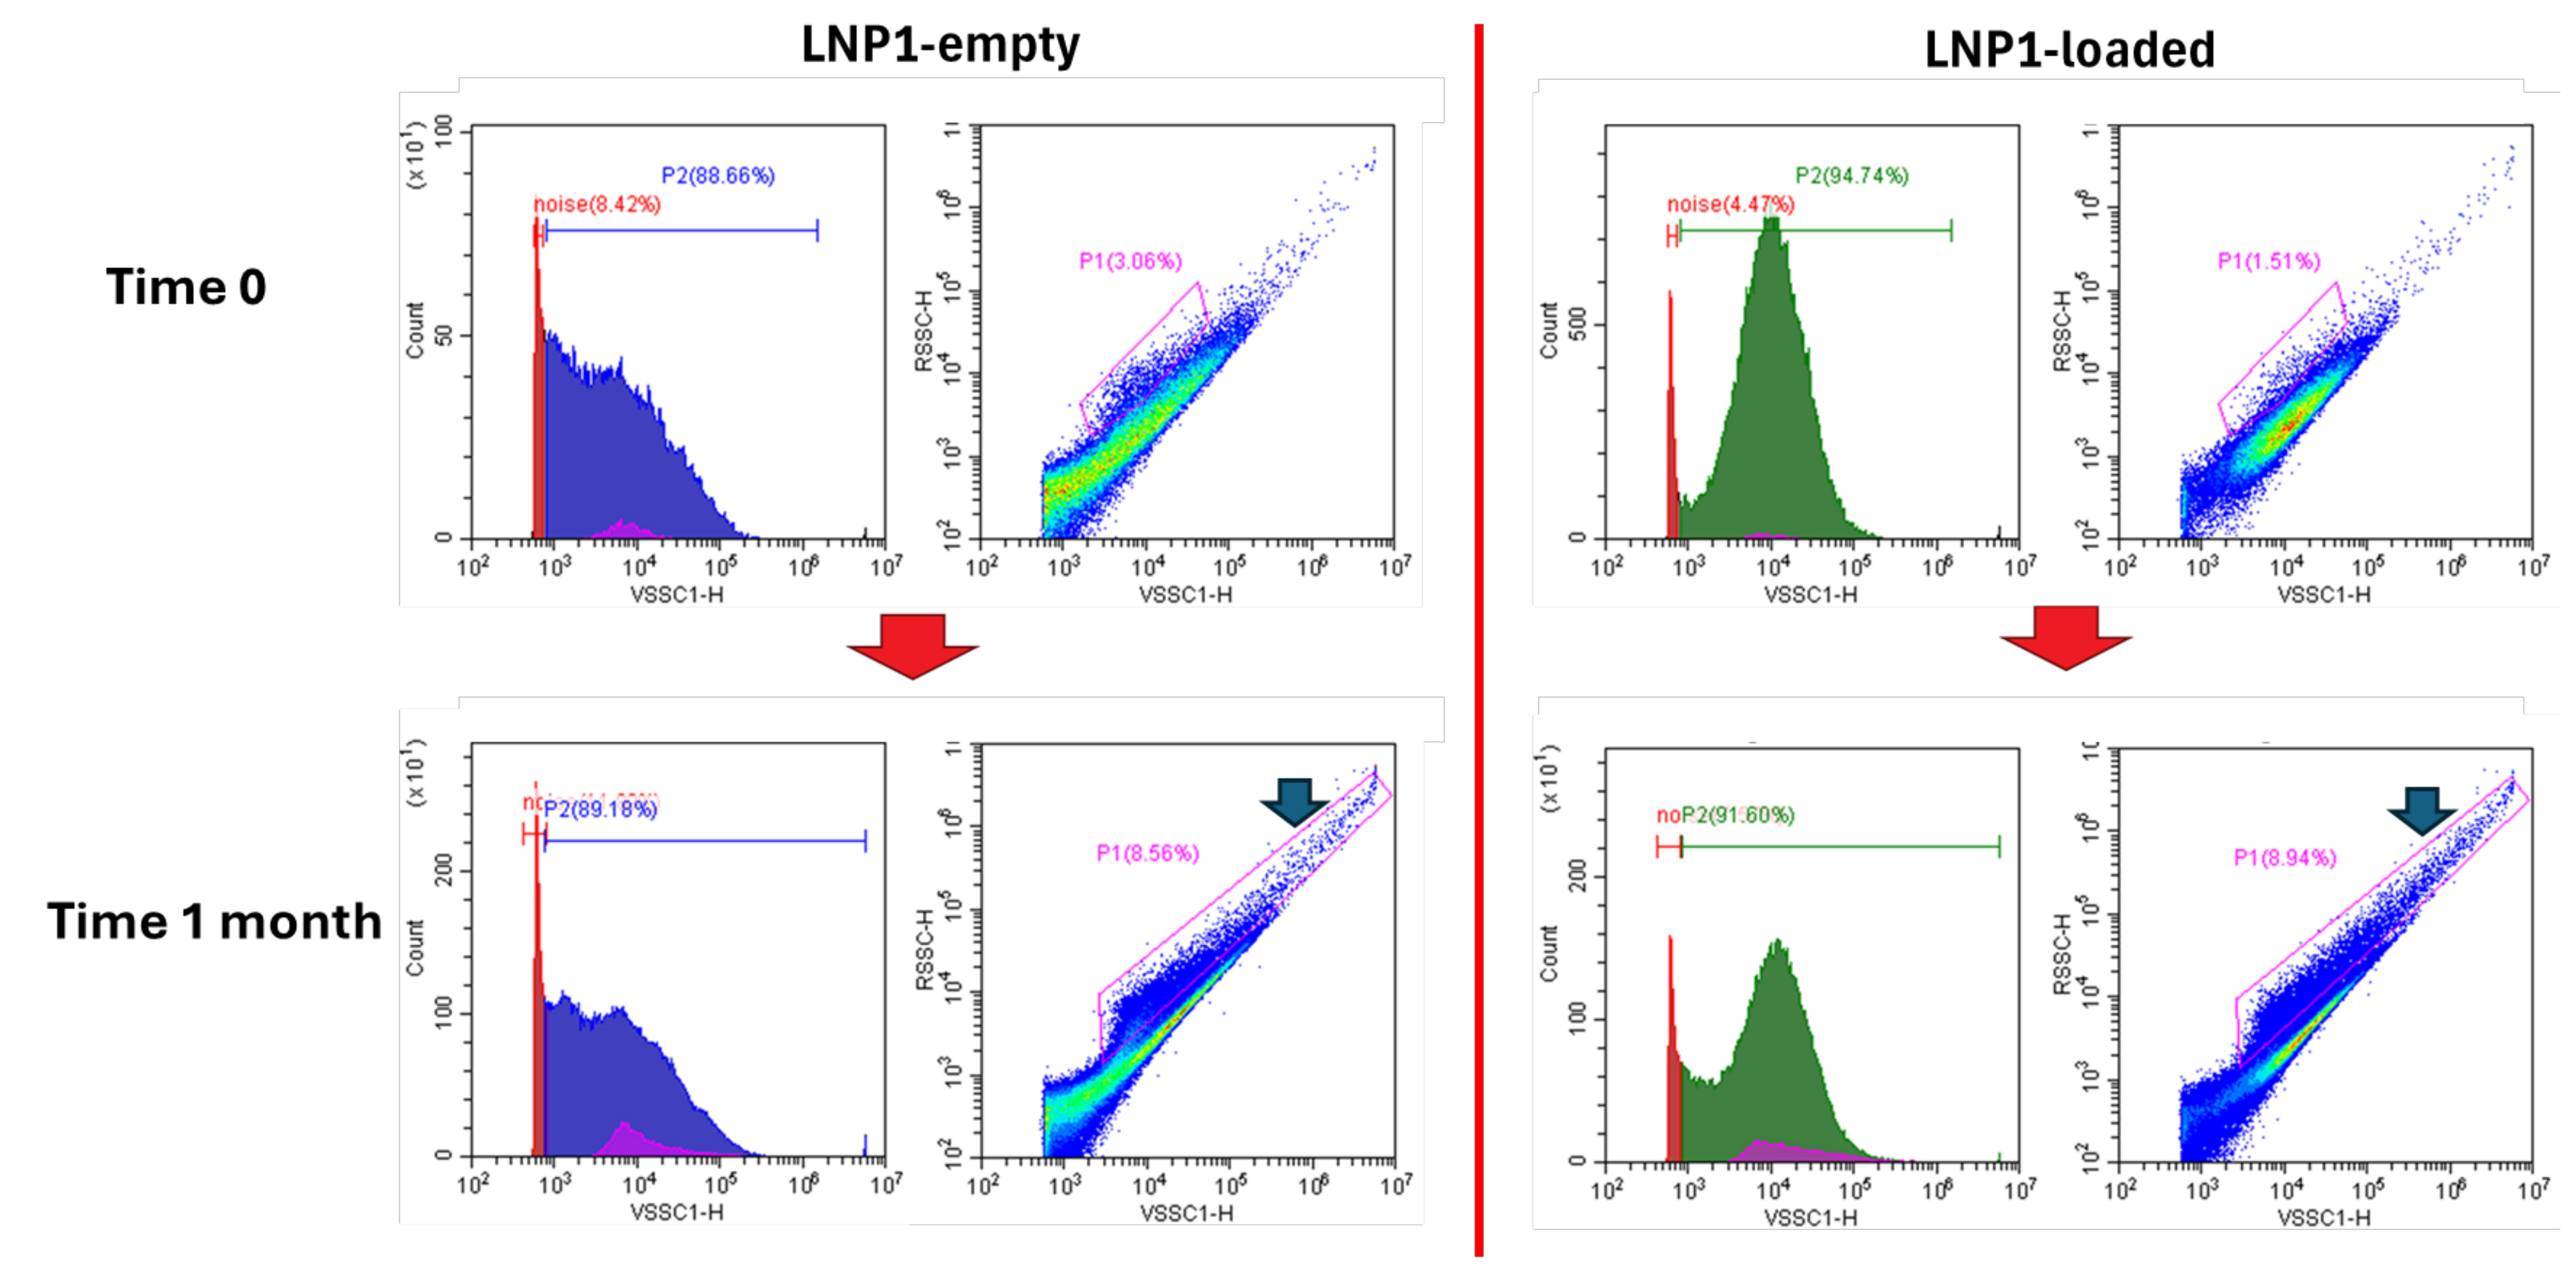
LNP1-empty
LNP1-loaded
Time 0
Time 1 month
noise(8.42%)
P2(88.66%)
P1(3.06%)
noise(4.47%)
P2(94.74%)
P1(1.51%)
P2(89.18%)
P1(8.56%)
P2(91.60%)
P1(8.94%)
VSSC1-H
RSSC-H
Count

Figure 2. Stability of Lipid nanoparticles (LNPs) over time. Empty and loaded LNPs were assessed and show degradation after one month. Violet-SSC1 and Red-SSC were used for this analysis.

