Advancing Unmixing Algorithms in Spectral Flow Cytometry

Anisha Jose[1], Fanuel Messaggio[2], Krittika Ralhan[1]
[1]Beckman Coulter Life Sciences India Pvt Ltd, Karnataka, 562149. [2]Beckman Coulter Life Sciences, Miami, FL 33196.

Introduction

Both spectral and conventional flow cytometers can generate high-resolution data with properly optimized panels. However, as researchers aim to evaluate more parameters in the (current) panel, spectral flow cytometry offers several advantages:

Advantage of Spectral	Description
Greater Flexibility	Detects a higher number of fluorochromes simultaneously, allowing the addition of more markers to previously used multicolor panels in conventional methods.
Detailed Analysis and Higher Resolution	It captures the entire fluorescence spectrum from each fluorochrome, creating unique spectral signatures and enhancing the resolution of individual markers.
Improved Autofluorescence Management	Effectively handles autofluorescence, which can hinder data analysis in conventional flow cytometry.

Methods of Data Collection: Spectral Flow Cytometer

Advantage of Spectral
Utilizes signals from all detection channels. With detectors typically exceeding the number of fluorochromes, each dye is measured across an entire spectrum from 351 nm to 950 nm.
Each dye, such as dye 1, uses all detection channels to produce a spectral emission signal called as reference spectrum for dye 1.
Uses reference spectrum for dye1, in the spectral unmixing calculation.

