Label-free Technology to Assess Protein Loadings on the Surface of Biological Sample

Evgenia Kim[1], Boi Hoa San[1], Anisha Jose[2], Sergei Gulnik[1],
[1]Research and Development, Beckman Coulter Life Sciences, Miami, FL 33196.
[2]Commercial Excellence, Beckman Coulter Life Sciences.

Introduction

Interest in identifying and characterizing nano-sized particles like extracellular vesicles (EVs), viruses, and liposomes is growing due to their wide applications in biomedicine. Flow cytometry offers a powerful, high-throughput method for single-particle detection, particularly exemplified by the CytoFLEX nano flow cytometer, which is designed for nanoparticle characterization. This instrument features two violet side scatter channels with a large dynamic range and additional channels for blue, yellow, and red scatter signals. The high scatter sensitivity of the CytoFLEX nano flow cytometer enables the detection of small changes in medium scatter intensities caused by varying amounts of protein loaded on the surface of a biological sample. In this study, we demonstrate the capabilities of the instrument to detect and analyze the change in refractive index caused by different numbers of antibodies bound to the surface of Murine Leukemia Virus (MLV).

Sample preparation

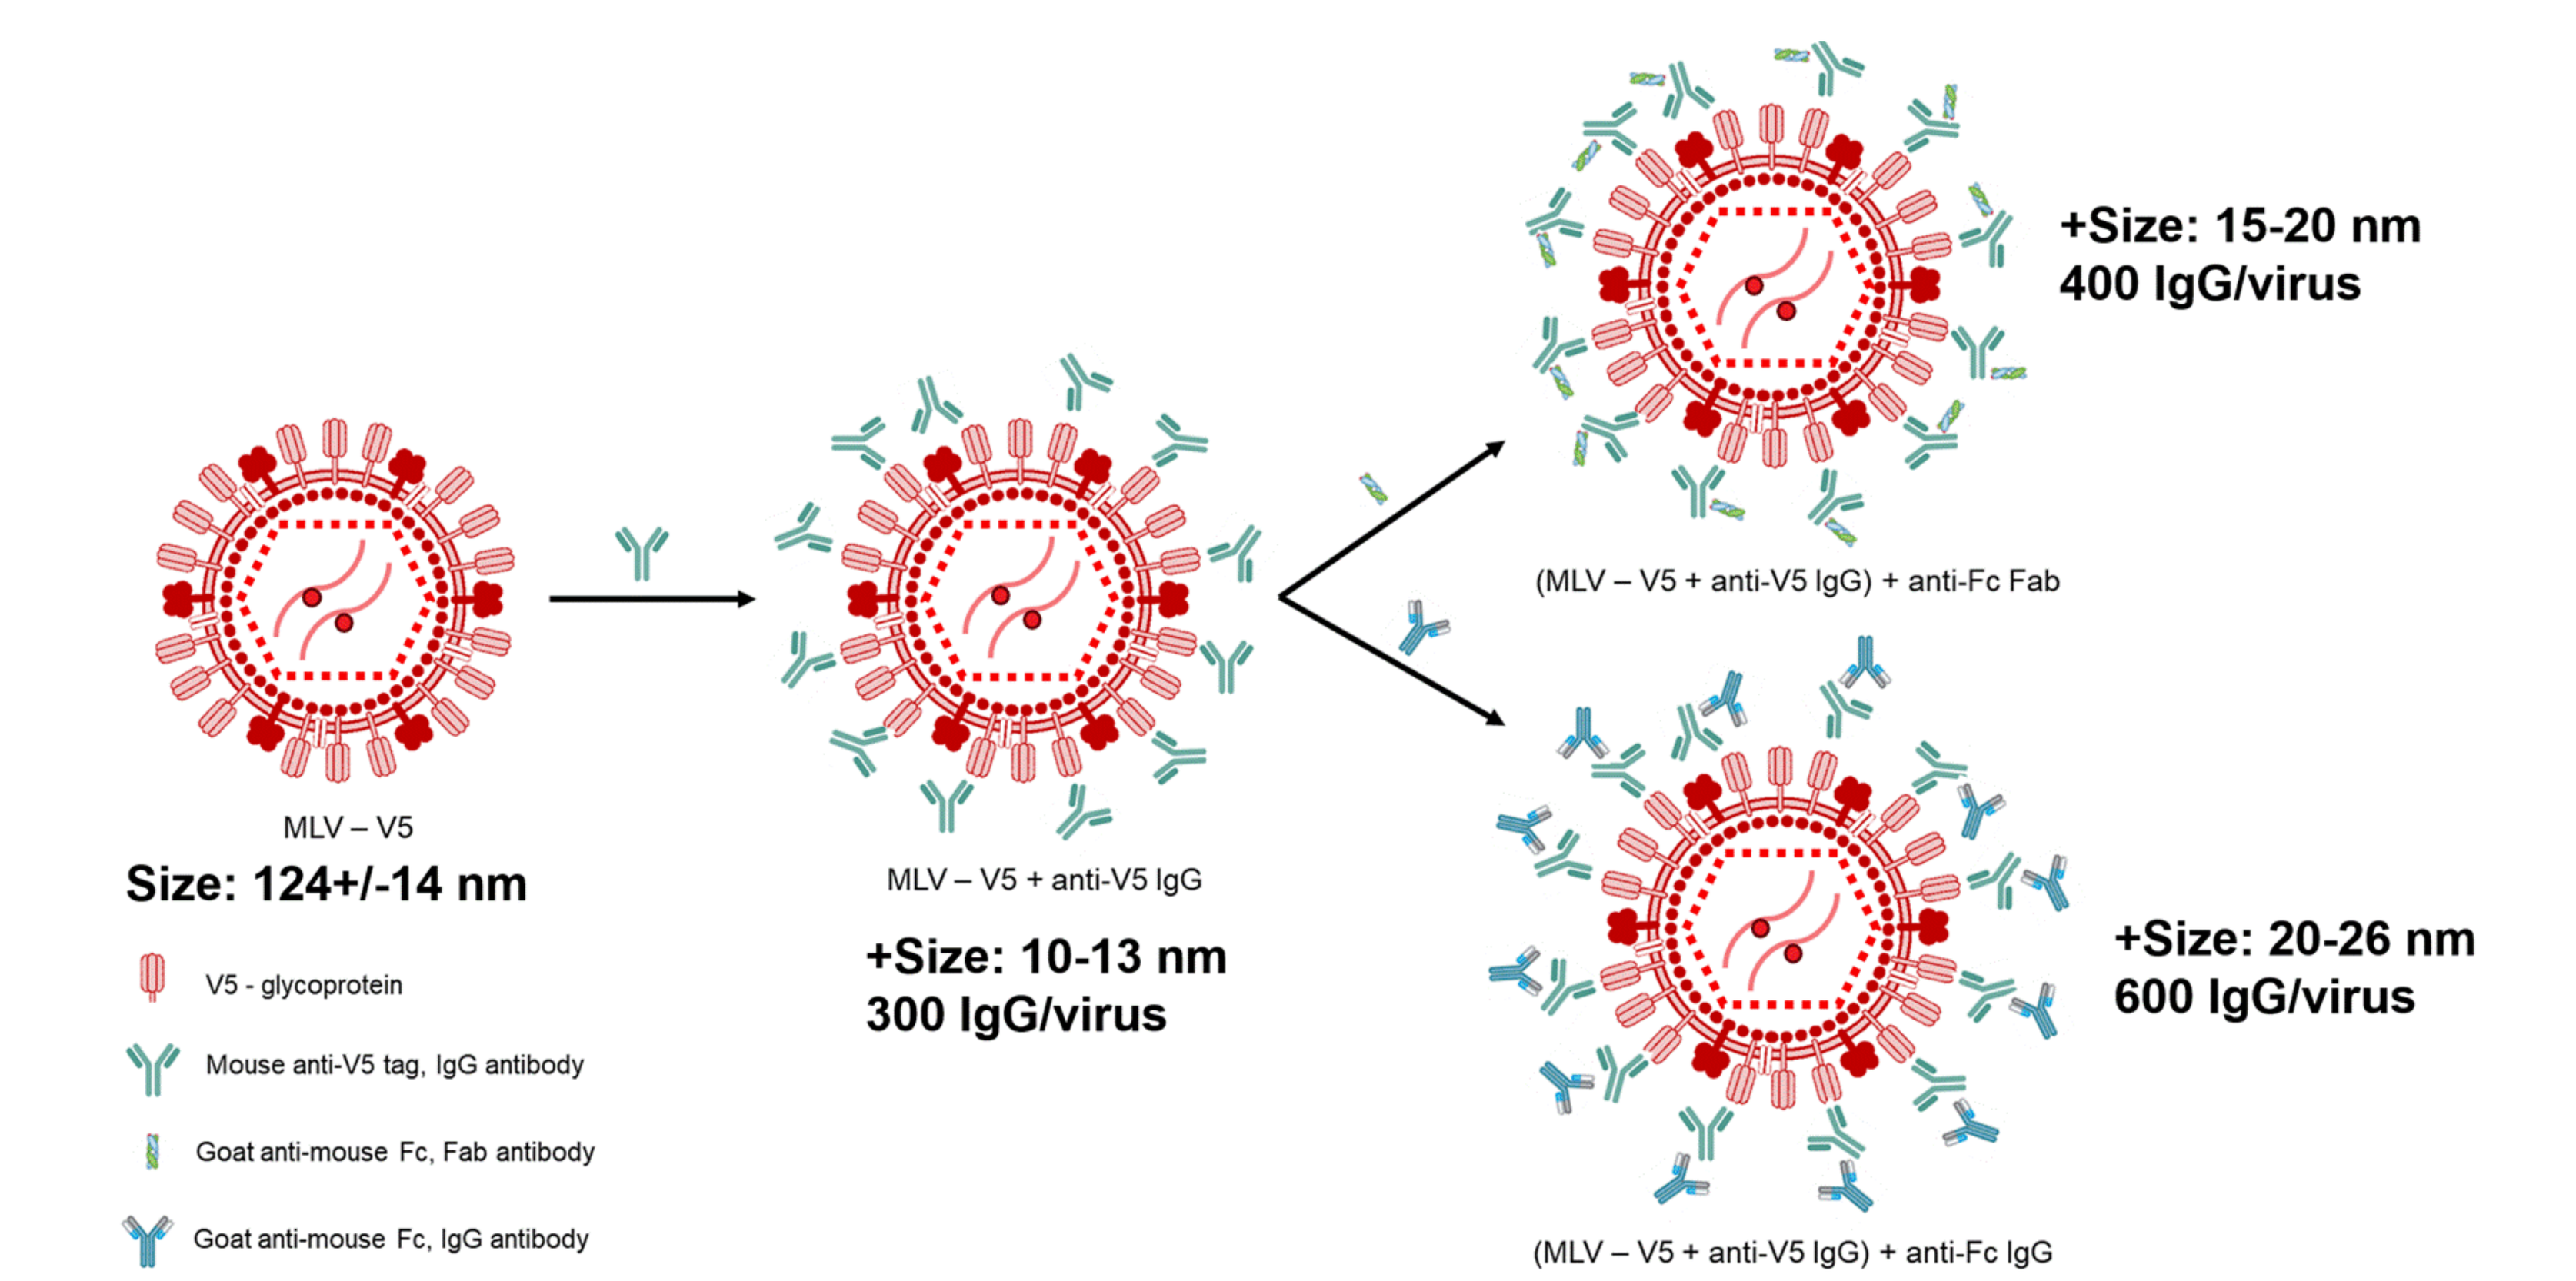
MLV – V5
Size: 124+/-14 nm
MLV – V5 + anti-V5 IgG
+Size: 10-13 nm
300 IgG/virus
(MLV – V5 + anti-V5 IgG) + anti-Fc Fab
+Size: 15-20 nm
400 IgG/virus
(MLV – V5 + anti-V5 IgG) + anti-Fc IgG
+Size: 20-26 nm
600 IgG/virus
V5 - glycoprotein
Mouse anti-V5 tag, IgG antibody
Goat anti-mouse Fc, Fab antibody
Goat anti-mouse Fc, IgG antibody