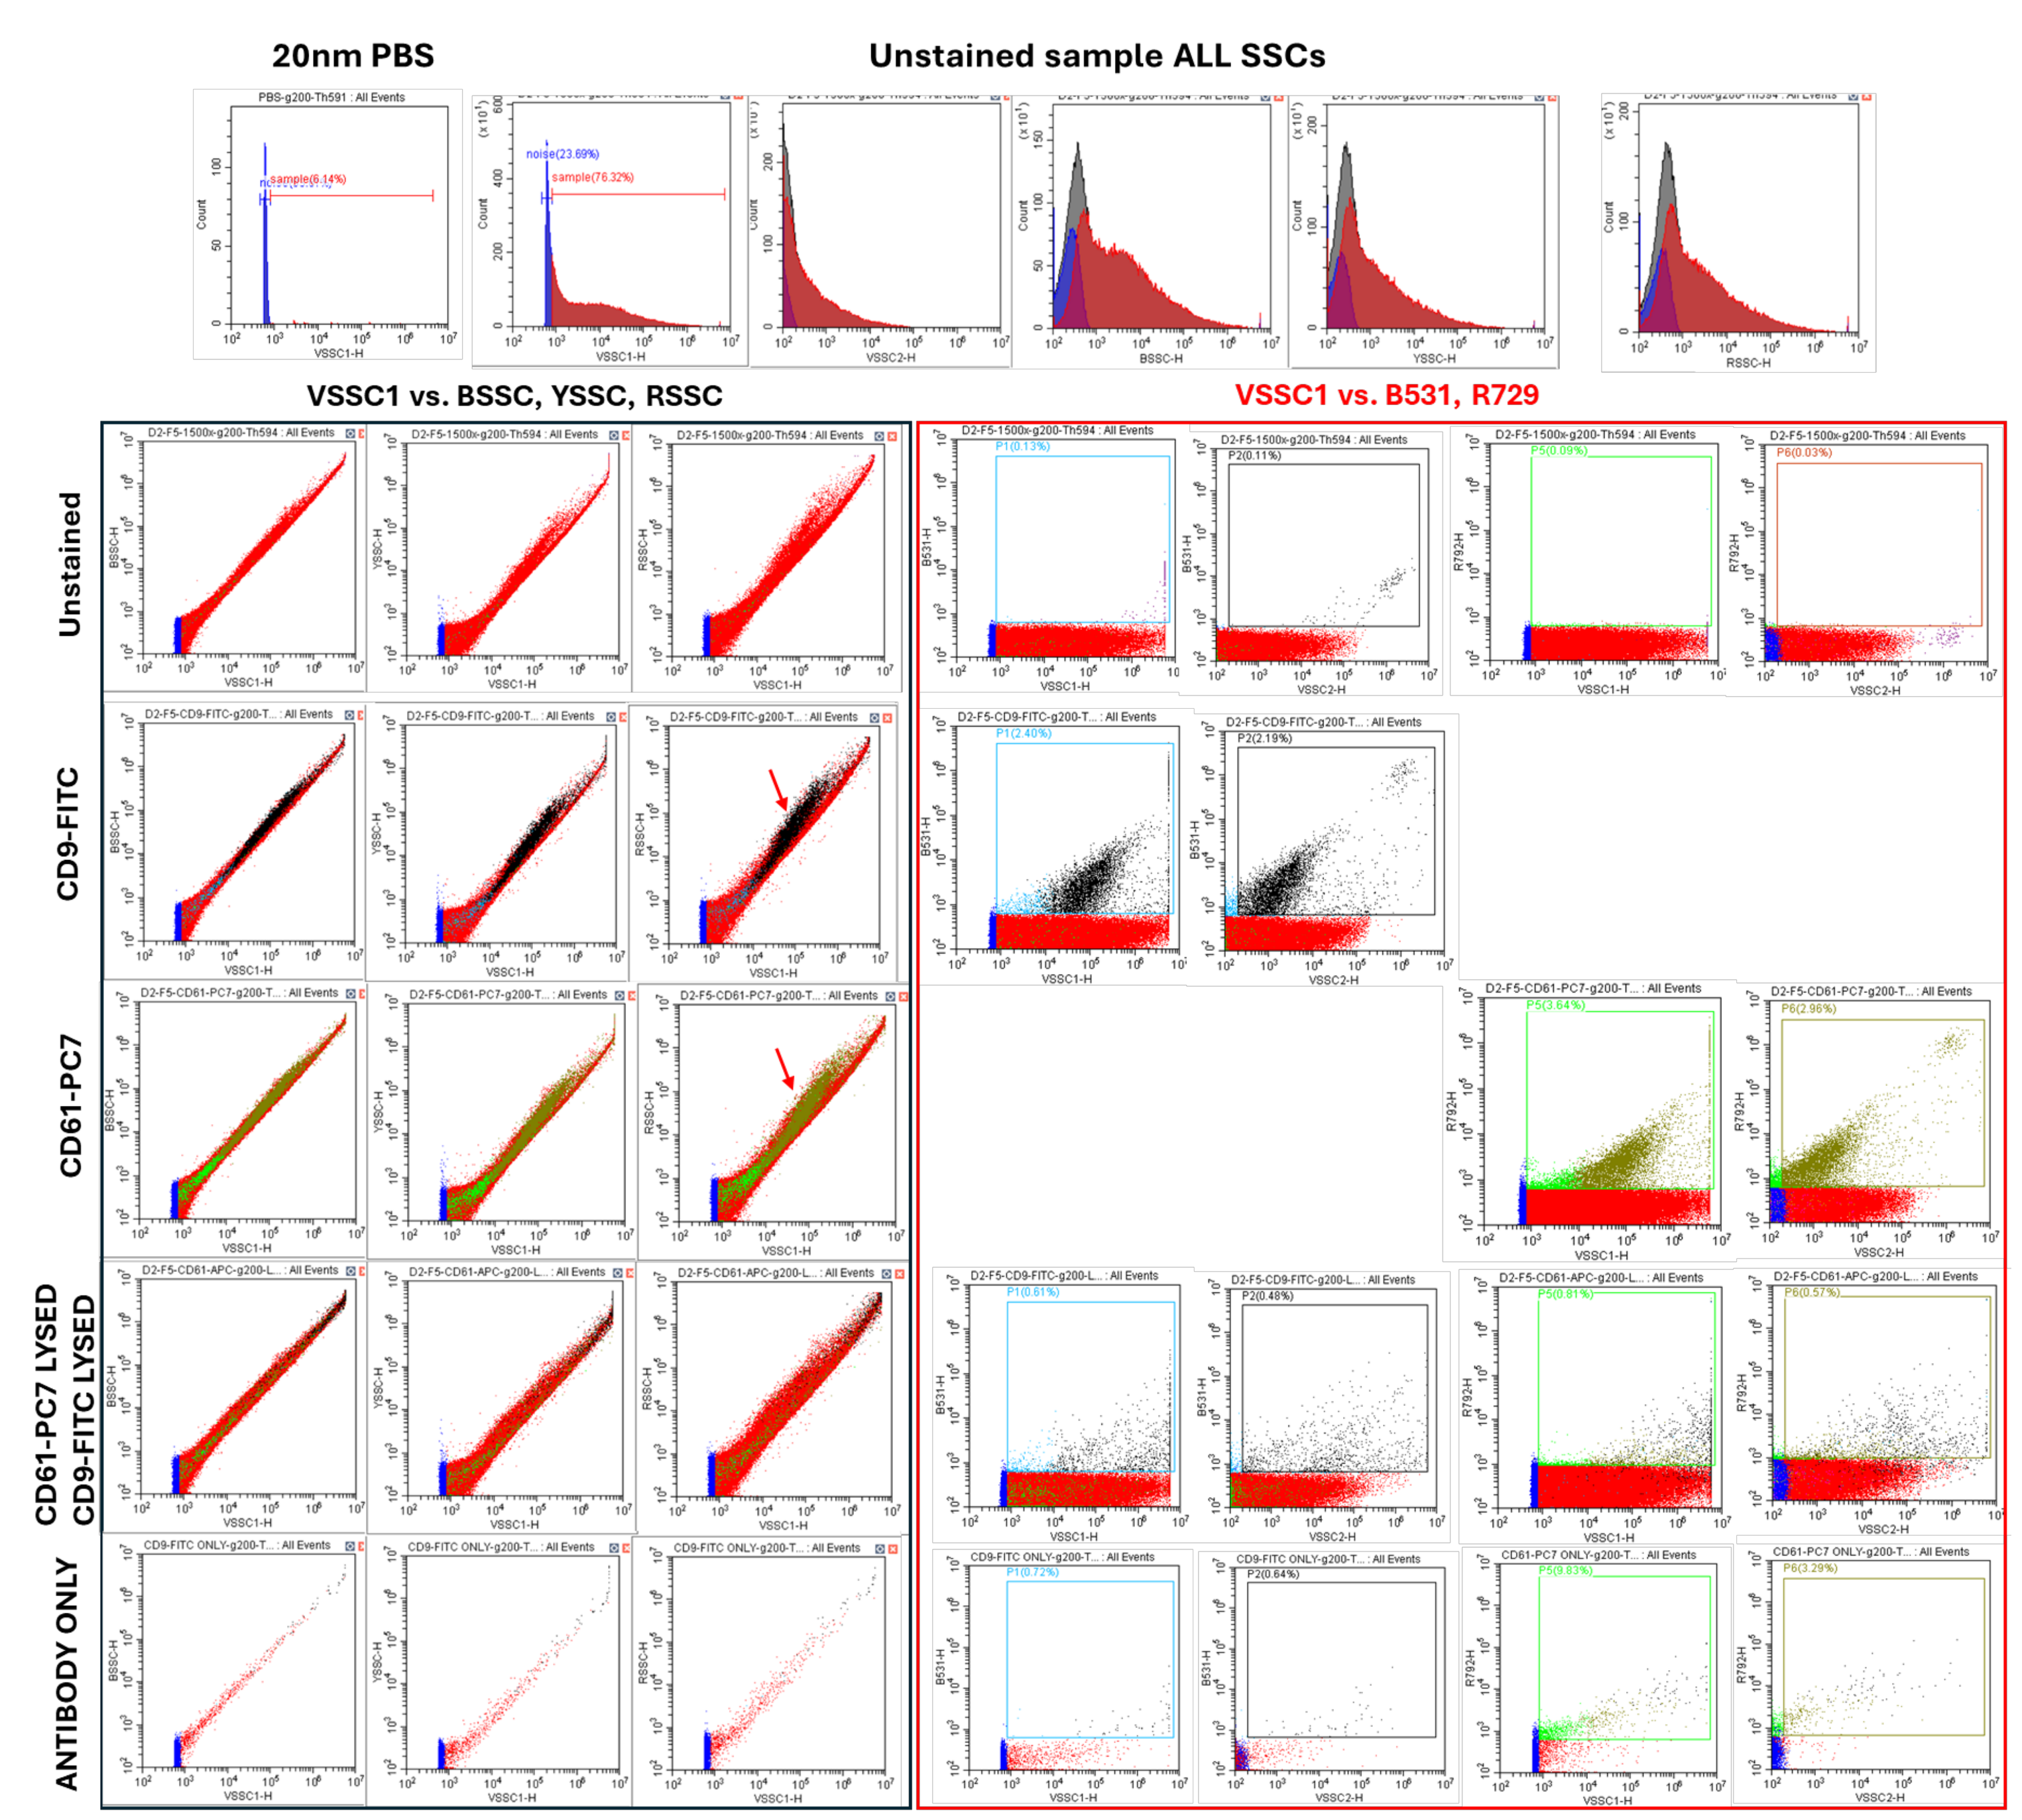
20nm PBS
Unstained sample ALL SSCs
VSSC1 vs. BSSC, YSSC, RSSC
VSSC1 vs. B531, R729
Unstained
CD9-FITC
CD61-PC7
CD61-PC7 LYSED
CD9-FITC LYSED
ANTIBODY ONLY

Figure 3. Antibody Staining of PPP derived EV. CD9-FITC, CD61-PC7 were used concurrently analyzed with VSSC1 vs. RSSC. The Unstained sample is depicted by the red arrow. Positive staining was ensured by controls.