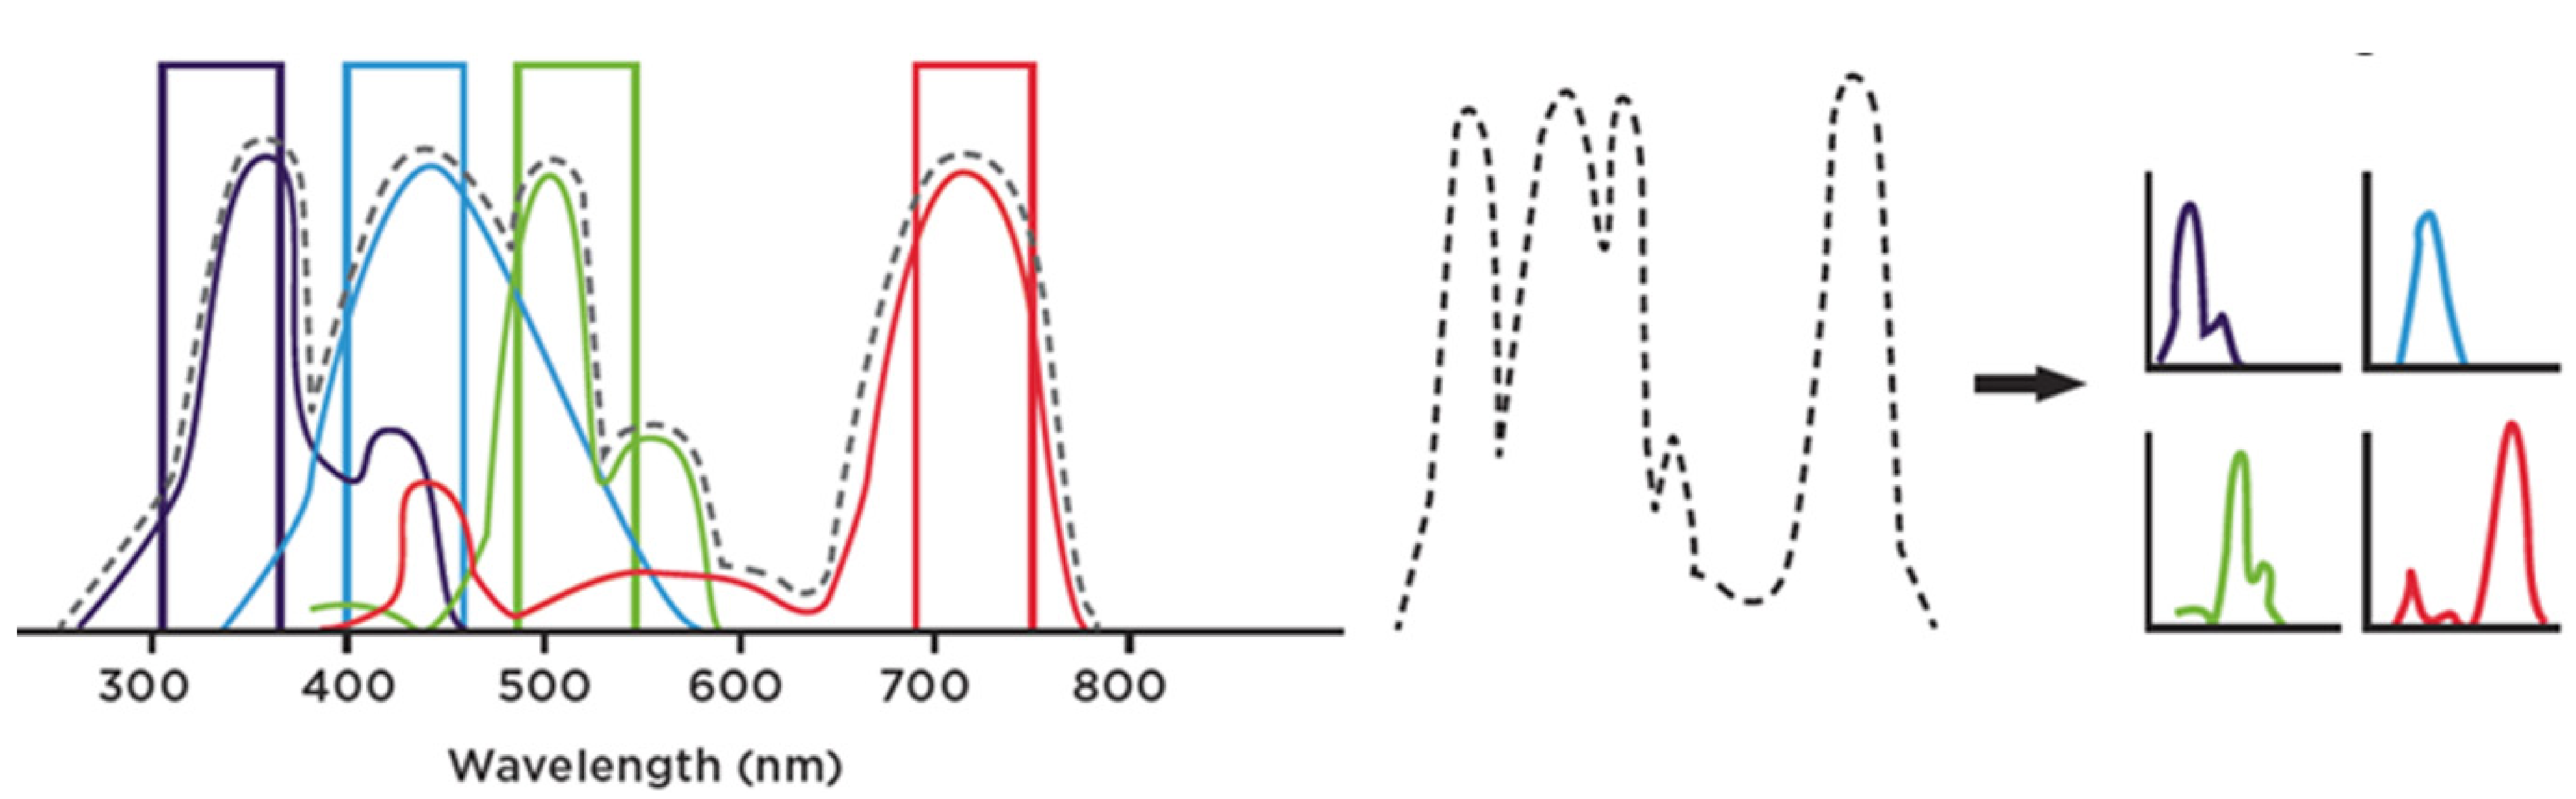
300
400
500
600
700
800
Wavelength (nm)