Figure 1. Sample preparation. MLV-V5 was stained with different coatings of antibodies.

Results

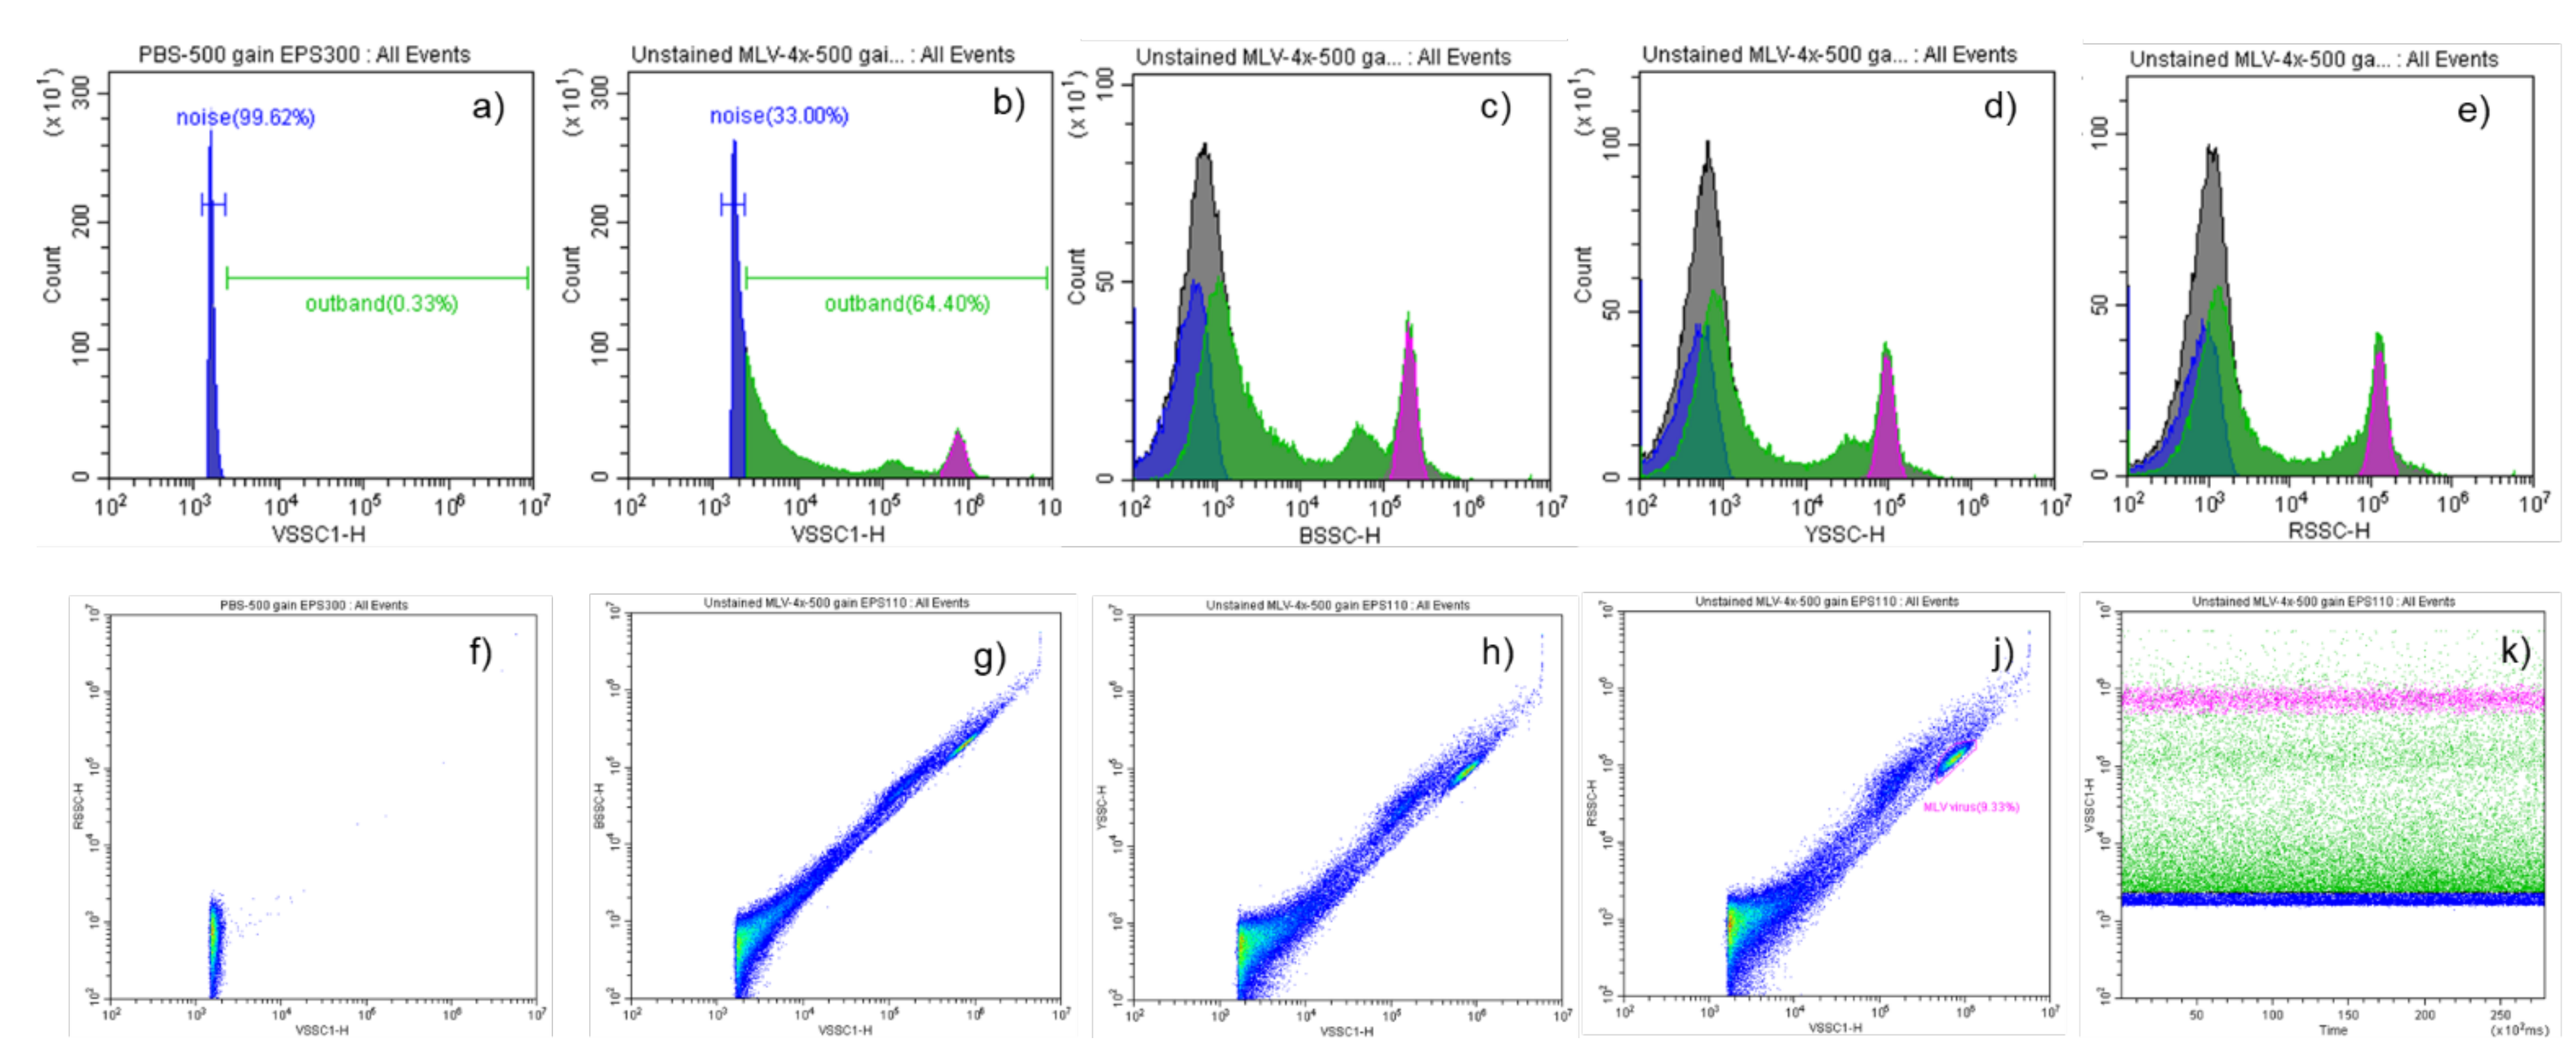
PBS-500 gain EPS300 : All Events
noise(99.62%)
outband(0.33%)
noise(33.00%)
outband(64.40%)
VSSC1-H
BSSC-H
YSSC-H
RSSC-H