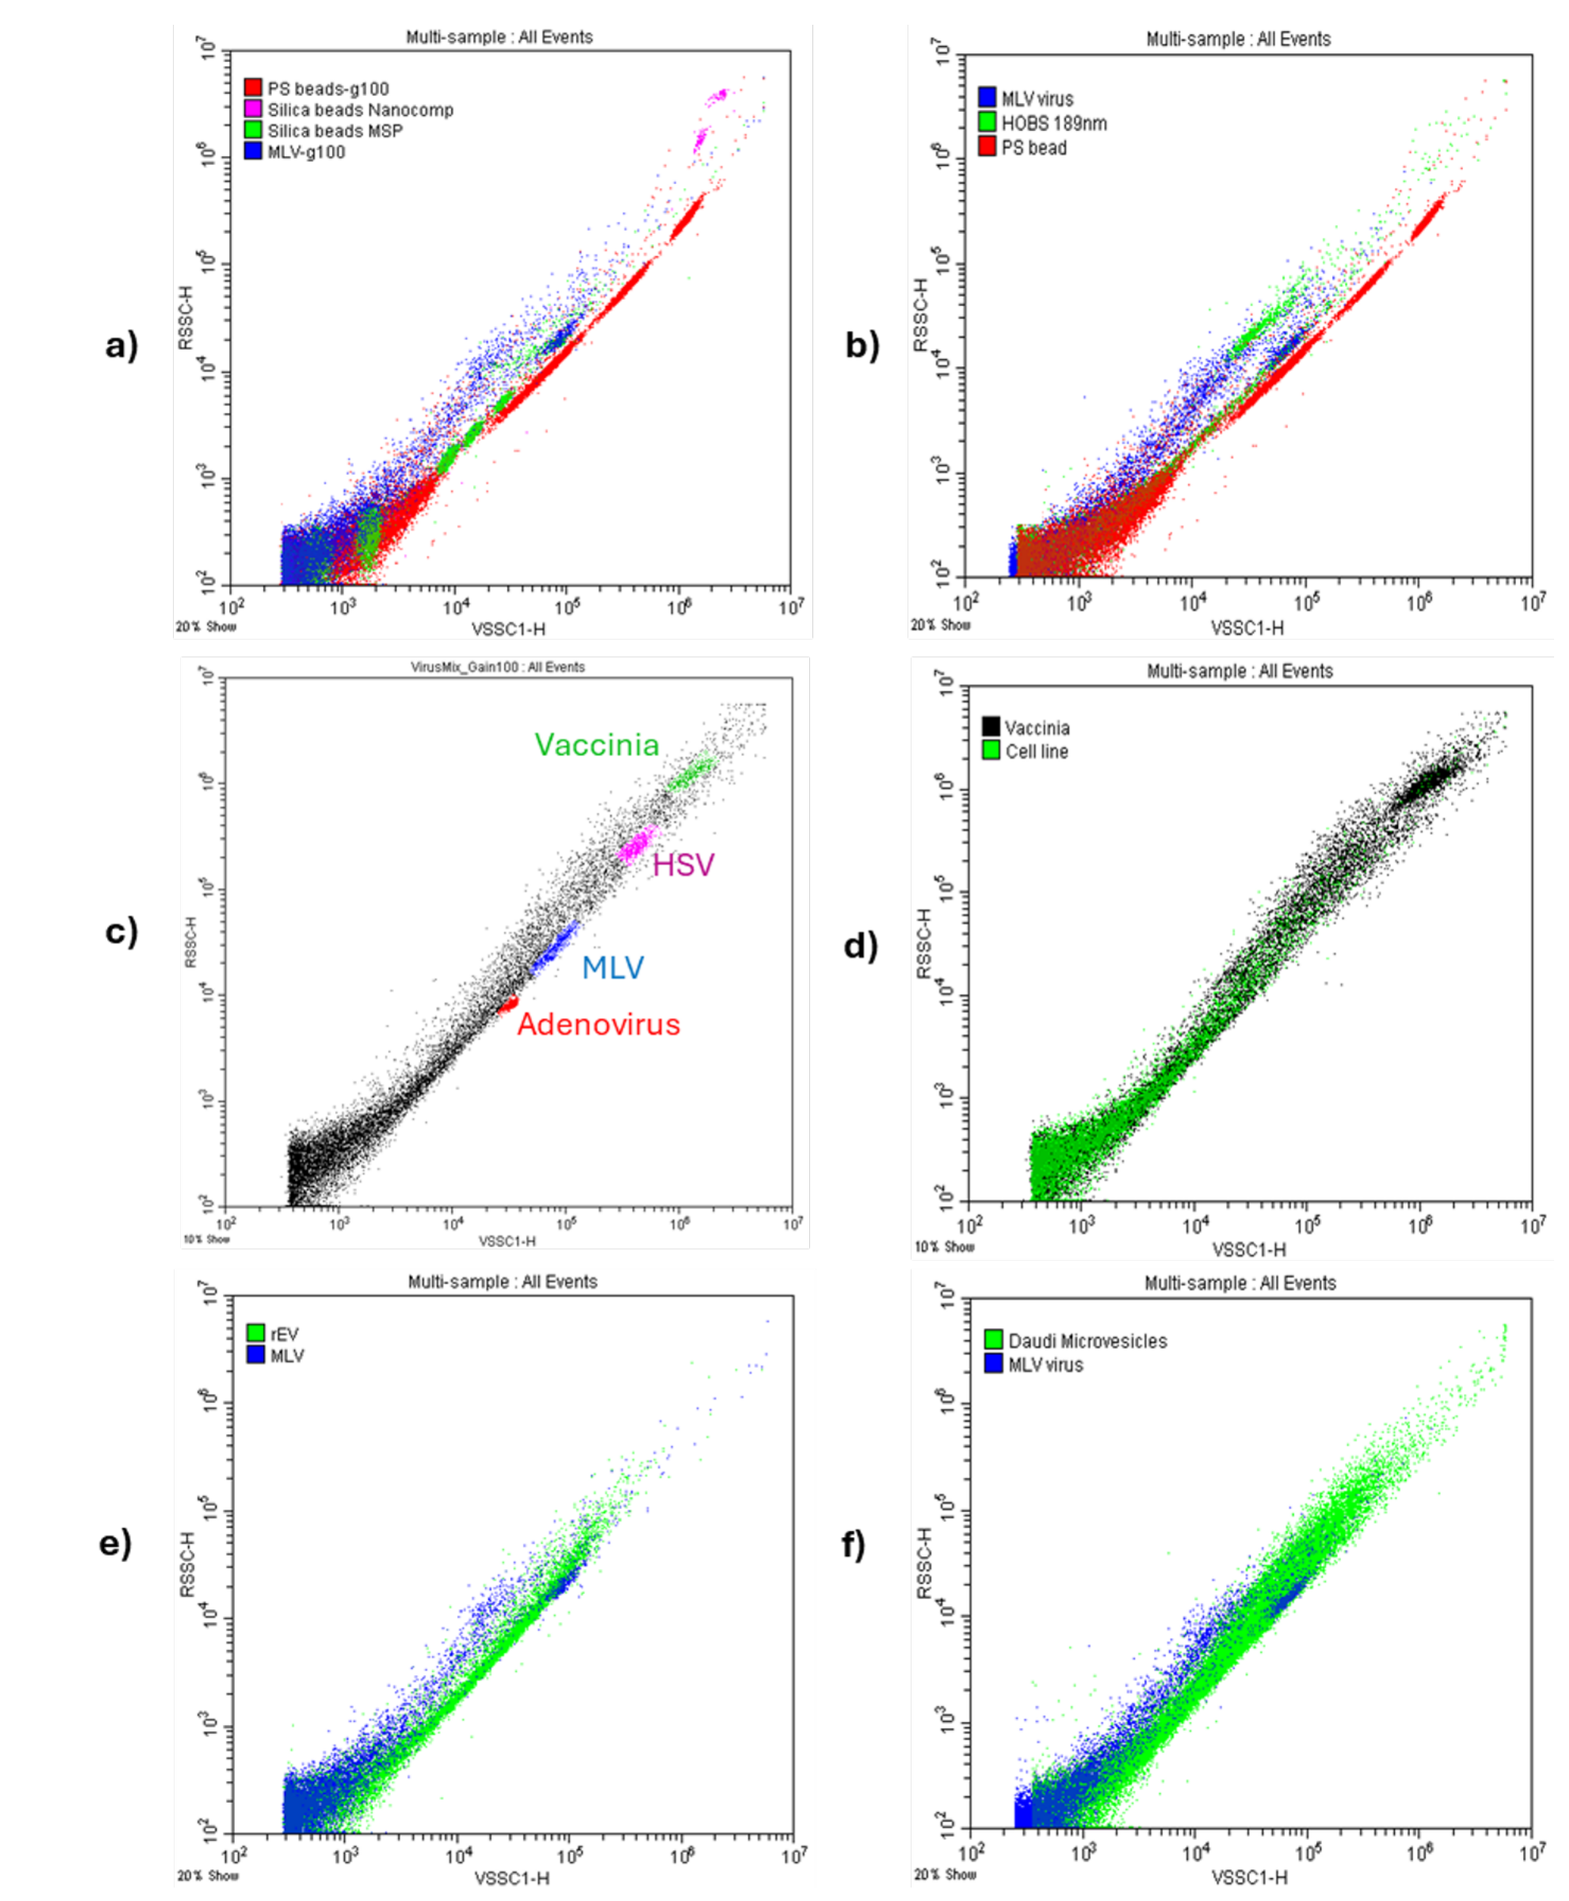
a)
b)
c)
d)
e)
f)
Vaccinia
HSV
MLV
Adenovirus

Figure 4. Multiple scatter analysis of small particles. The combined use of use of VSSC1-H and RSSC-H enables to identify the various controls run. Polystyrene NIST beads with different sizes 44 nm, 80 nm, 100 nm, 144 nm; Silica MSP beads with different sizes: 50 nm, 60 nm, 80 nm, 88 nm, 99 nm; Silica nanoComposix beads with sizes: 309 nm, 510nm, MLV virus (a); Polystyrene NIST mixer beads, MLV virus, Hollow organic beads 189 nm (b); Mixer of MLV virus, Adenovirus, HSV, Vaccinia (c); Vaccinia and cell line control (d); MLV virus and recombinant EVs (e); MLV virus and Daudi microvesicles (f).