Unmixing in Spectral Flow Cytometry: Spectral flow cytometers use a detector array to capture the full spectral signature of all fluorochromes. These combined signals create a single complex waveform, which is then separated into individual fluorochrome signatures using an unmixing algorithm

Mathematics of Compensation and Unmixing, and Noise Management

- The goal of this approach is to reduce noise in dye intensity measurements.
- In flow cytometry, noise primarily originates from two sources:
 - Instrument noise, which includes electronics and optics.
 - Biological variation, due to differences in emission between individual cells from sample and single stained control, following Poisson statistics, where emissions are never same.

Compensation	Spectral Flow Cytometer
The number of detectors matches the number of dyes, resulting in a method that typically provides a unique solution by solving a set of mathematical equations. Hence, this compensation model is called a full-rank system of equations.	Spectral analysis accommodates more detectors than dyes, resulting in an overdetermined system. Consequently, the values we determine for unmixed fluorescence depend on the chosen unmixing algorithm used to solve the equations and obtain optimal results.

Mathematics of Compensation and Unmixing, and Noise Management

Unmixing Algorithm	Features
Least square method (LSM)	• Simplest and most common method • Assumes mean of noise is constant and same variance across all detectors, regardless of signal level • In practical scenario this mismatch can cause population distortion (spreading)
Weighted least square method (WLSM)	• Extension of LSM • Gives more weight to some errors/noises over others • Weights are inversely proportional to the error variance • Improves accuracy, especially in data with varying variability
Poisson	• Unmixing approach incorporates the Poisson nature of signal and noise • It can help in distinguishing true signals from background noise, useful where the signal-to-noise ratio is low
Poisson hybrid	• Combination of Poisson and LSM • It takes LSM's result as the initial estimated parameter and incorporates the Poisson nature of signal and noise • Improved efficiency and accuracy in data with varying variability

Unmixing Algorithm in CytoFLEX mosaic Spectral Detection Module

Our approach includes two algorithms: LSM and our Poisson hybrid. LSM is well-known in the industry. The Poisson hybrid is useful for larger panels with multiple fluorochromes, overlapping dyes, or varied sample types, even when antibodies are not fully titrated. It may reduce spreading errors in these scenarios. We suggest that users try both unmixing algorithms to find the one that best suits their specific needs and experimental conditions.