Figure 2. Analysis of Unstained MLV sample. Single SSC wavelength histograms (b–e) and multiwavelength dot plots (g–j). Histogram (a) and dot plot (f) of noise from PBS. Time monitor graph (k).

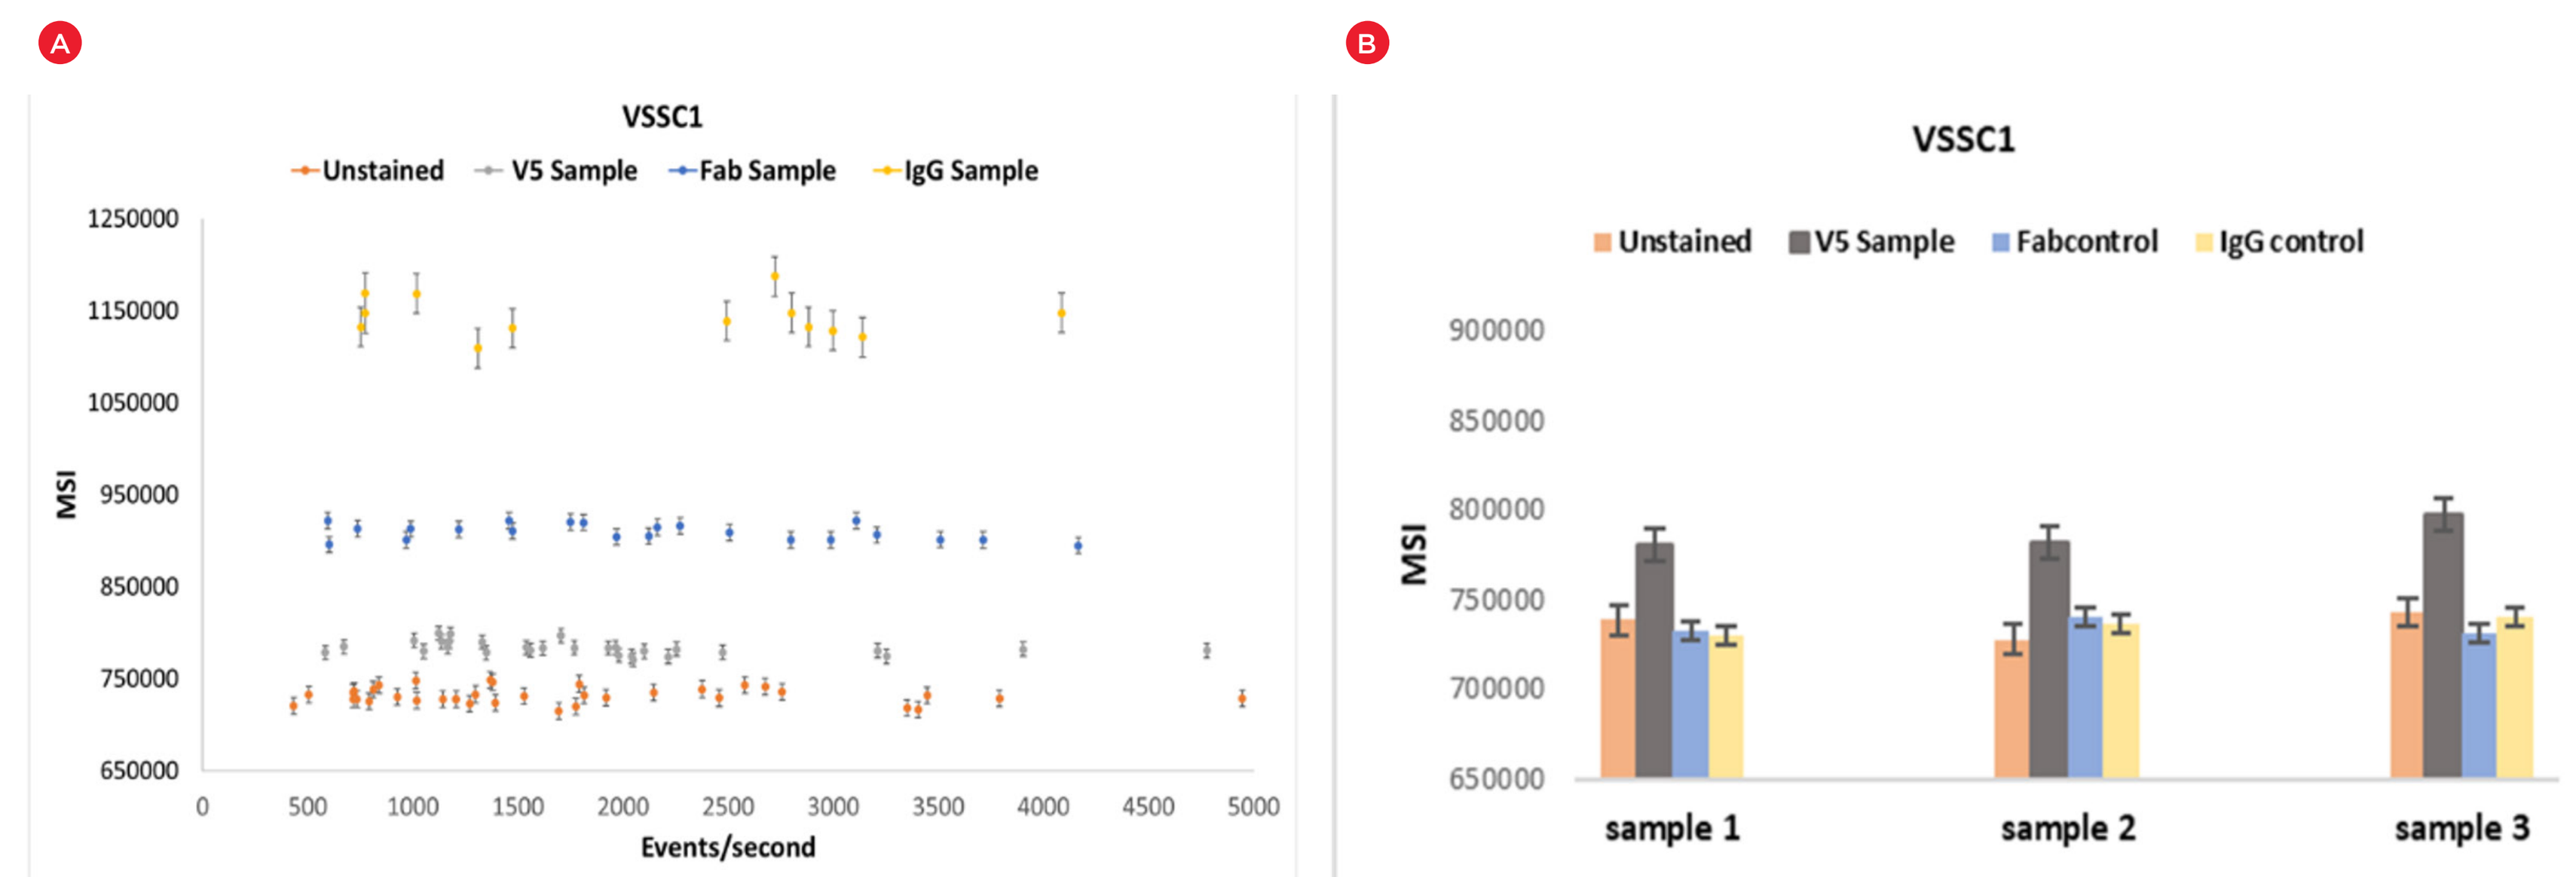
VSSC1
Unstained
V5 Sample
Fab Sample
IgG Sample
MSI
Events/second
Fabcontrol
IgG control
sample 1
sample 2
sample 3