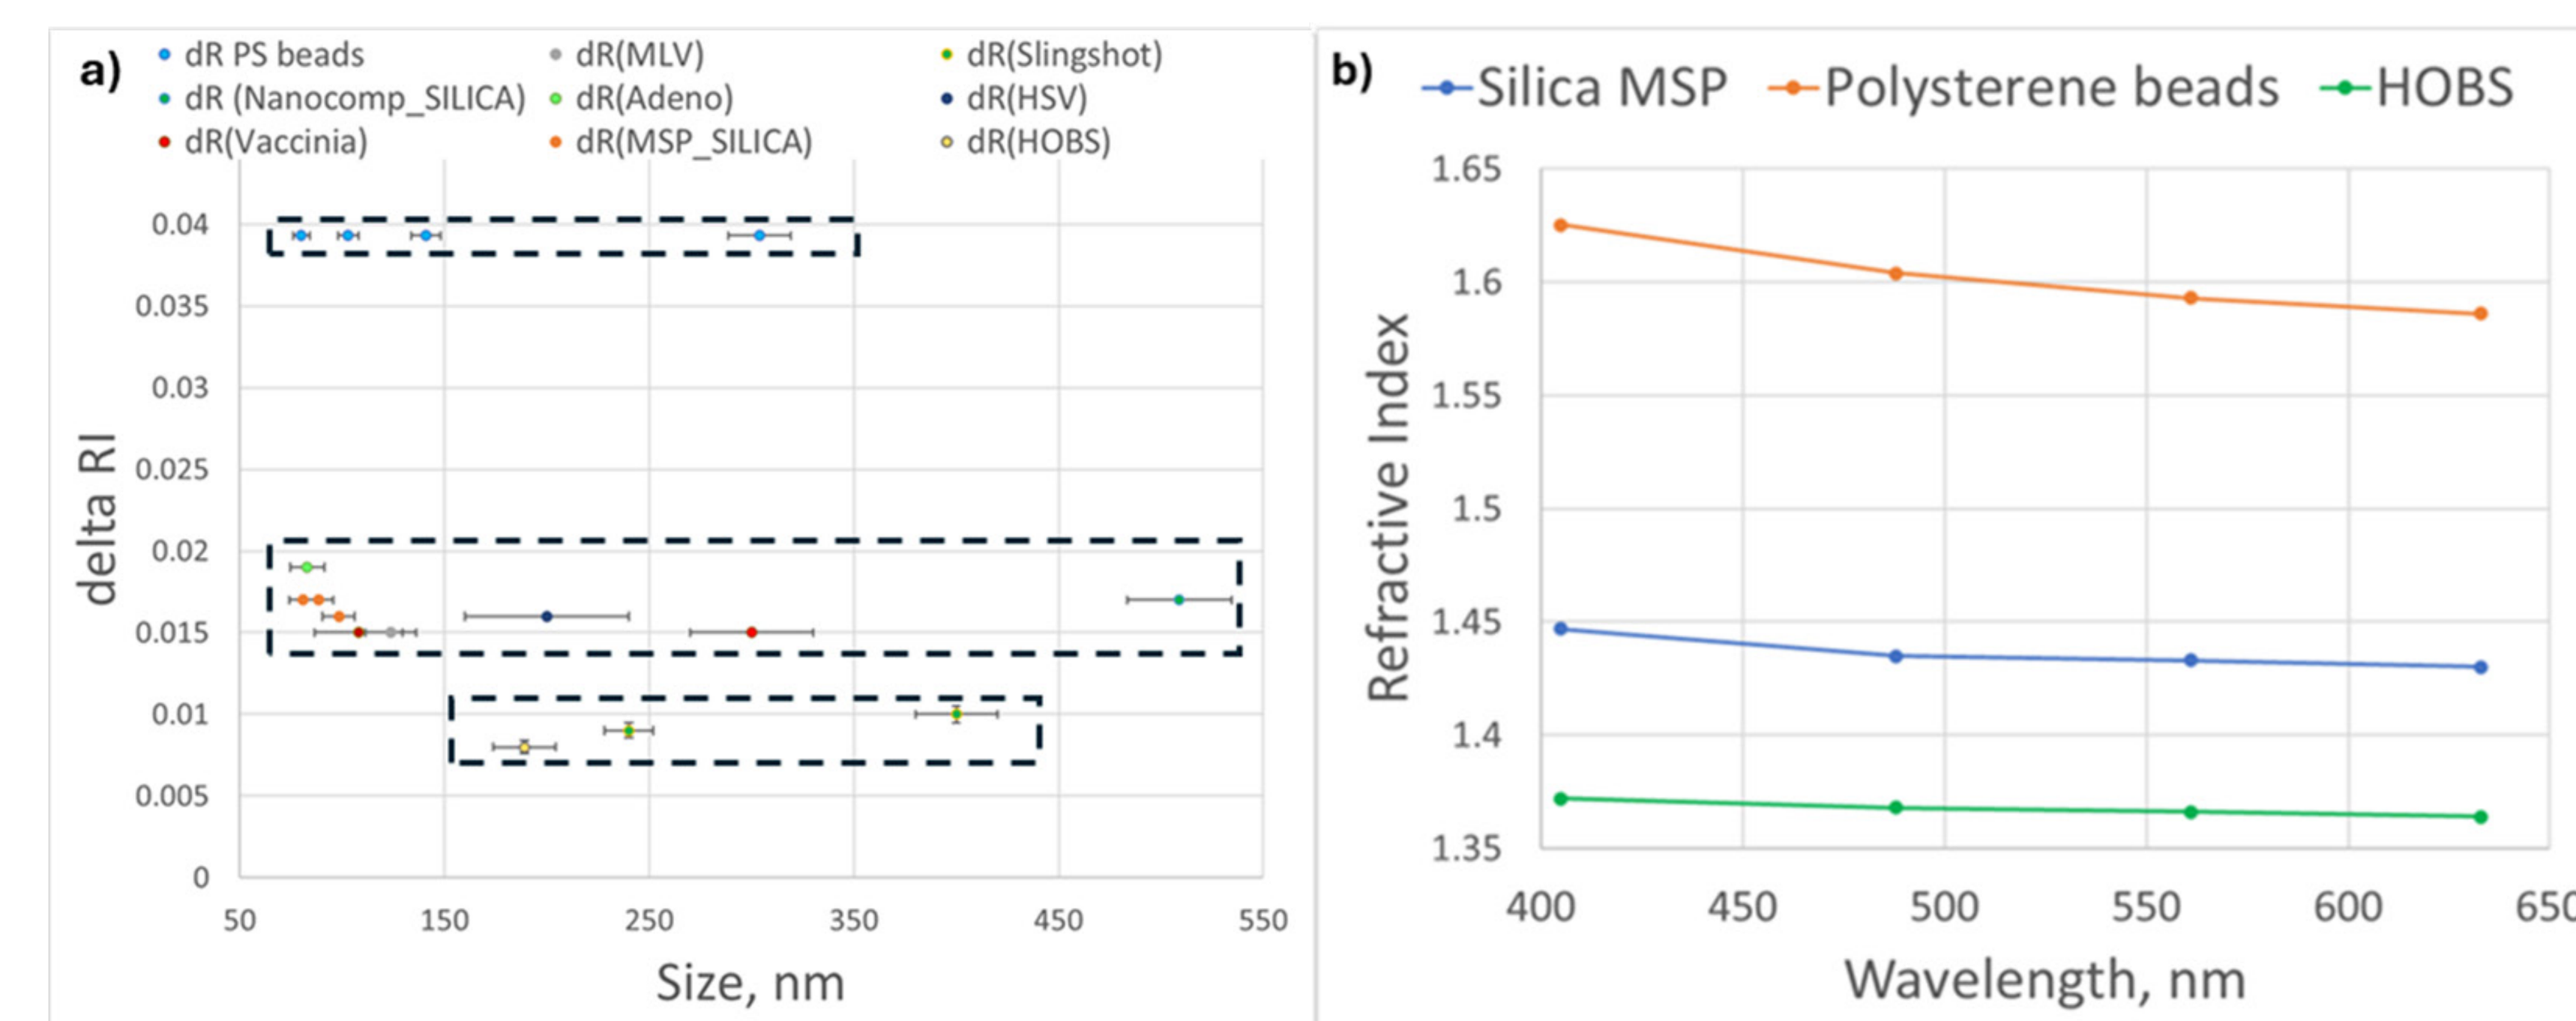
a)
dR PS beads
dR (Nanocomp_SILICA)
dR(Vaccinia)
dR(MLV)
dR(Adeno)
dR(MSP_SILICA)
dR(Slingshot)
dR(HSV)
dR(HOBS)
delta RI
Size, nm
b)
Silica MSP
Polysterene beads
HOBS
Refractive Index
Wavelength, nm

Figure 5. Delta refractive index change between Violet and Red wavelengths. NIST PS beads with different sizes: 44 nm, 80 nm, 100 nm, 144 nm; Silica MSP beads with different sizes: 50 nm, 60 nm, 80 nm, 88 nm, 99 nm; Silica nanoComposix beads with size 510 nm, MLV virus, HSV, Adenovirus, Vaccinia, Hollow organic beads 189 nm. Refractive index vs. Wavelength (b) for PS beads, silica and HOBs beads; Data was generated using FCMPASSTM software.

Conclusions

The CytoFLEX nano Flow Cytometer enables the detection of small changes in refractive indices across multiple wavelength scatters, allowing for the differentiation of populations of different natures. It also enables the detection of smaller-sized populations compared to conventional flow cytometers, improving the ability to characterize samples. We demonstrate the importance of calibrating arbitrary units of MSI to absolute units of size and refractive index.

Acknowledgements

Cytiva R&D team Hailey Kim, Andrew Kondratowicz, Adam Crowe

For Research Use Only. Not for use in diagnostic procedures.
© 2025 Beckman Coulter, Inc. All rights reserved. Beckman Coulter, the stylized logo, and the Beckman Coulter product and service marks mentioned herein are trademarks or registered trademarks of Beckman Coulter, Inc. in the United States and other countries.
All other trademarks are the property of their respective owners.

For Beckman Coulter's worldwide office locations and phone numbers, please visit contact us at **beckman.com**
2025-GBL-EN-107667-v2