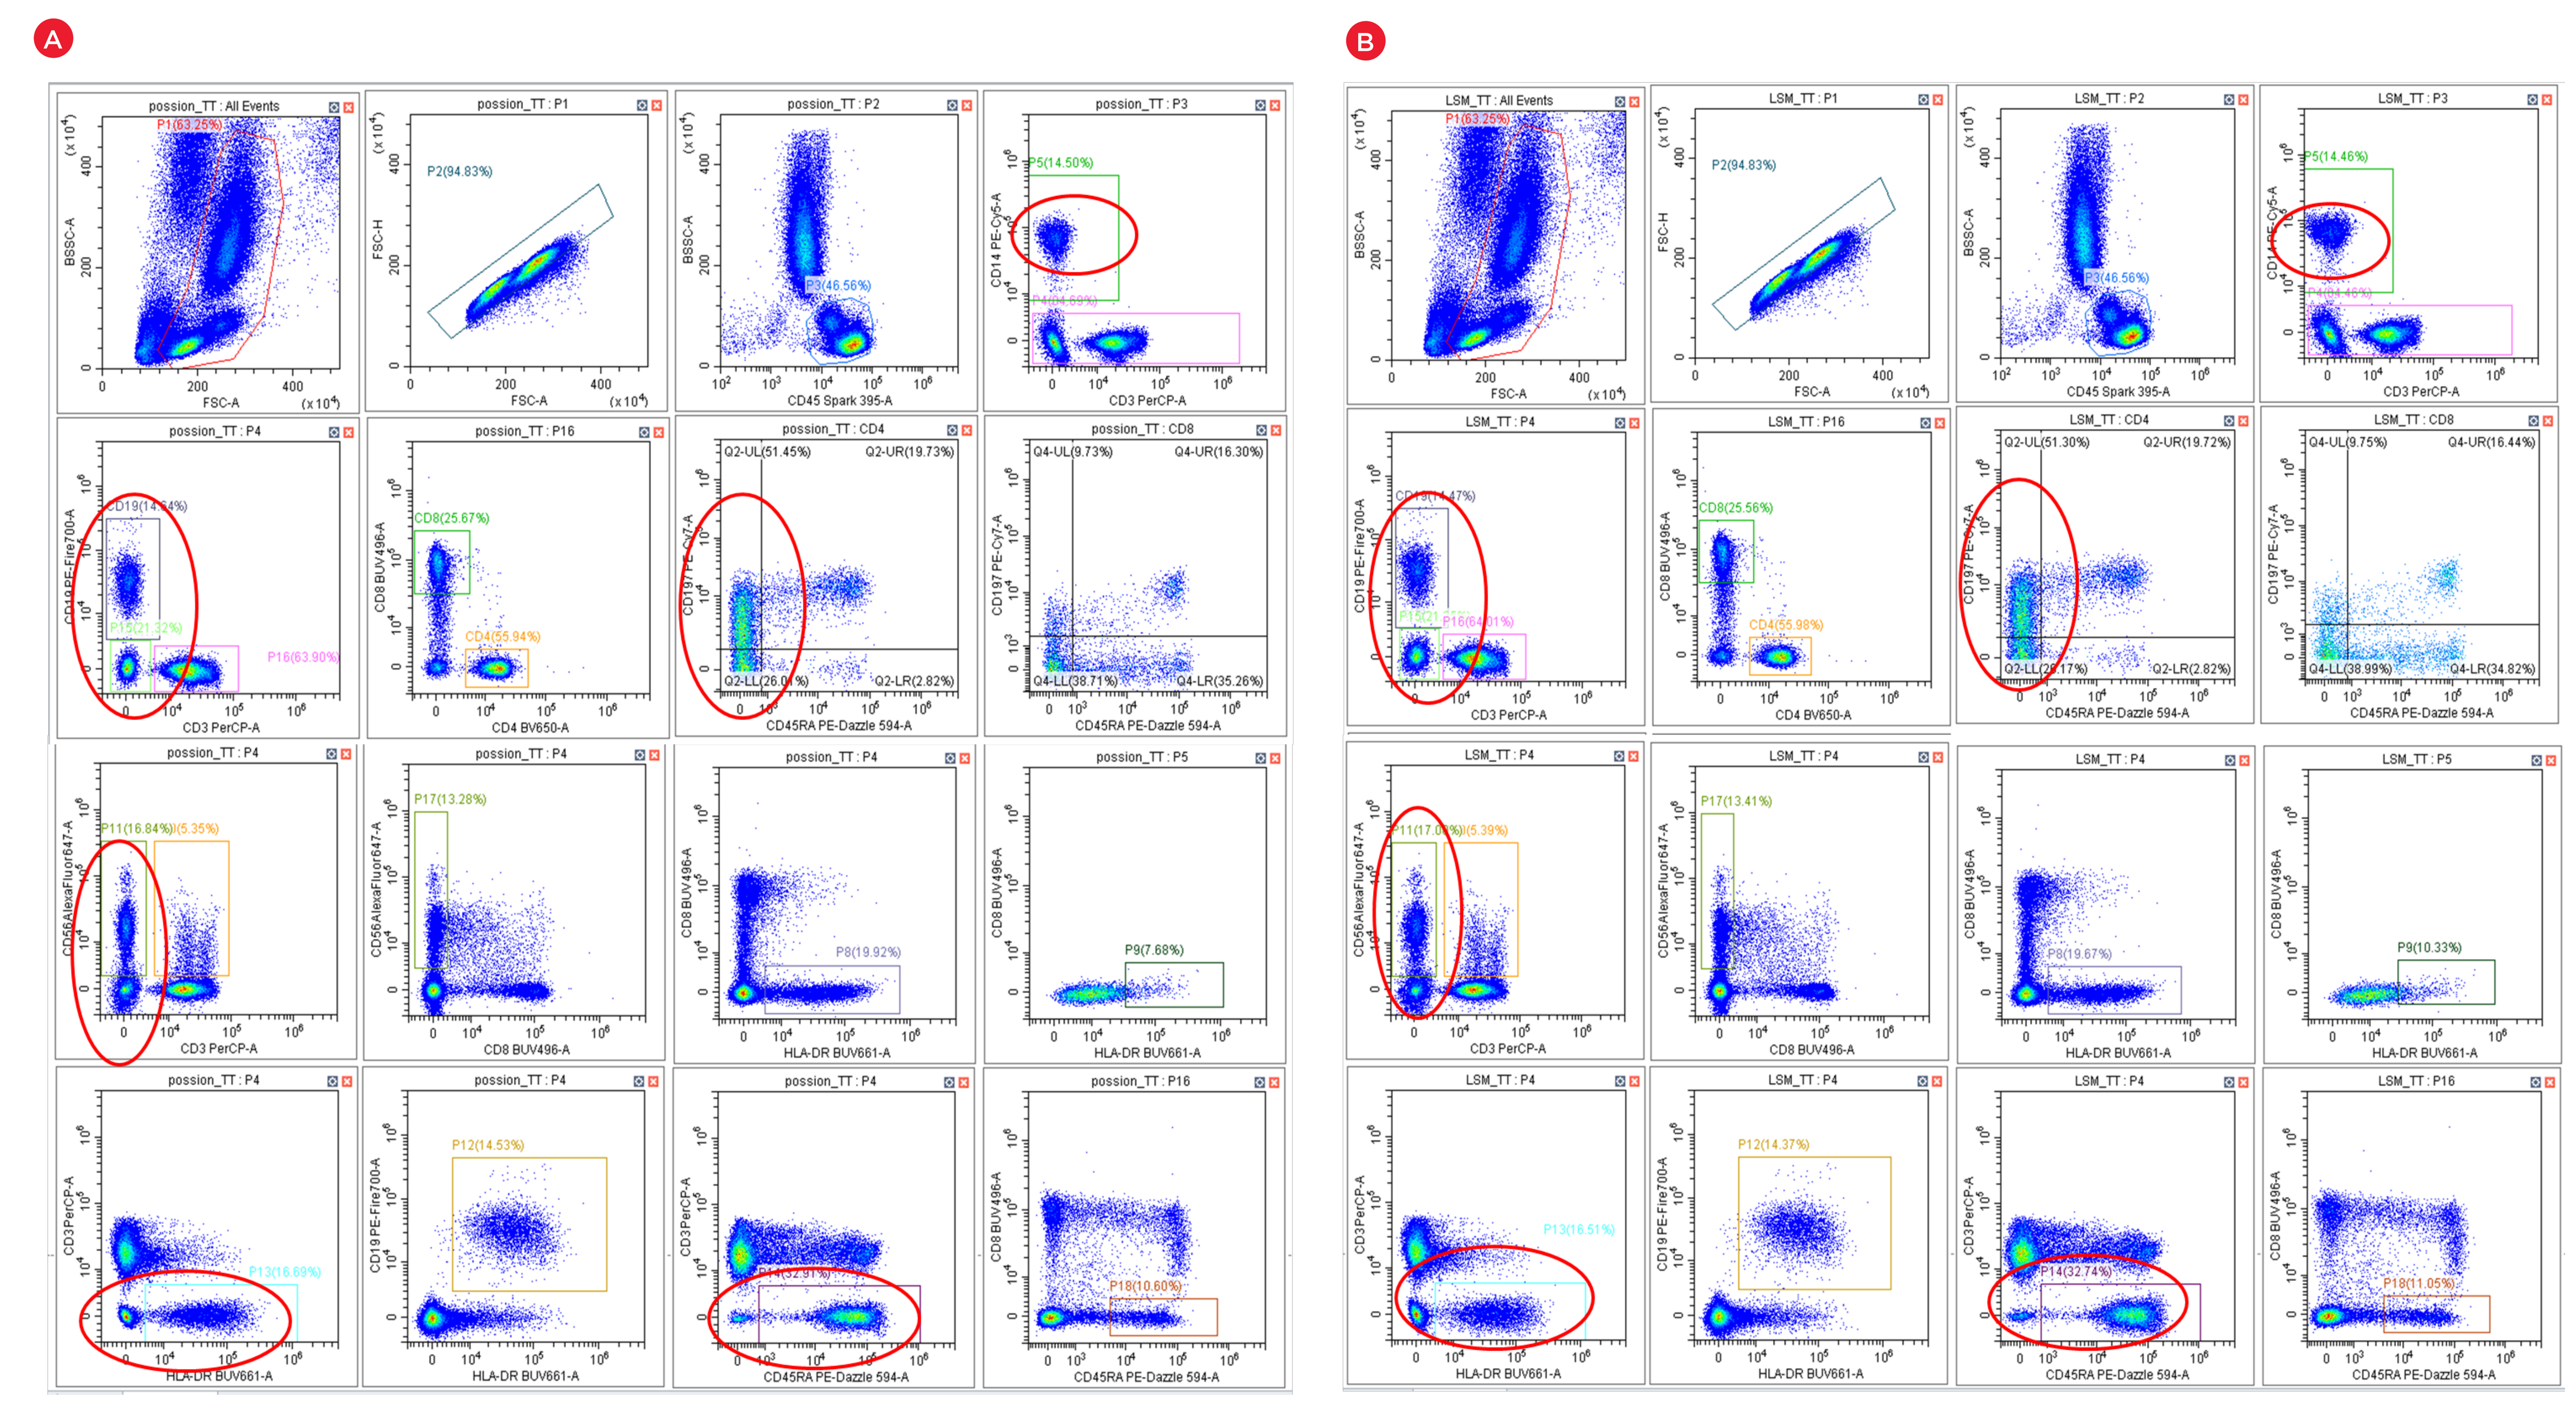
A
B

Figure 1: Comparison of Poisson hybrid and LSM Algorithms in Spread Error Reduction. A 10-marker panel was used to detect major cell subpopulations in human peripheral blood, unmixed using Poisson hybrid and LSM algorithms. Our findings show that the Poisson hybrid algorithm (Figure 1A) reduces spread error and enhances signal resolution compared to LSM (Figure 1B). Note that results may vary with larger panels, overlapping dyes, or different samples, regardless of antibody titration. Users should switch between algorithms to find the best fit for their specific needs and conditions.

AlexaFluor (“AF”) is a trademark or registered trademark of Thermo Fisher Scientific Inc. Brilliant Ultraviolet (“BUV”) and PerCP are trademarks or registered trademarks of Becton, Dickinson and Company. PE-Fire and PE-Dazzle are trademarks or registered trademarks of BioLegend. PE-Cy5 is a trademark or registered trademark of GE Healthcare.

Summary

Spectral flow cytometry analyzes multiple cellular features in one experiment using various fluorochromes. It employs spectral unmixing to separate these signals and create a matrix estimating each fluorochrome's abundance on a cell. Accurate results require good reference controls and an understanding of the unmixing algorithm, which should accurately reflect the physics of signal formation.

For Research Use Only. Not for use in diagnostic procedures.
© 2025 Beckman Coulter, Inc. All rights reserved. Beckman Coulter, the stylized logo, and the Beckman Coulter product and service marks mentioned herein are trademarks or registered trademarks of Beckman Coulter, Inc. in the United States and other countries. All other trademarks are the property of their respective owners.

For Beckman Coulter’s worldwide office locations and phone numbers, please visit Contact Us at beckman.com

2025-GBL-EN-107735-v1