Figure 3. Event per second (EPS) rate to prevent swarming across lasers. An EPS rate to prevent swarming across all lasers when plotting mean side scatter intensities (MSI) against events per second against a) Violet laser. Unstained MLV virus control, virus with mouse anti-V5 tag antibody, virus with Goat anti-mouse Fc-Fab (without anti-V5 IgG), virus with Goat anti-mouse Fc-IgG (without anti-V5 IgG) for b) Violet laser.

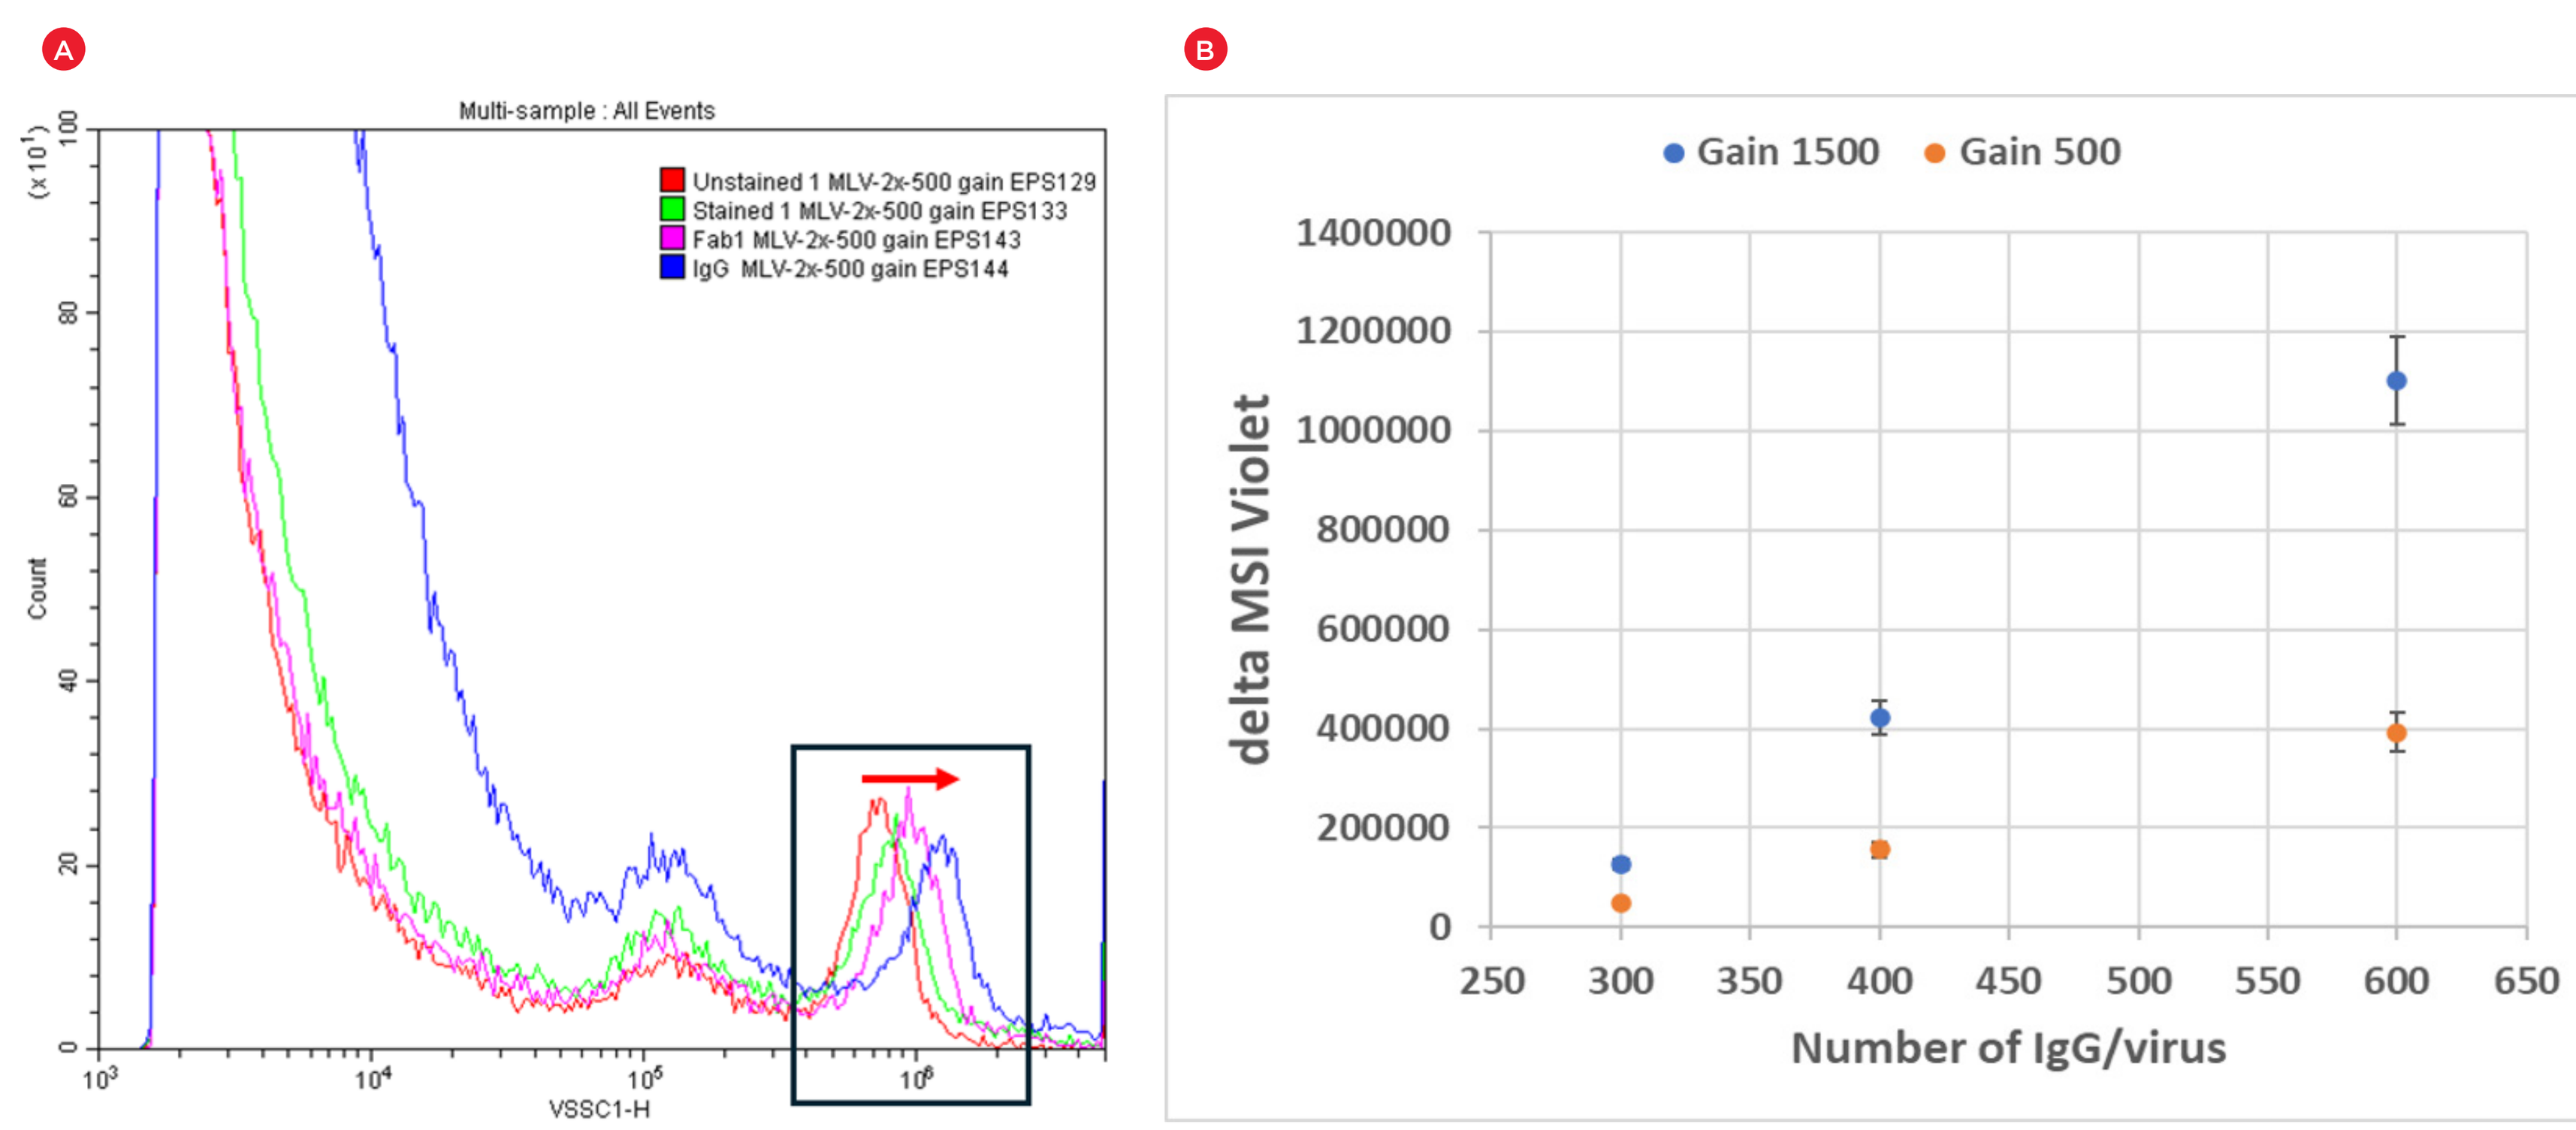
Multi-sample : All Events
Unstained 1 MLV-2x-500 gain EPS129
Stained 1 MLV-2x-500 gain EPS133
Fab1 MLV-2x-500 gain EPS143
IgG MLV-2x-500 gain EPS144
Count
VSSC1-H
Gain 1500
Gain 500
delta MSI Violet
Number of IgG/virus

Figure 4. Violet scatter and delta medium scatter intensity of samples. Overlay of histograms from Violet (a) scatter for Unstained MLV, Stained anti-V5 IgG, virus with anti-V5+ anti-Fc-Fab, virus with anti-V5+ anti-Fc-IgG; b) delta medium scatter intensity as a function of number of IgG/virus.

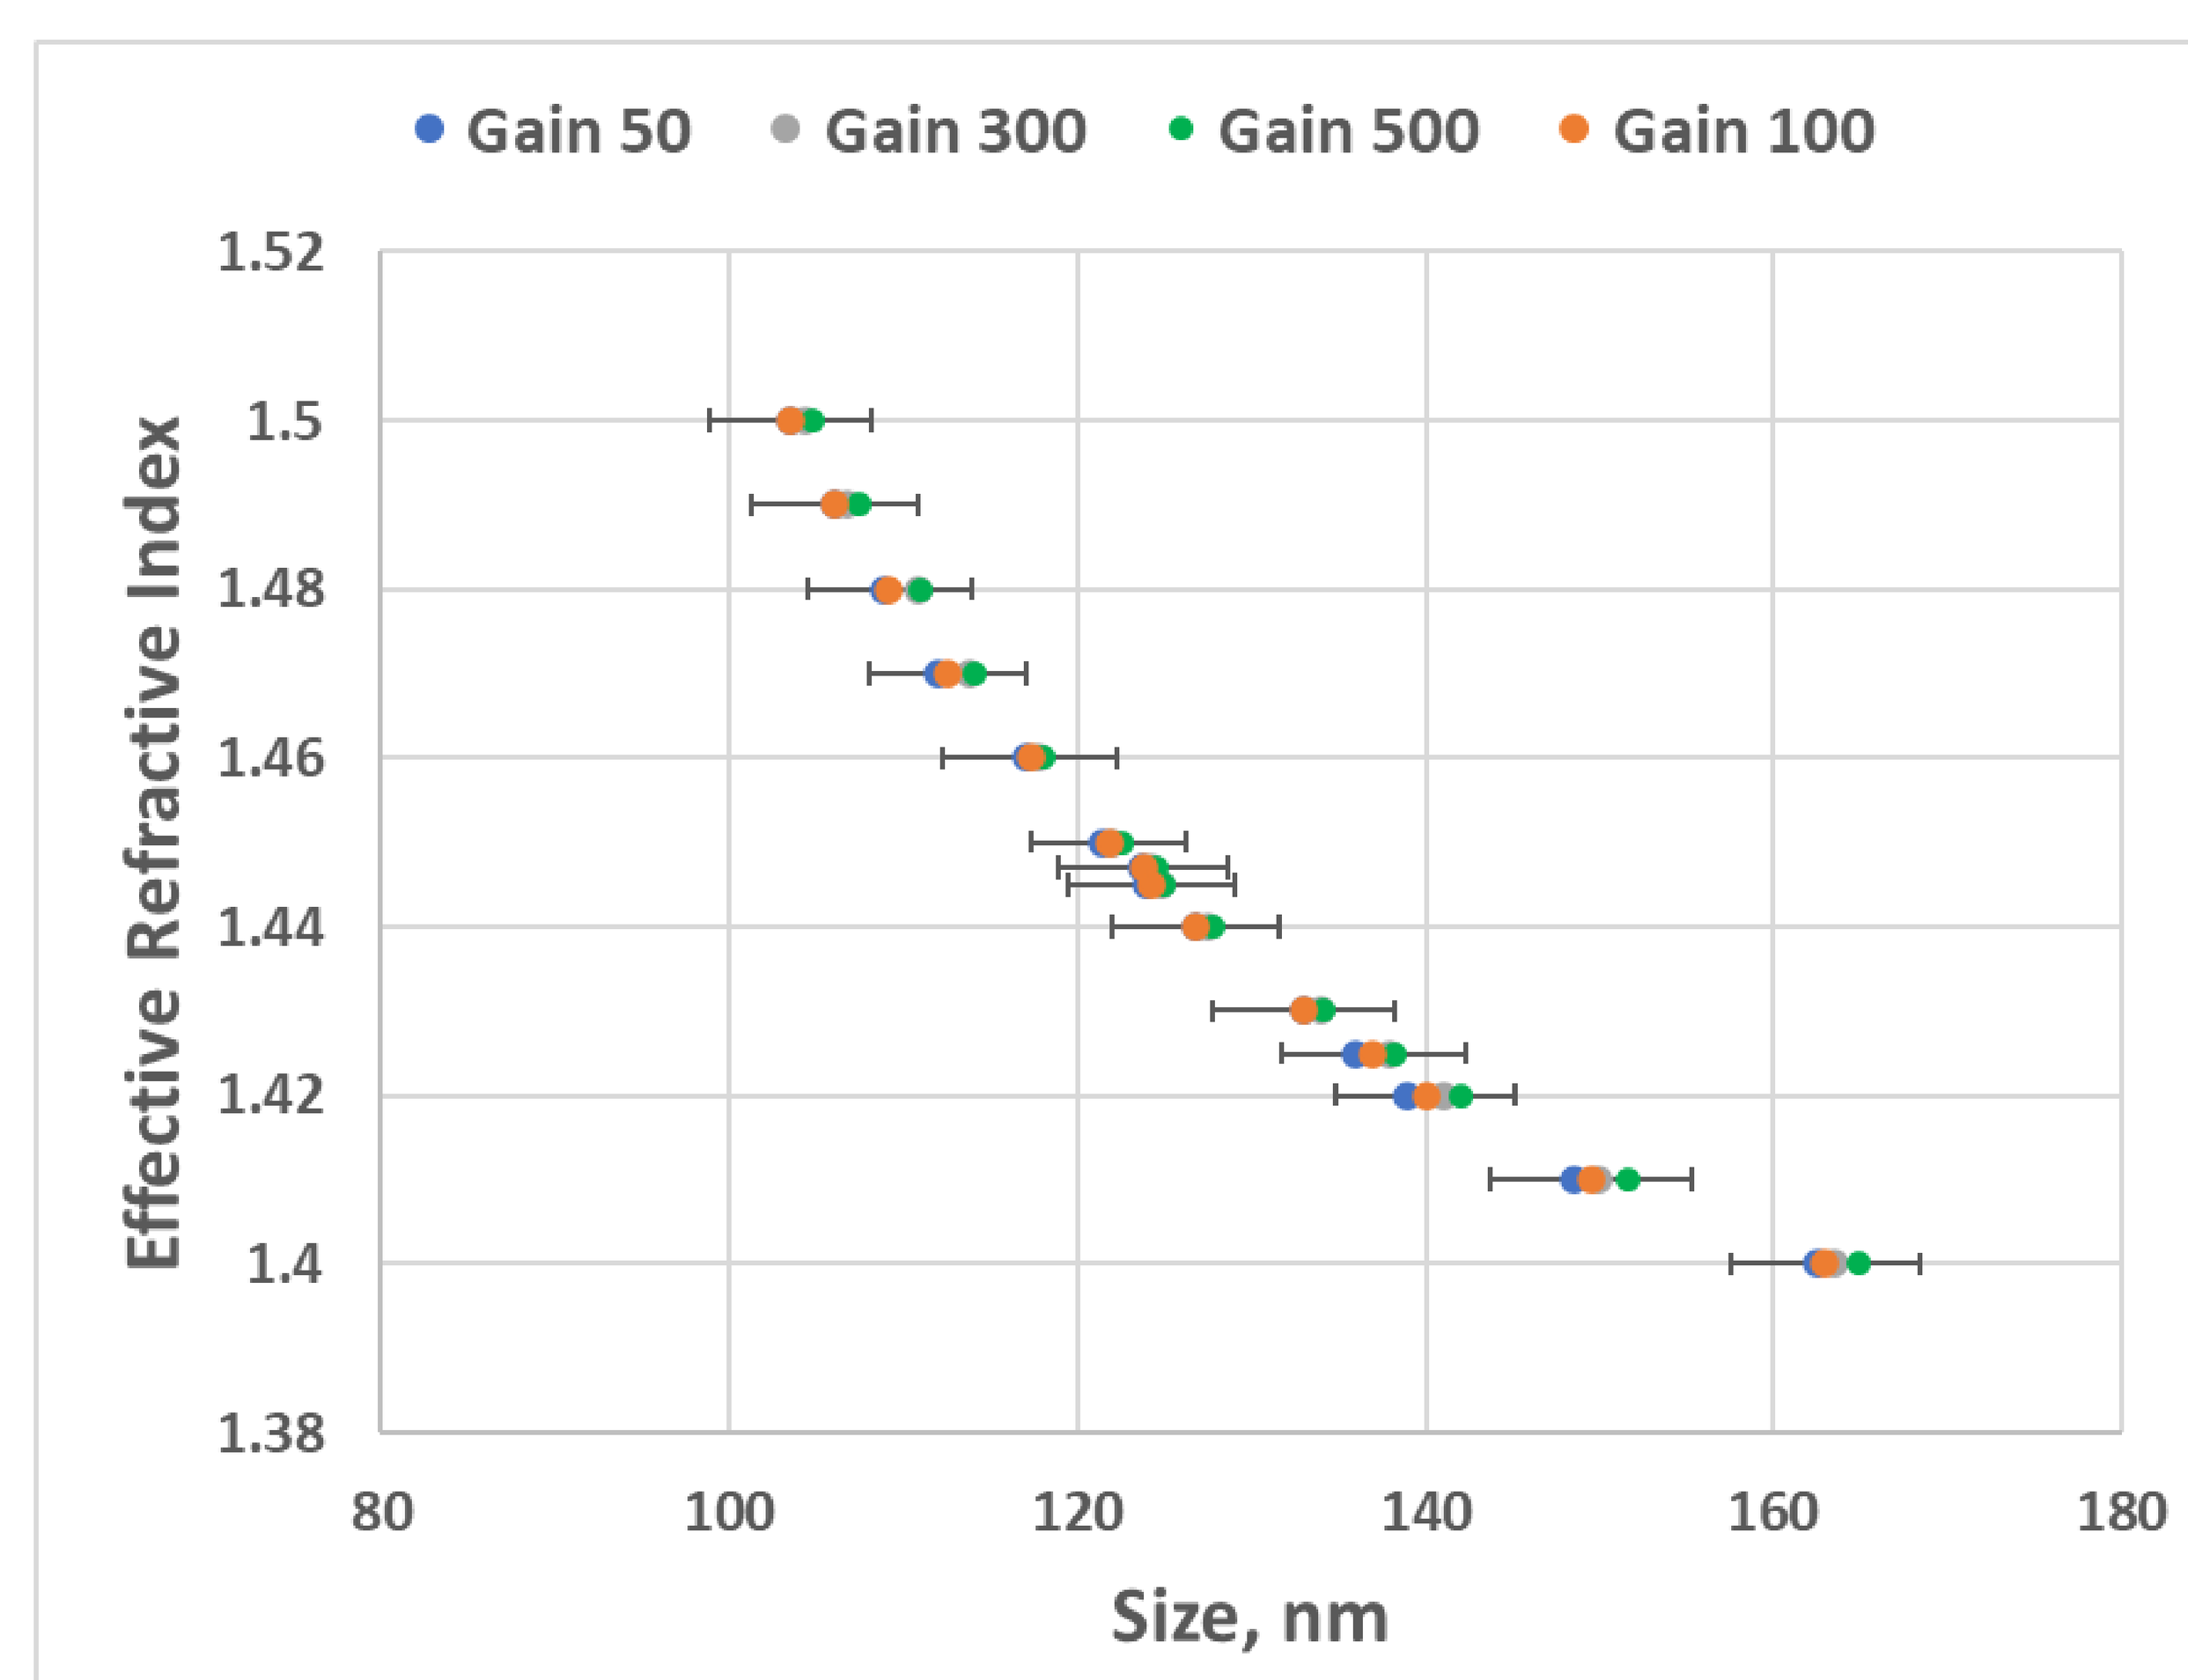
Gain 50
Gain 300
Gain 500
Gain 100
Effective Refractive Index
Size, nm

Figure 5. FCM$_{PASS}$™ software calibration. Calibration for unstained MLV virus using solid modeling.

Solid modeling	Gain 50	Gain 100	Gain 300	Gain 500	CV
Effective RI	Size, nm	Size, nm	Size, nm	Size, nm	4.7
1.5	103.5	103.5	104.3	104.83	4.8
1.49	106	106	106.8	107.41	4.7
1.48	108.9	109.2	110.8	111	4.5
1.47	112	112.5	113.8	114.15	5
1.46	117	117.3	117.8	118	4.44
1.45	121.5	121.8	122	112.6	4.9
1.447	123.7	123.8	124.2	124.5	4.8
1.445	124	124.2	124.8	125	4.8
1.44	126.8	126.8	127.5	127.8	5.2
1.43	133	133	134	134.2	5.3
1.425	136	137	138	138.3	5.1
1.42	139	140	141	142	5.8
1.41	148.5	149.5	150	151.6	5.4
1.4	162.5	163	163.5	164.9	5.5

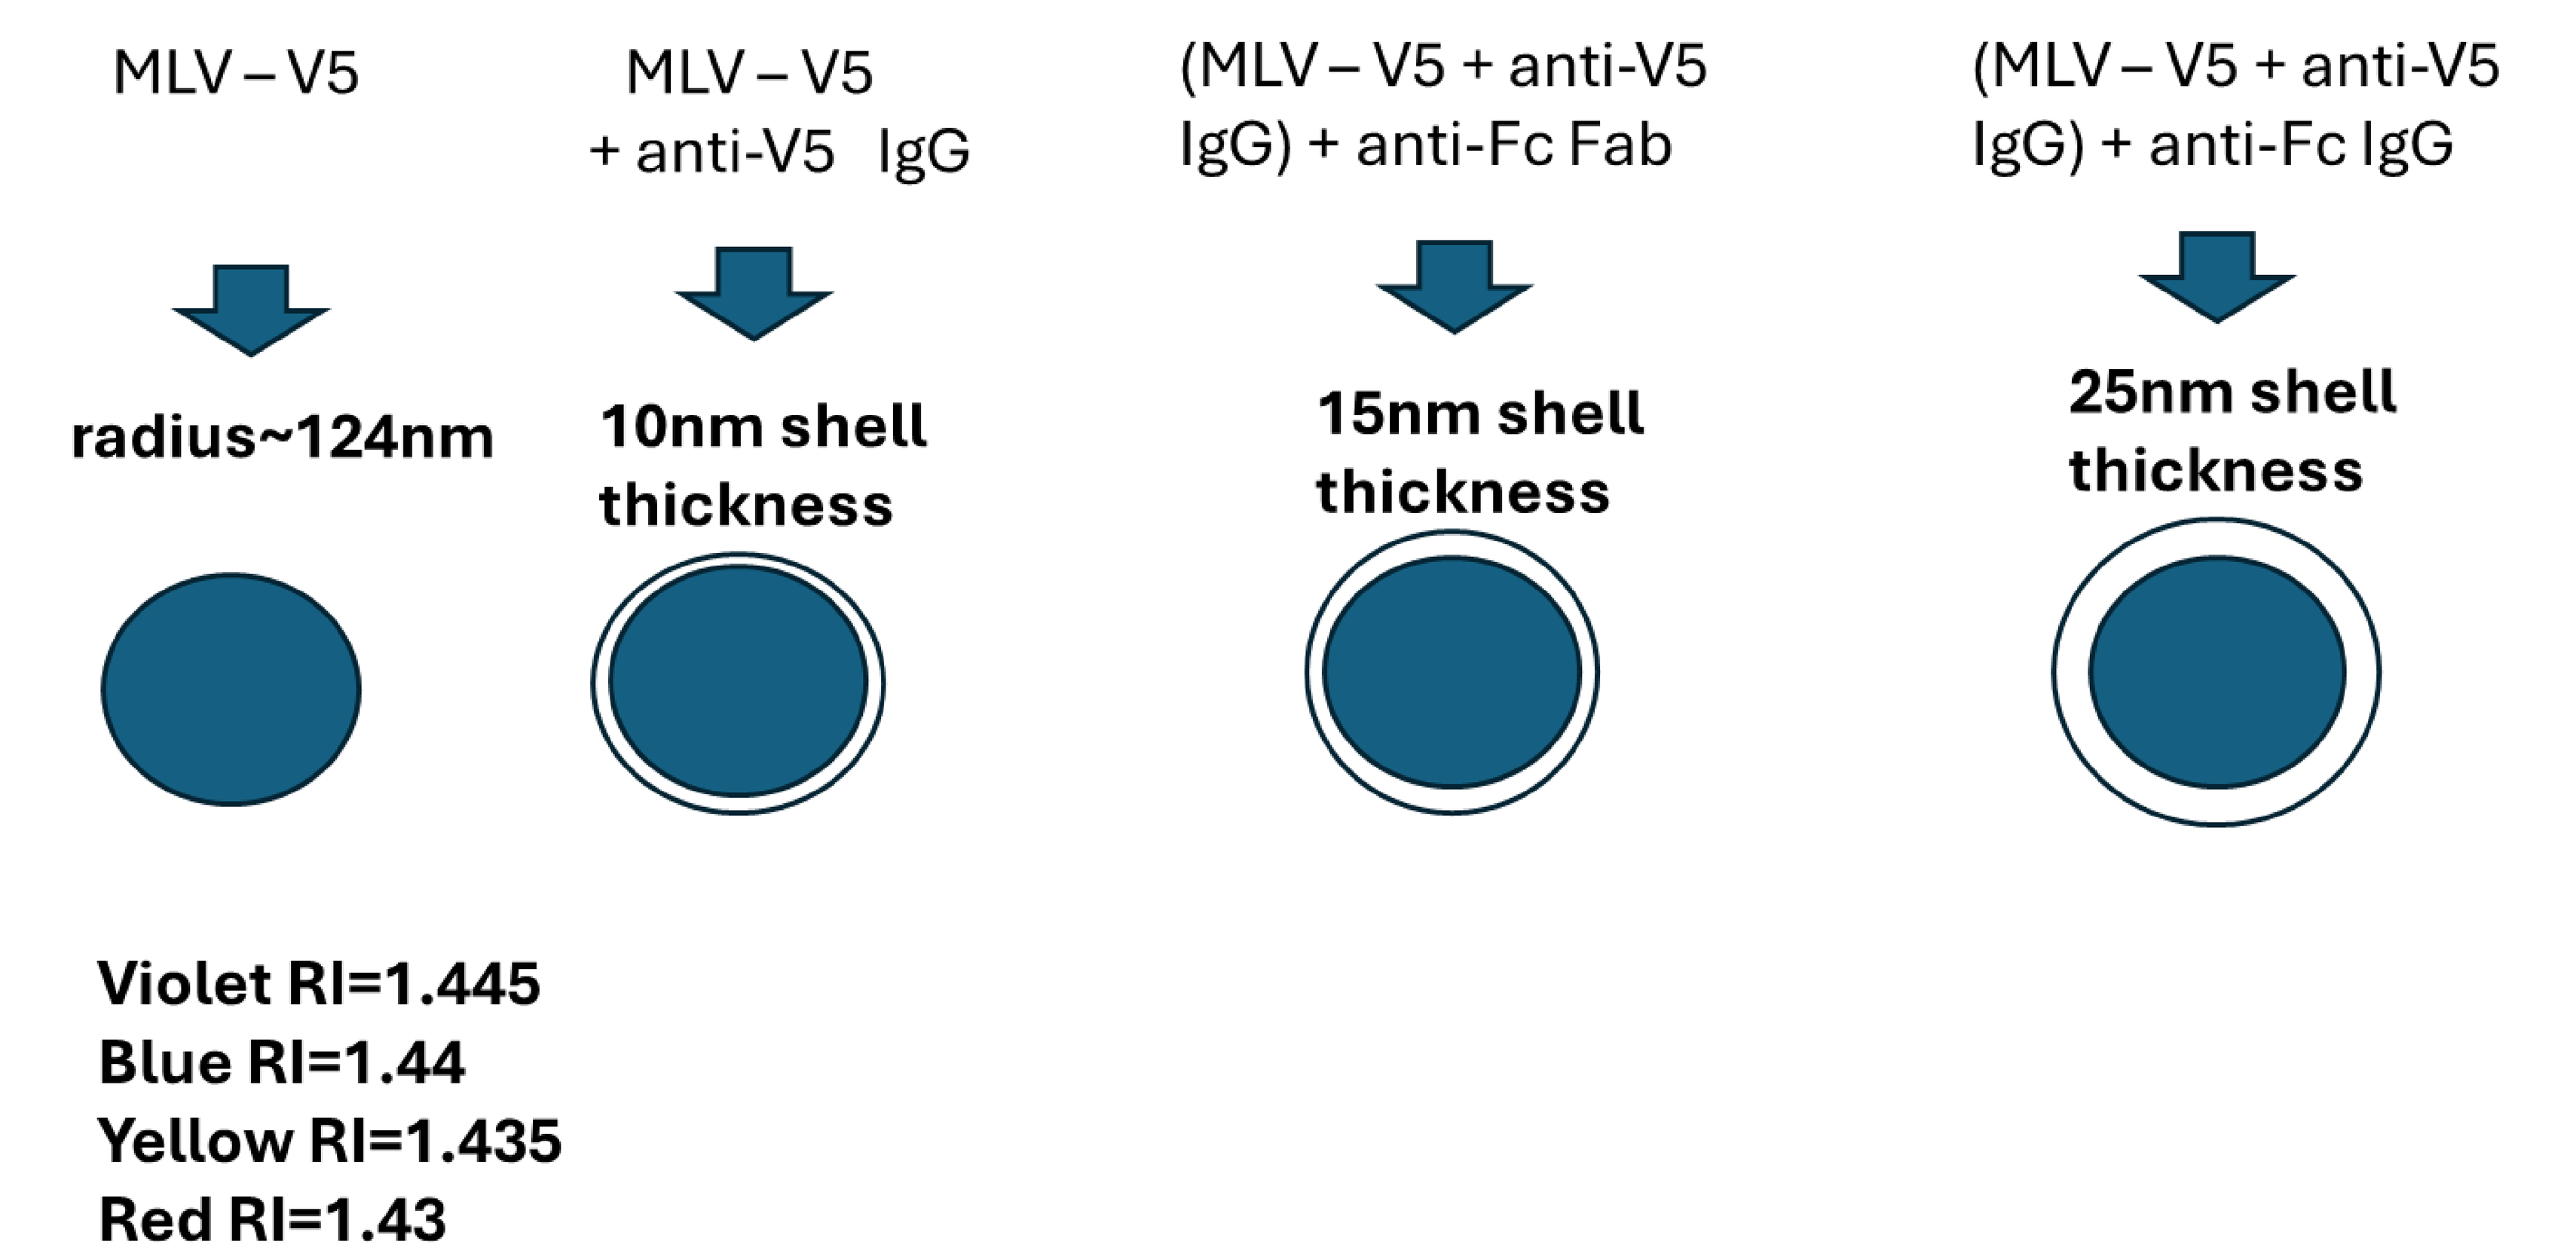
MLV – V5
MLV – V5 + anti-V5 IgG
(MLV – V5 + anti-V5 IgG) + anti-Fc Fab
(MLV – V5 + anti-V5 IgG) + anti-Fc IgG
radius~124nm
10nm shell thickness
15nm shell thickness
25nm shell thickness
Violet RI=1.445
Blue RI=1.44
Yellow RI=1.435
Red RI=1.43

Figure 6. Schematic of core-shell modeling with different shell thicknesses.

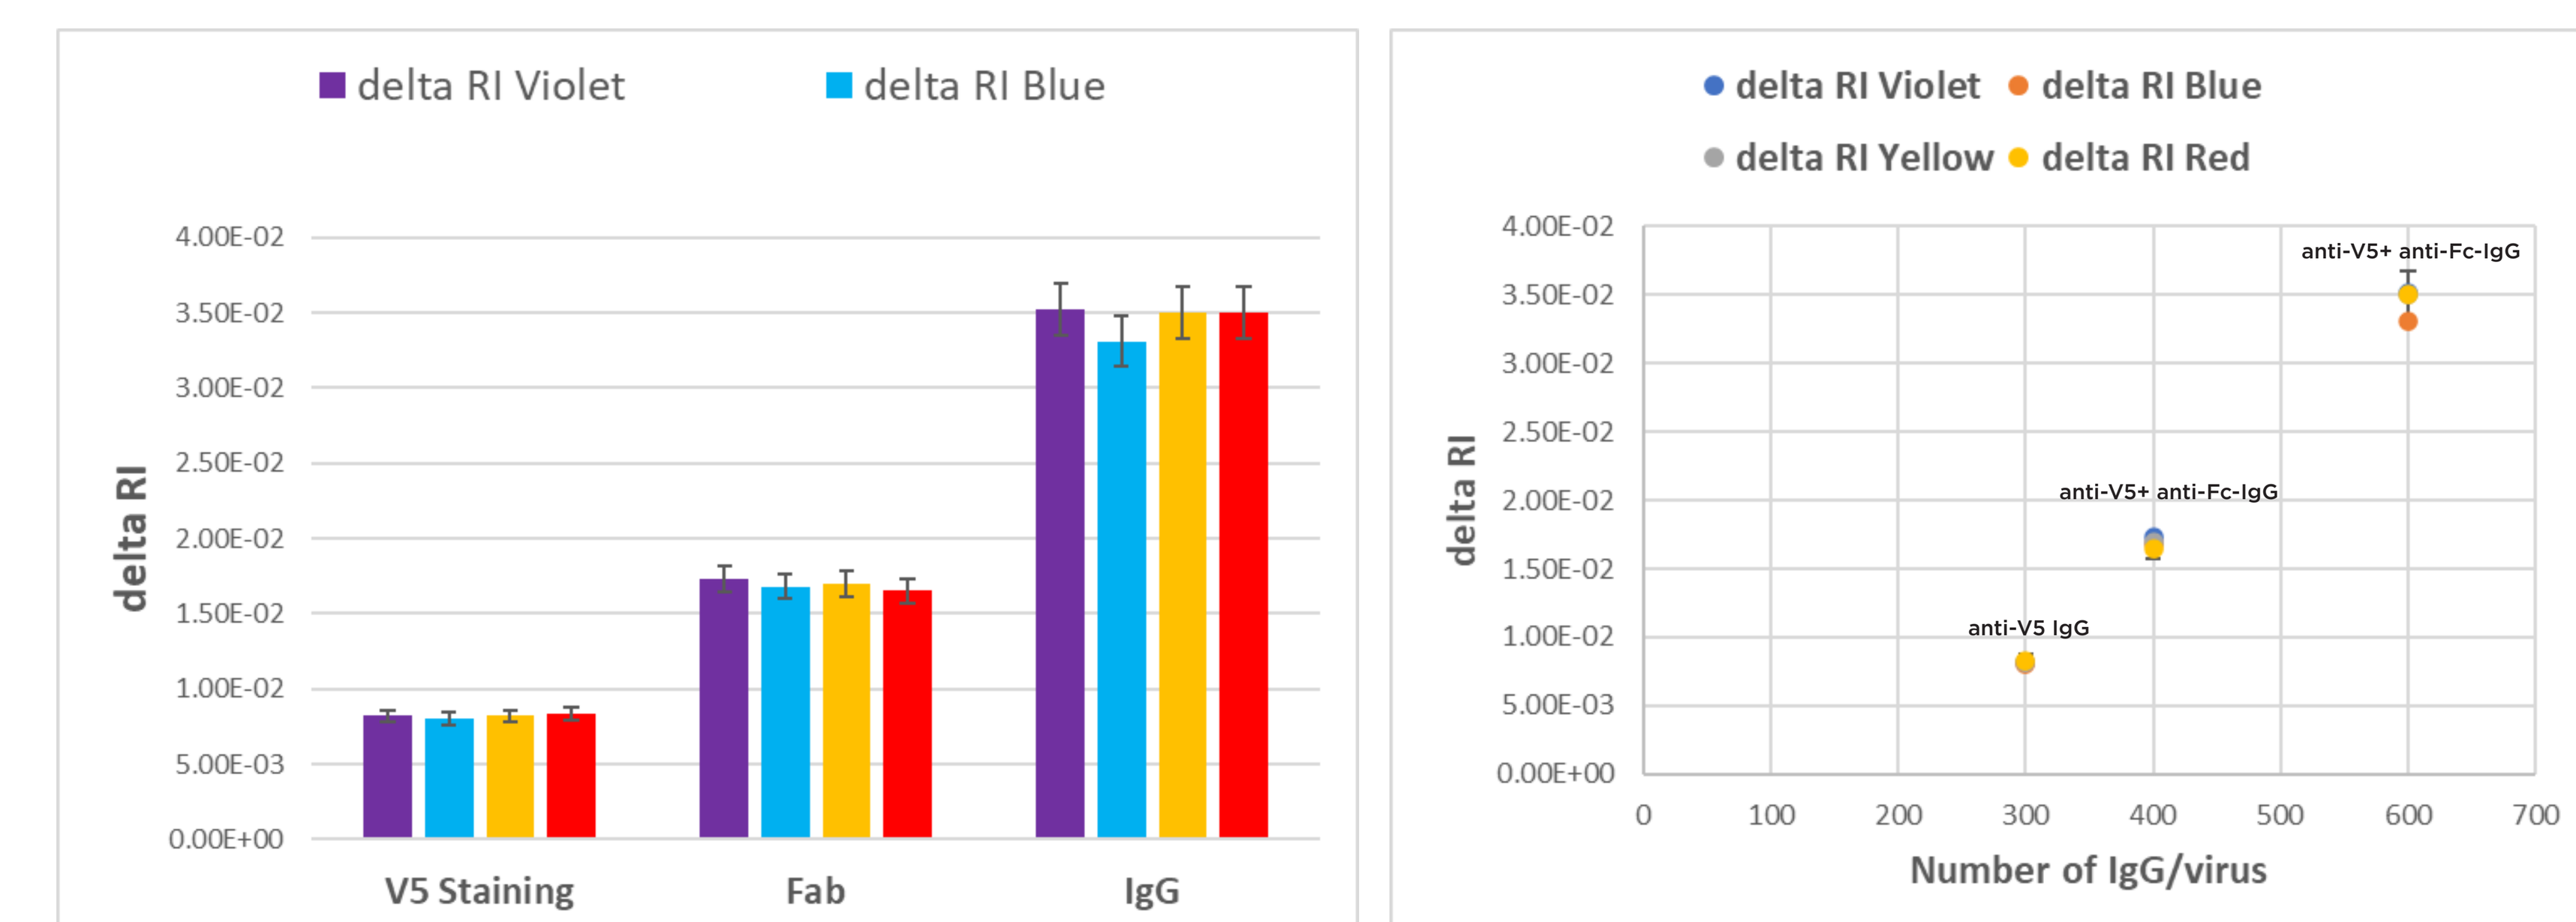
delta RI Violet
delta RI Blue
delta RI Yellow
delta RI Red
delta RI
V5 Staining
Fab
IgG
anti-V5 IgG
anti-V5+ anti-Fc-IgG
Number of IgG/virus

Figure 7. Delta refractive index change for three coatings of antibodies at four different wavelengths of scatter. (a); delta refractive index change as a function of number of IgG/virus (b).

Conclusions

We demonstrate the capabilities of the CytoFLEX nano instrument to detect and analyze the change in refractive index down to <0.01 caused by different numbers of antibodies bound to the surface of MLV.

For Research Use Only. Not for use in diagnostic procedures.
© 2025 Beckman Coulter, Inc. All rights reserved. Beckman Coulter, the stylized logo, and the Beckman Coulter product and service marks mentioned herein are trademarks or registered trademarks of Beckman Coulter, Inc. in the United States and other countries. All other trademarks are the property of their respective owners.

For Beckman Coulter's worldwide office locations and phone numbers, please visit contact us at **beckman.com**

2025-GBL-EN-107666-v4